Poster # 732

Immune Activation with Plinabulin Enhances Anti-tumor Response Combining Radiation with Immune Checkpoint Blockade

Steven H. Lin, MD, PhD[1], Evan Cohen, PhD[2], Ziyi Li, PhD[3], James Reuben, PhD, MBA[2], Hua Gao, PhD[2], Huiying Sun, MS[4], June Lu, PhD[5], Ken Lloyd, PhD[5], James Tonra, PhD[5], Lan Huang, PhD[5], Siqing Fu, MD, PhD[4], and Vivek Subbiah, MD[4].

Departments of Radiation Oncology[1], Hematopathology[2], and Biostatistics and Quantitative Sciences[3], The University of Texas M. D. Anderson Cancer Center, Houston Texas 77030 USA

Beyond Spring Pharmaceuticals, Inc.[4], New York, NY 10005 USA

Abstract

Background

Plinabulin is a selective immunomodulating microtubule-binding agent that exerts direct anti-cancer activity as a single agent, as well as enhancing the immune response, primarily by inducing dendritic cell (DC) maturation and T-cell activation. Radiation can liberate local antigen release, and when coupled to dendritic cell maturation, can potentiate systemic immunity with immune checkpoint blockade, even in the immunotherapy refractory setting. We set out to test this hypothesis preclinically and clinically in a phase I basket study in immunotherapy refractory cancers.

Methods

In vitro assays involved combining plinabulin with radiation in different timings before and after radiation. DC activation was assessed with flow cytometry evaluating CD80/CD86/MHC-II expression. Using syngeneic TSA breast cancer model, we irradiated tumors with 8Gy x 3, and treated them with or without plinabulin and/or αPD1. We initiated an open-label, single-center, phase I study to evaluate the safety of plinabulin in combination with radiation and immunotherapy in patients with select advanced malignancies and a majority had progressed on PD-1, PD-L1 and/or CTLA-4 targeted antibodies (NCT04902040).

Results

In SP37A3 and XS106 DC lines, we found that DC maturation was enhanced by combining radiation with plinabulin, particularly when radiation was added 3-6 hours prior to plinabulin, but not when plinabulin was added first before radiation. In the TSA model, there were minimal anti-tumoral effects with αPD1 alone, plinabulin alone, and αPD1+plinabulin. However, triple therapy triggered a stronger abscopal effect than irradiation plus αPD1. The percentage of CD8+ T cells and CD86+ DCs in the tumor were significantly increased in the triple combination group that were not seen for the monotherapy or bimodal therapy groups (p<0.05, Dunnett's test). In addition, we initiated a phase I trial testing the triple combination approach and have enrolled six tumor types with ten ICI-refractory patients (PD-1/PD-L1, CTLA-4). Eight patients received at least 3 cycles with no dose-limiting toxicities. Per RECIST criteria, there were 8 responders and 2 non-responders. In responders, whole blood analyses indicate positive trends towards increased myeloid-derived DC (mDC) numbers and maturation markers (CD40/CD80/CD83/CD86), increased CCR7 expression on plasmacytoid DC (pDC), and monocyte shift from classical to inflammatory phenotype. Single-cell RNAseq analysis of tumor biopsies at pre/post treatment indicates activation of a GEF-H1-dependent immune signature in subtypes of DCs and monocyte-derived macrophages in responder patients.

Conclusions

Plinabulin in combination with radiation and immune checkpoint inhibitor (ICI) was able to induce systemic immune response in immunotherapy-refractory tumors, possibly by enhancing the coactivation of DCs to generate a systemic immune response.

Plinabulin

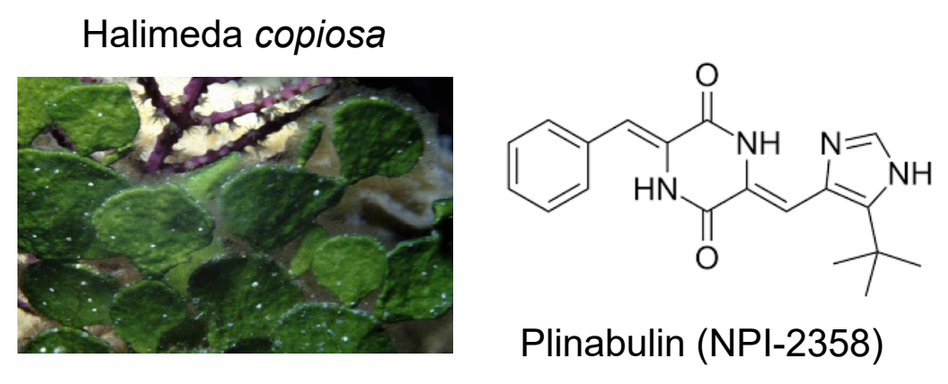
Halimeda copiosa
Plinabulin (NPI-2358)

Plinabulin is derived from the natural marine product Halimide, a fungal metabolite on green algae Halimeda copiosa.

Screening of anti-cancer effects discovers Plinabulin, a small molecule that had single agent activity against various cancer cell lines.

Plinabulin was found to bind to tubulin, at a Distinct site on beta-tubulin that is distinct from Other tubulin agents, like taxanes, vinca Alkaloids, colchicine.

Plinabulin prevents polymerization Of microtubules, which leads to GEF-H1 Release.

Liberated GEF-H1 is activated upon release Which binds and activates RhoA, leading to Downstream JNK activation.

Activation of JNK causes dendritic cell maturation In vitro, and in vivo, this boosts antigen induced CD8 T cell activation.

Plinabulin alters the tumor microenvironment to reduce tumor associated macrophages and enhance M1/M2 ratio.

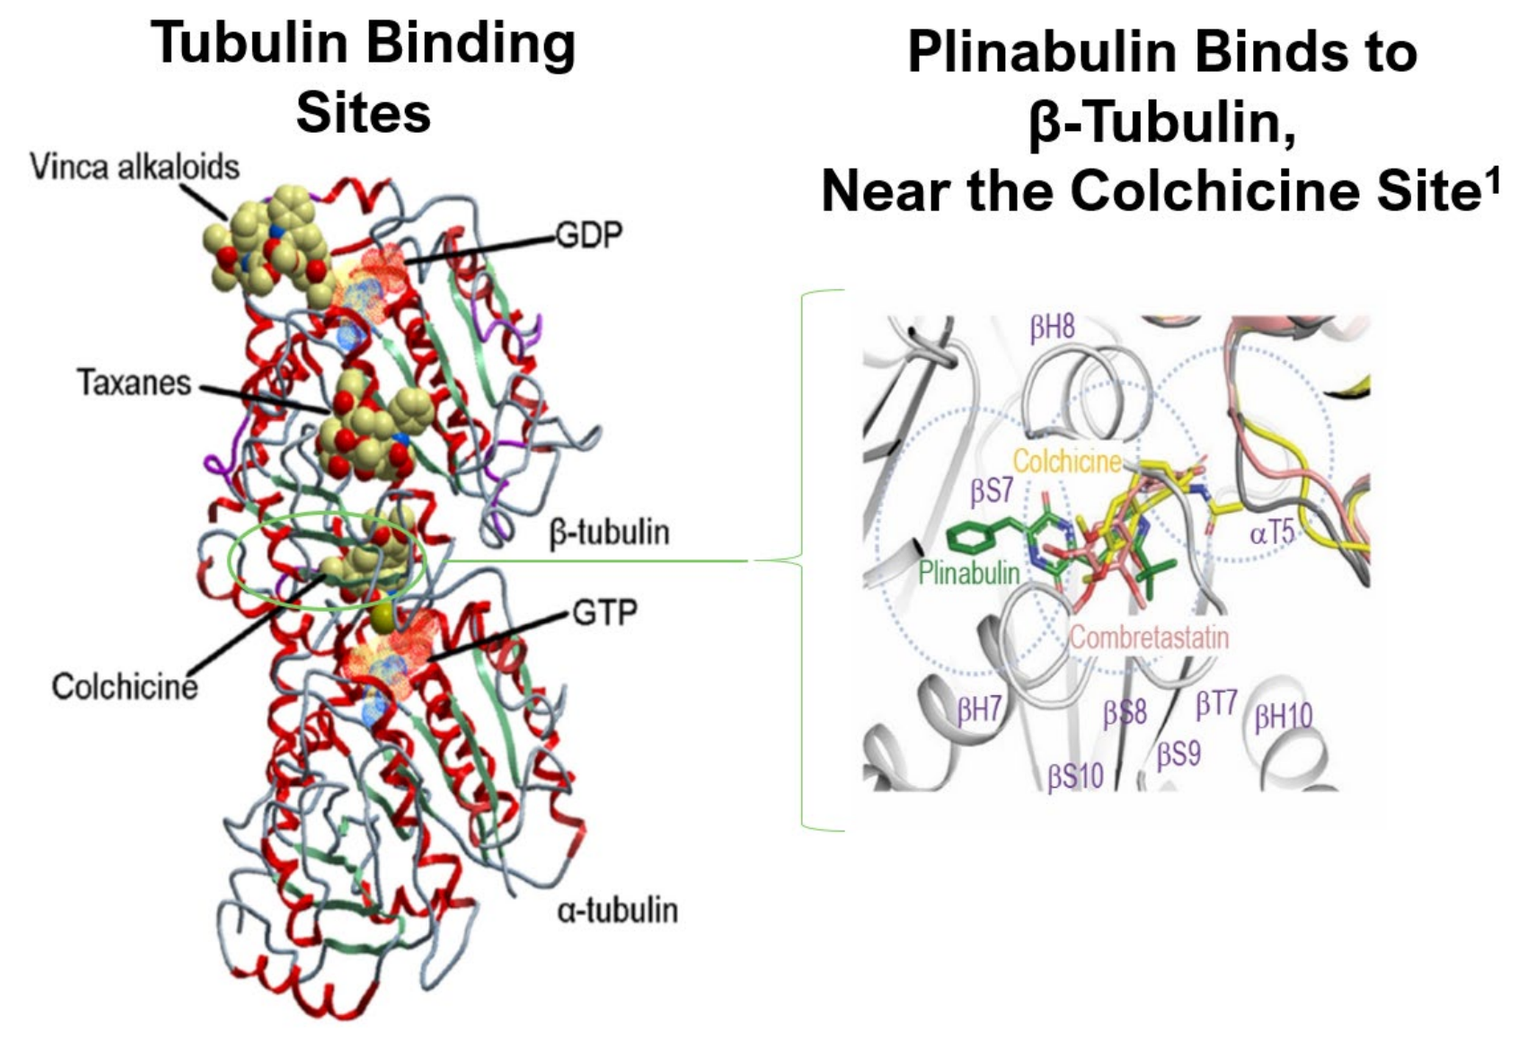
Tubulin Binding Sites
Plinabulin Binds to β-Tubulin, Near the Colchicine Site[1]
Vinca alkaloids
GDP
Taxanes
β-tubulin
GTP
Colchicine
α-tubulin

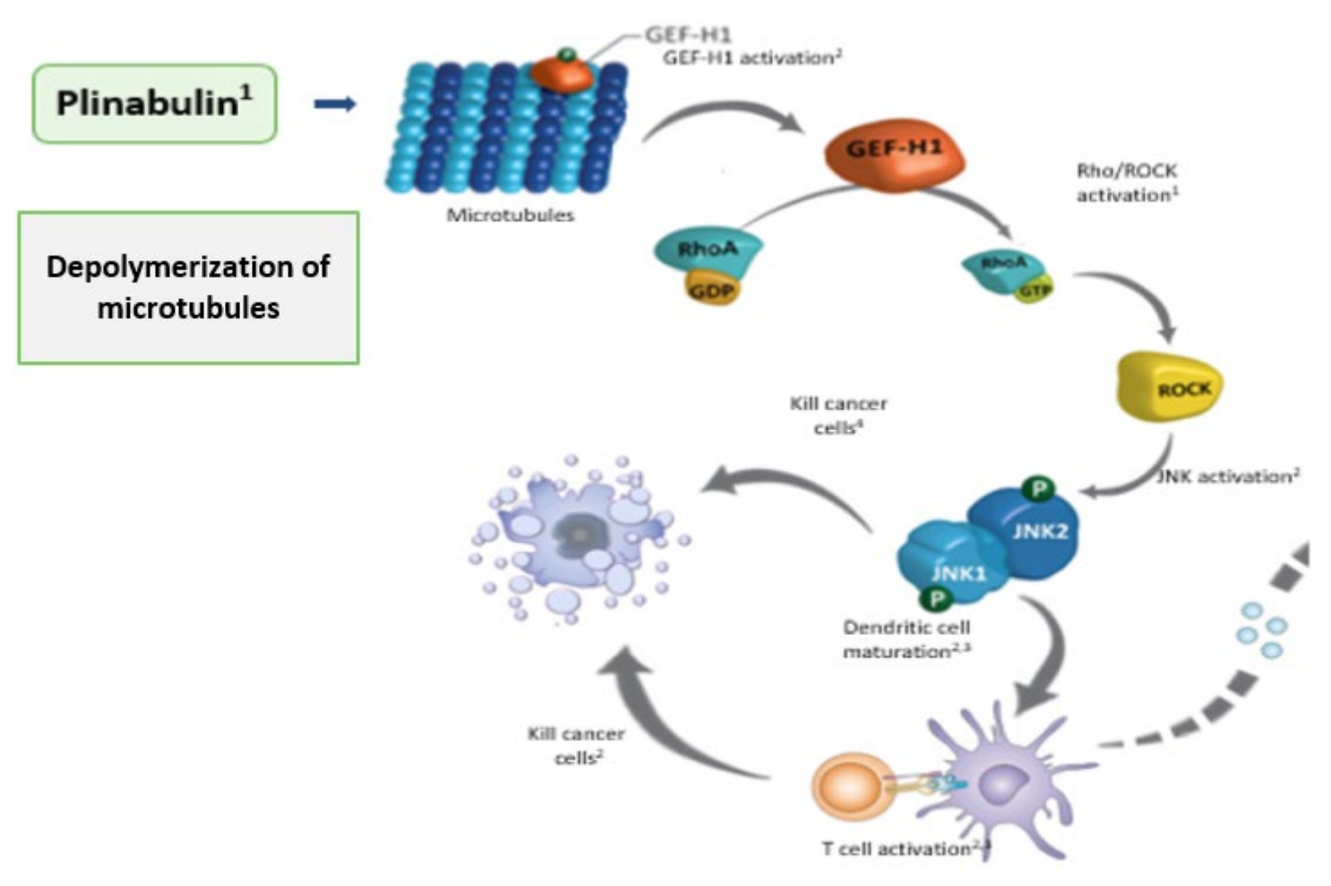
Plinabulin[1]
Depolymerization of microtubules
Microtubules
GEF-H1
ROCK
JNK1
JNK2
Dendritic cell maturation
Kill cancer cells
T cell activation

Preclinical Rationale

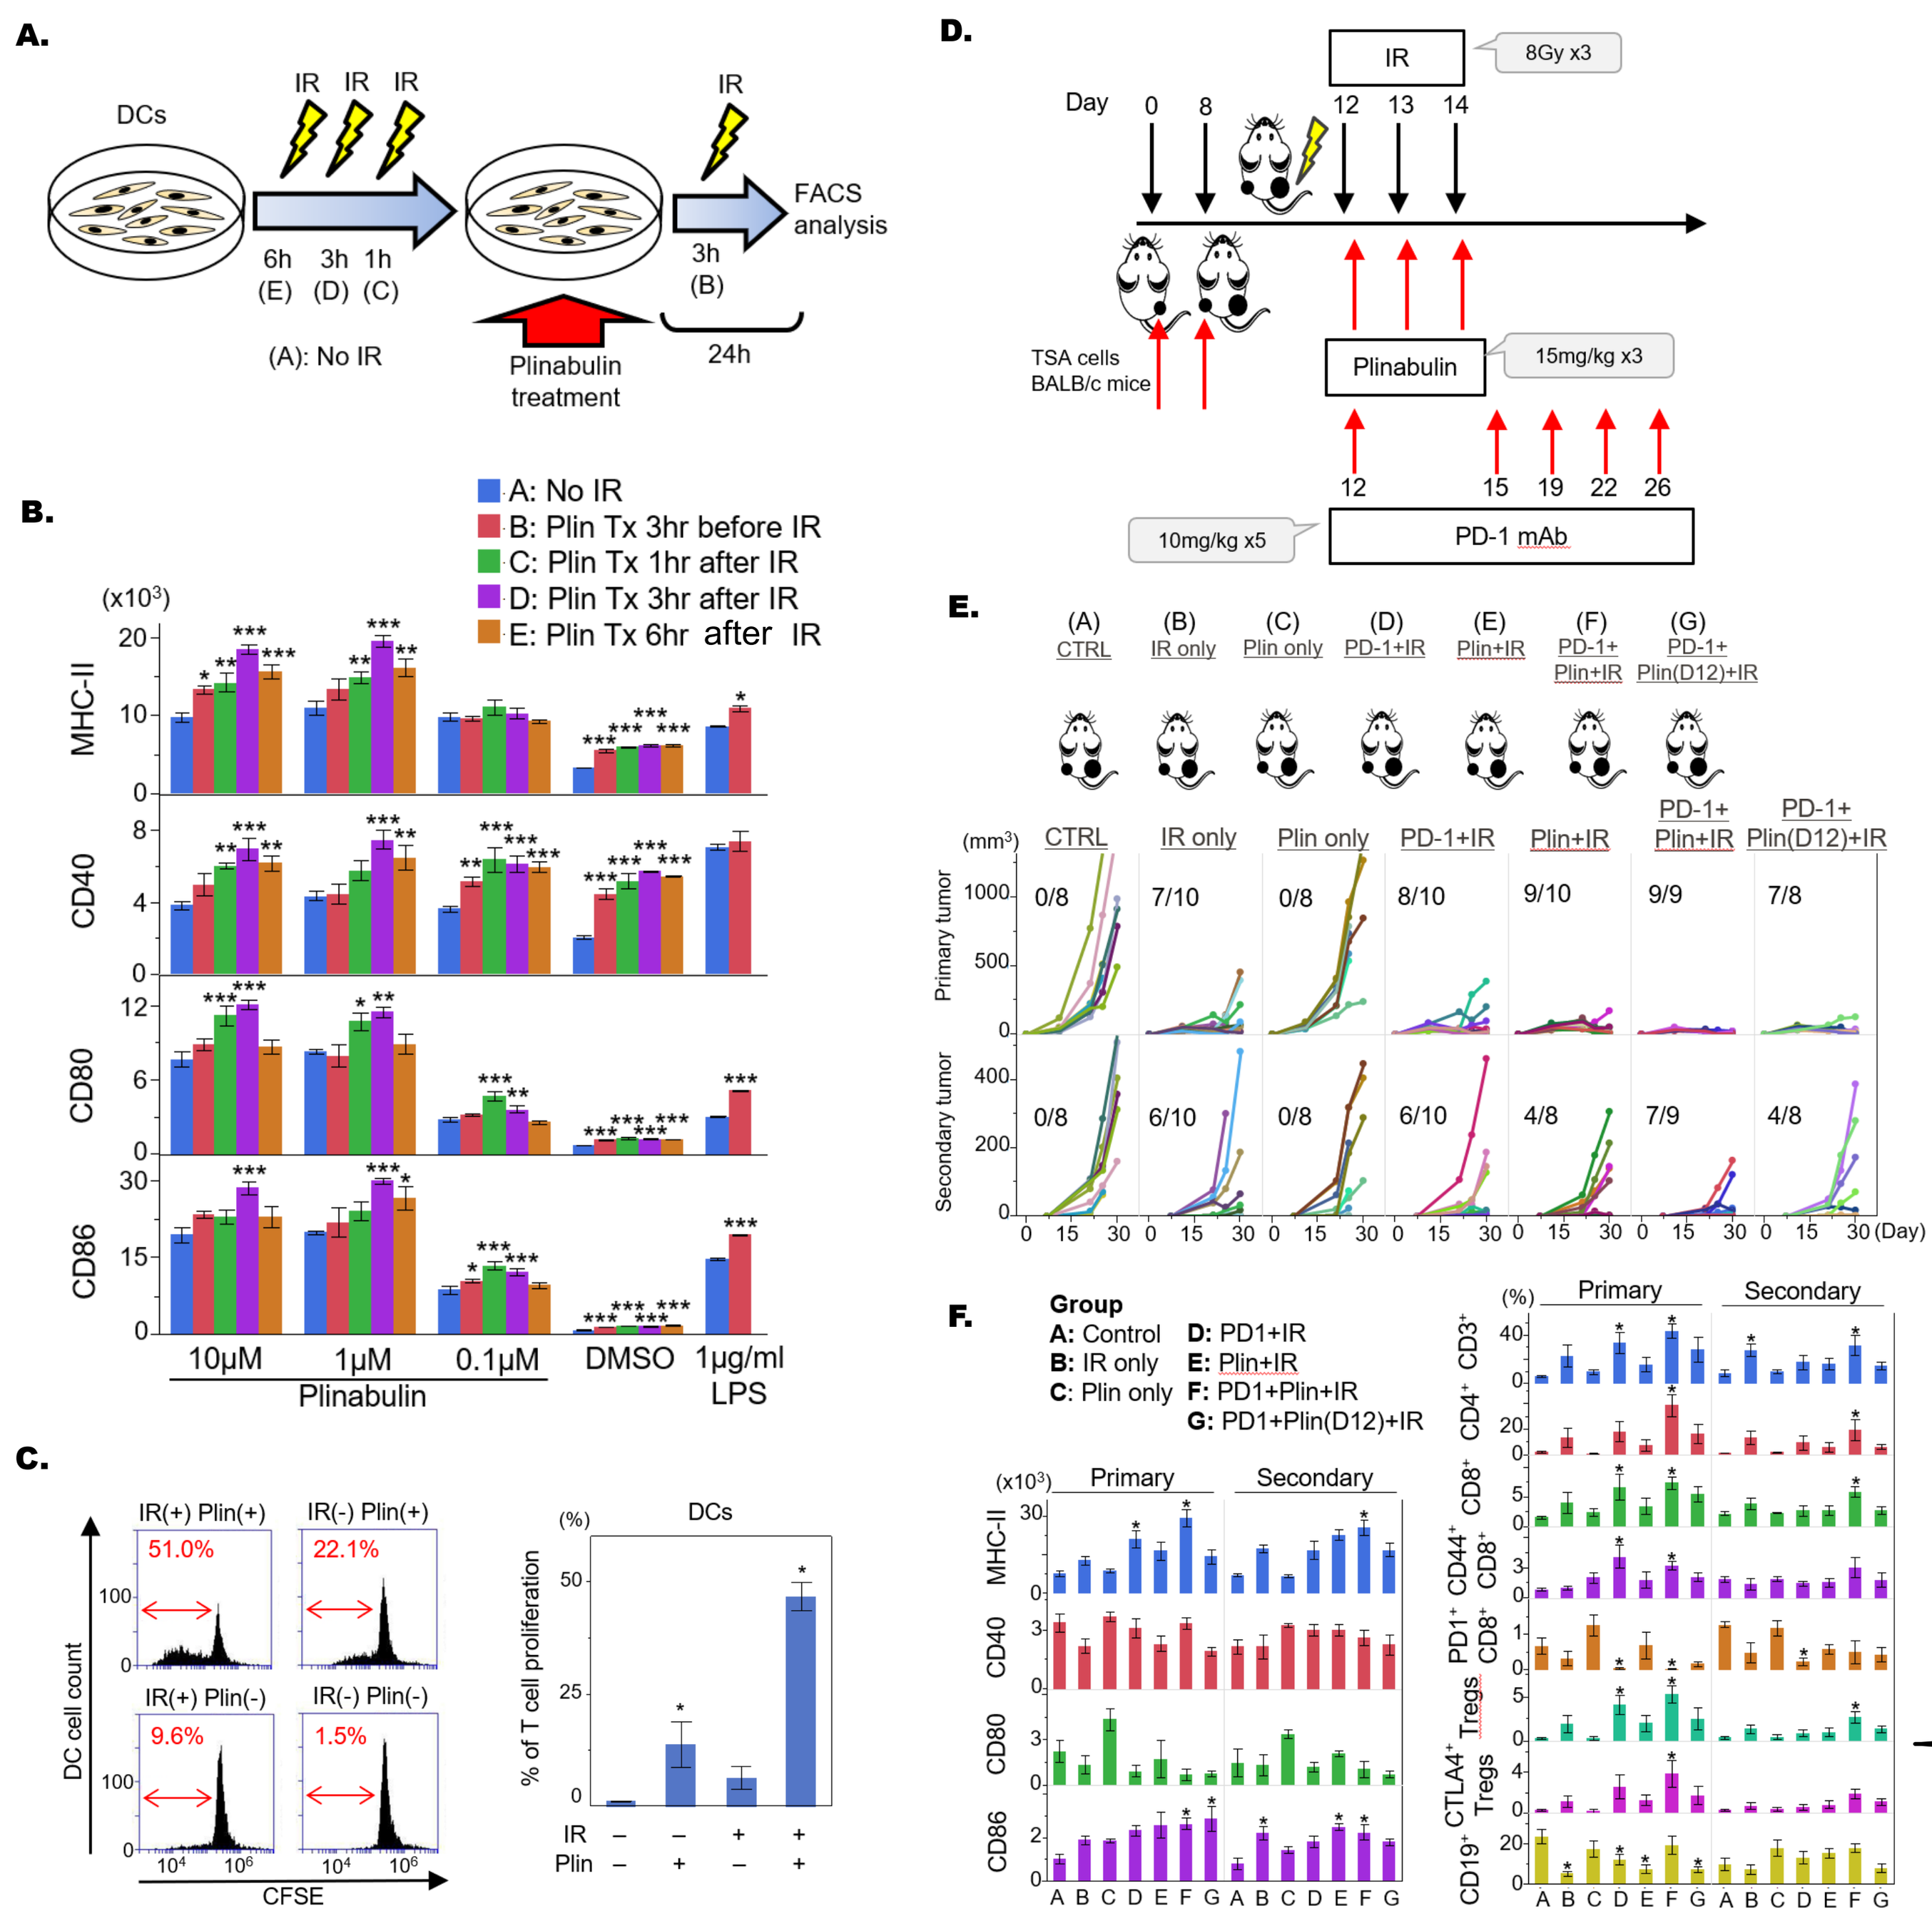
A.
DCs
IR IR IR
6h 3h 1h
(E) (D) (C)
(A): No IR
Plinabulin treatment
3h (B)
24h
FACS analysis
B.
A: No IR
B: Plin Tx 3hr before IR
C: Plin Tx 1hr after IR
D: Plin Tx 3hr after IR
E: Plin Tx 6hr after IR
MHC-II
CD40
CD80
CD86
10μM 1μM 0.1μM DMSO 1μg/ml
Plinabulin LPS
C.
IR(+) Plin(+) 51.0%
IR(-) Plin(+) 22.1%
IR(+) Plin(-) 9.6%
IR(-) Plin(-) 1.5%
DC cell count
CFSE
DCs
% of T cell proliferation
D.
IR 8Gy x3
Day 0 8 12 13 14
TSA cells BALB/c mice
Plinabulin 15mg/kg x3
10mg/kg x5
PD-1 mAb
E.
(A) CTRL (B) IR only (C) Plin only (D) PD-1+IR (E) Plin+IR (F) PD-1+ Plin+IR (G) PD-1+ Plin(D12)+IR
Primary tumor
Secondary tumor
F.
Group
A: Control D: PD1+IR
B: IR only E: Plin+IR
C: Plin only F: PD1+Plin+IR
G: PD1+Plin(D12)+IR
Primary
Secondary

Preclinical PoC of plinabulin combinatorial effect with radiation and immune checkpoint blockade.

(A) Scheme representing the *in vitro* DC stimulation assay with Plinabulin administration 6 h, 3 h, 1h after irradiation or 3 h before irradiation.
(B) Mean fluorescence intensity (MFI) levels of surface expressions of MHC-II, CD40, CD80 and CD86 on XS106 DCs using flow cytometry (gating: CD45+CD11c+).
(C) Representative histograms of CSFE(+) PI(-) CD3+ T cell count co-cultured with DCs. Percentages of T cell proliferation co-cultured with DCs. Error bars indicate ±SD, *p < 0.05.
(D) Design of combination therapy of irradiation, Plinabulin and PD-1 mAb inhibits tumor growth and induces MHC-II expression on DCs and T cell proliferation in TS/A tumor-bearing mice.
(E) Tumor growth delay of the various combination groups, showing that triple combination strategy has the greatest anti-tumor and abscopal effect.
(F) MFI levels of MHC-II, CD40, CD80 and CD86 expressions on CD11c+ DCs in the tumor. (right panel) Percentages of lymphocytes to live cells in the tumor. Error bars indicate ±SEM, *p < 0.05, **p < 0.01, ***p < 0.001.

Phase 1 Trial Schema (NCT04902040)

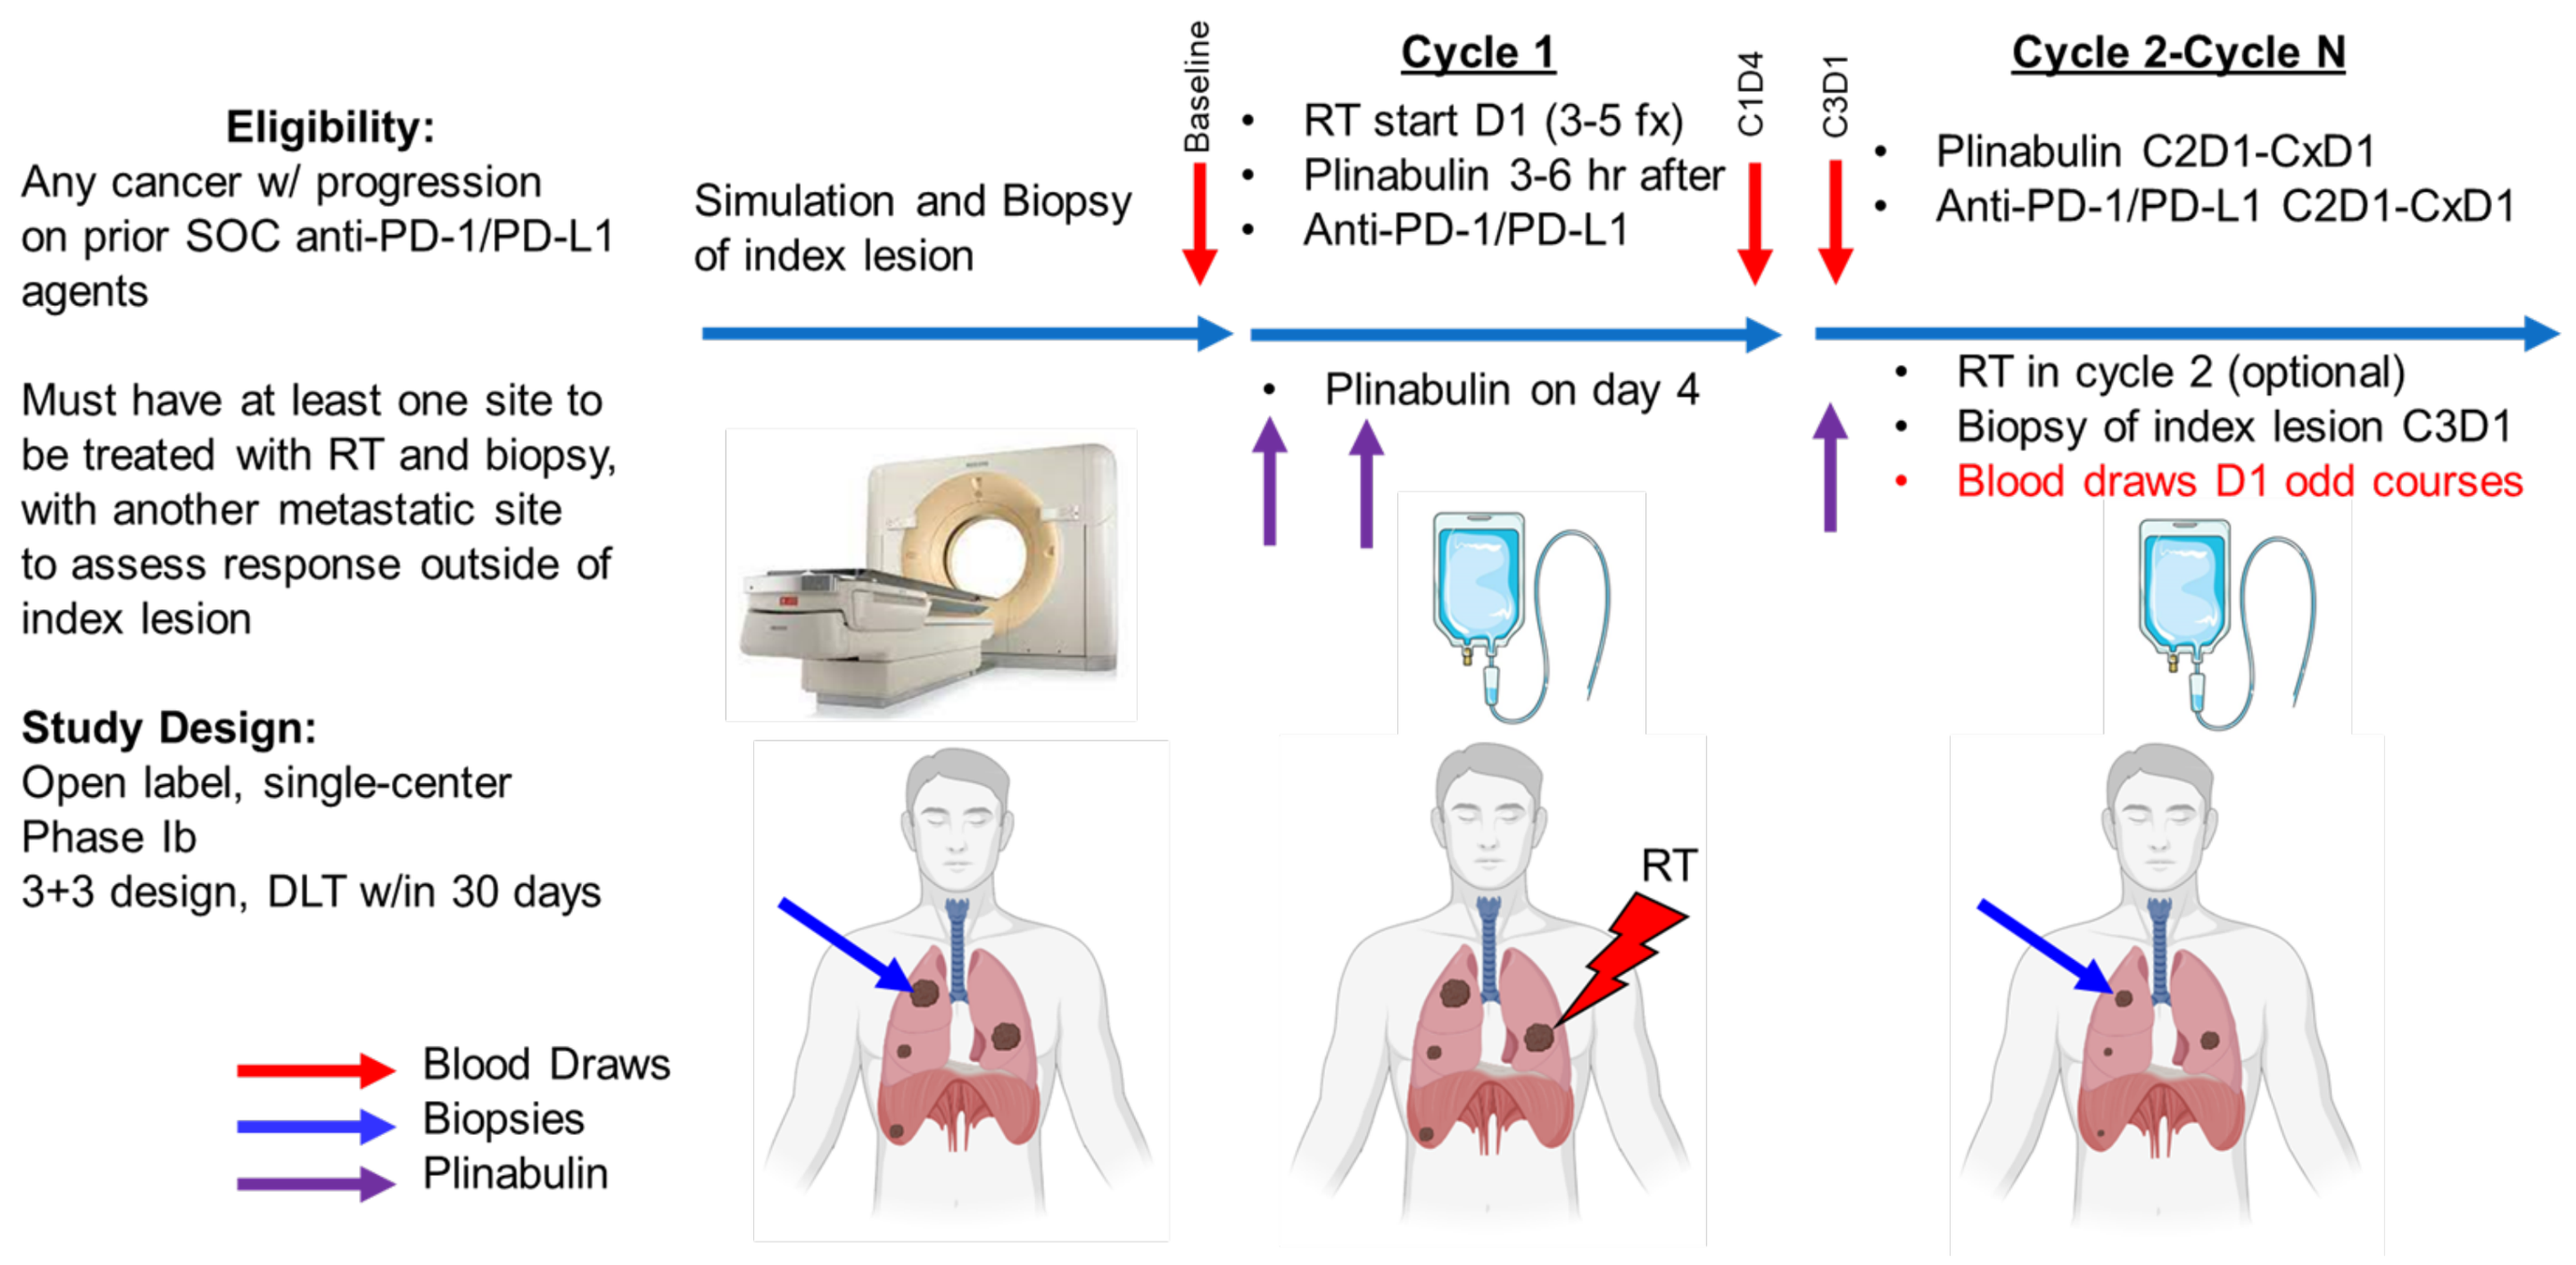
Eligibility:
Any cancer w/ progression on prior SOC anti-PD-1/PD-L1 agents
Must have at least one site to be treated with RT and biopsy, with another metastatic site to assess response outside of index lesion
Study Design:
Open label, single-center
Phase Ib
3+3 design, DLT w/in 30 days
Simulation and Biopsy of index lesion
Baseline
Cycle 1
RT start D1 (3-5 fx)
Plinabulin 3-6 hr after
Anti-PD-1/PD-L1
C1D4
C3D1
Cycle 2-Cycle N
Plinabulin C2D1-CxD1
Anti-PD-1/PD-L1 C2D1-CxD1
Plinabulin on day 4
RT in cycle 2 (optional)
Biopsy of index lesion C3D1
Blood draws D1 odd courses
RT
Blood Draws
Biopsies
Plinabulin

Primary Objectives

- To assess the safety and tolerability of plinabulin when administered in combination with a radiation/immunotherapy regimen in subjects with select advanced solid malignancies after progression on anti-PD-1/PD-L1 mAb
- To assess the objective tumor response rate (ORR) (complete response + partial response)

Secondary Objectives

- To assess disease control rate (complete response, partial response + stable disease)
- To determine progression-free survival (PFS)
- To assess overall survival (OS)

Exploratory Objectives

- Immune repertoire TCR sequencing change in peripheral blood
- Immune phenotypes in tumor tissue, including DC, T cells, TAMs, pre and post treatment
- Phenotypes of Immune cells in peripheral blood
- Dendritic cell activation in peripheral blood
- To explore predictive and response biomarkers for treatment response based on the collected biomarkers

Eligibility

- Age 18 years or older
- Subjects must have one of several like histologically or cytologically confirmed malignant neoplasms (non-small cell lung cancer, small cell lung cancer, renal cell cancer, bladder cancer, Merkle cell cancer, MSI-H cancer (any histology), patients with melanoma and any other tumor type that has an approval for immune checkpoint inhibitor (ICI), who may or may not have progressed on previous anti-PD-1/PD-L1 mAb treatment +/- chemotherapy or anti-CTLA4 requiring further treatment.
- At least one lesion is amenable to radiation
- At least one additional non-contiguous lesion that has not been irradiated amenable to radiographic evaluation
- Have measurable disease based on immune-related response criteria (irRECIST).
- Tissue must be newly obtained as a core needle biopsy (not FNA) of the lesion being evaluated
- Adequate liver (AST, ALT, Alk Phos, and Tbili <2 fold upper limit) and kidney function (Cr < 2.5 limit of normal and Cr clearance >30)
- ECOG 0-2
- No history of clinically significant autoimmune disease, Crohn's disease, ulcerative colitis, or inflammatory disease.
- No prior diagnosis of hepatitis B or C
- No grade 4 toxicities during chemoradiation not resolved to grade ≤ 1 by the end of chemoradiation

Patient Enrollment

Patients Screened for the Study
N =29

Number of Subject Treated
N =19

Number of Patients Not Receiving Study Medication
Screen Failure – 9
Withdrew Consent - 1

Reason for Study Termination
Disease Progression - 8
Withdrew Consent - 4
Adverse Event - 1
Other complicating disease - 1
Death 2

Best Tumor Response per RECIST
N =14

PD (new brain lesion) -1
PD (insurance denial) - 1
PD (<1 cycle) - 1

Patients Ongoing as of OCT2023
N = 2

Best Target Lesion Response
N =11
(#15 – ICI-naïve)

Cancer types & Treatment Cycles

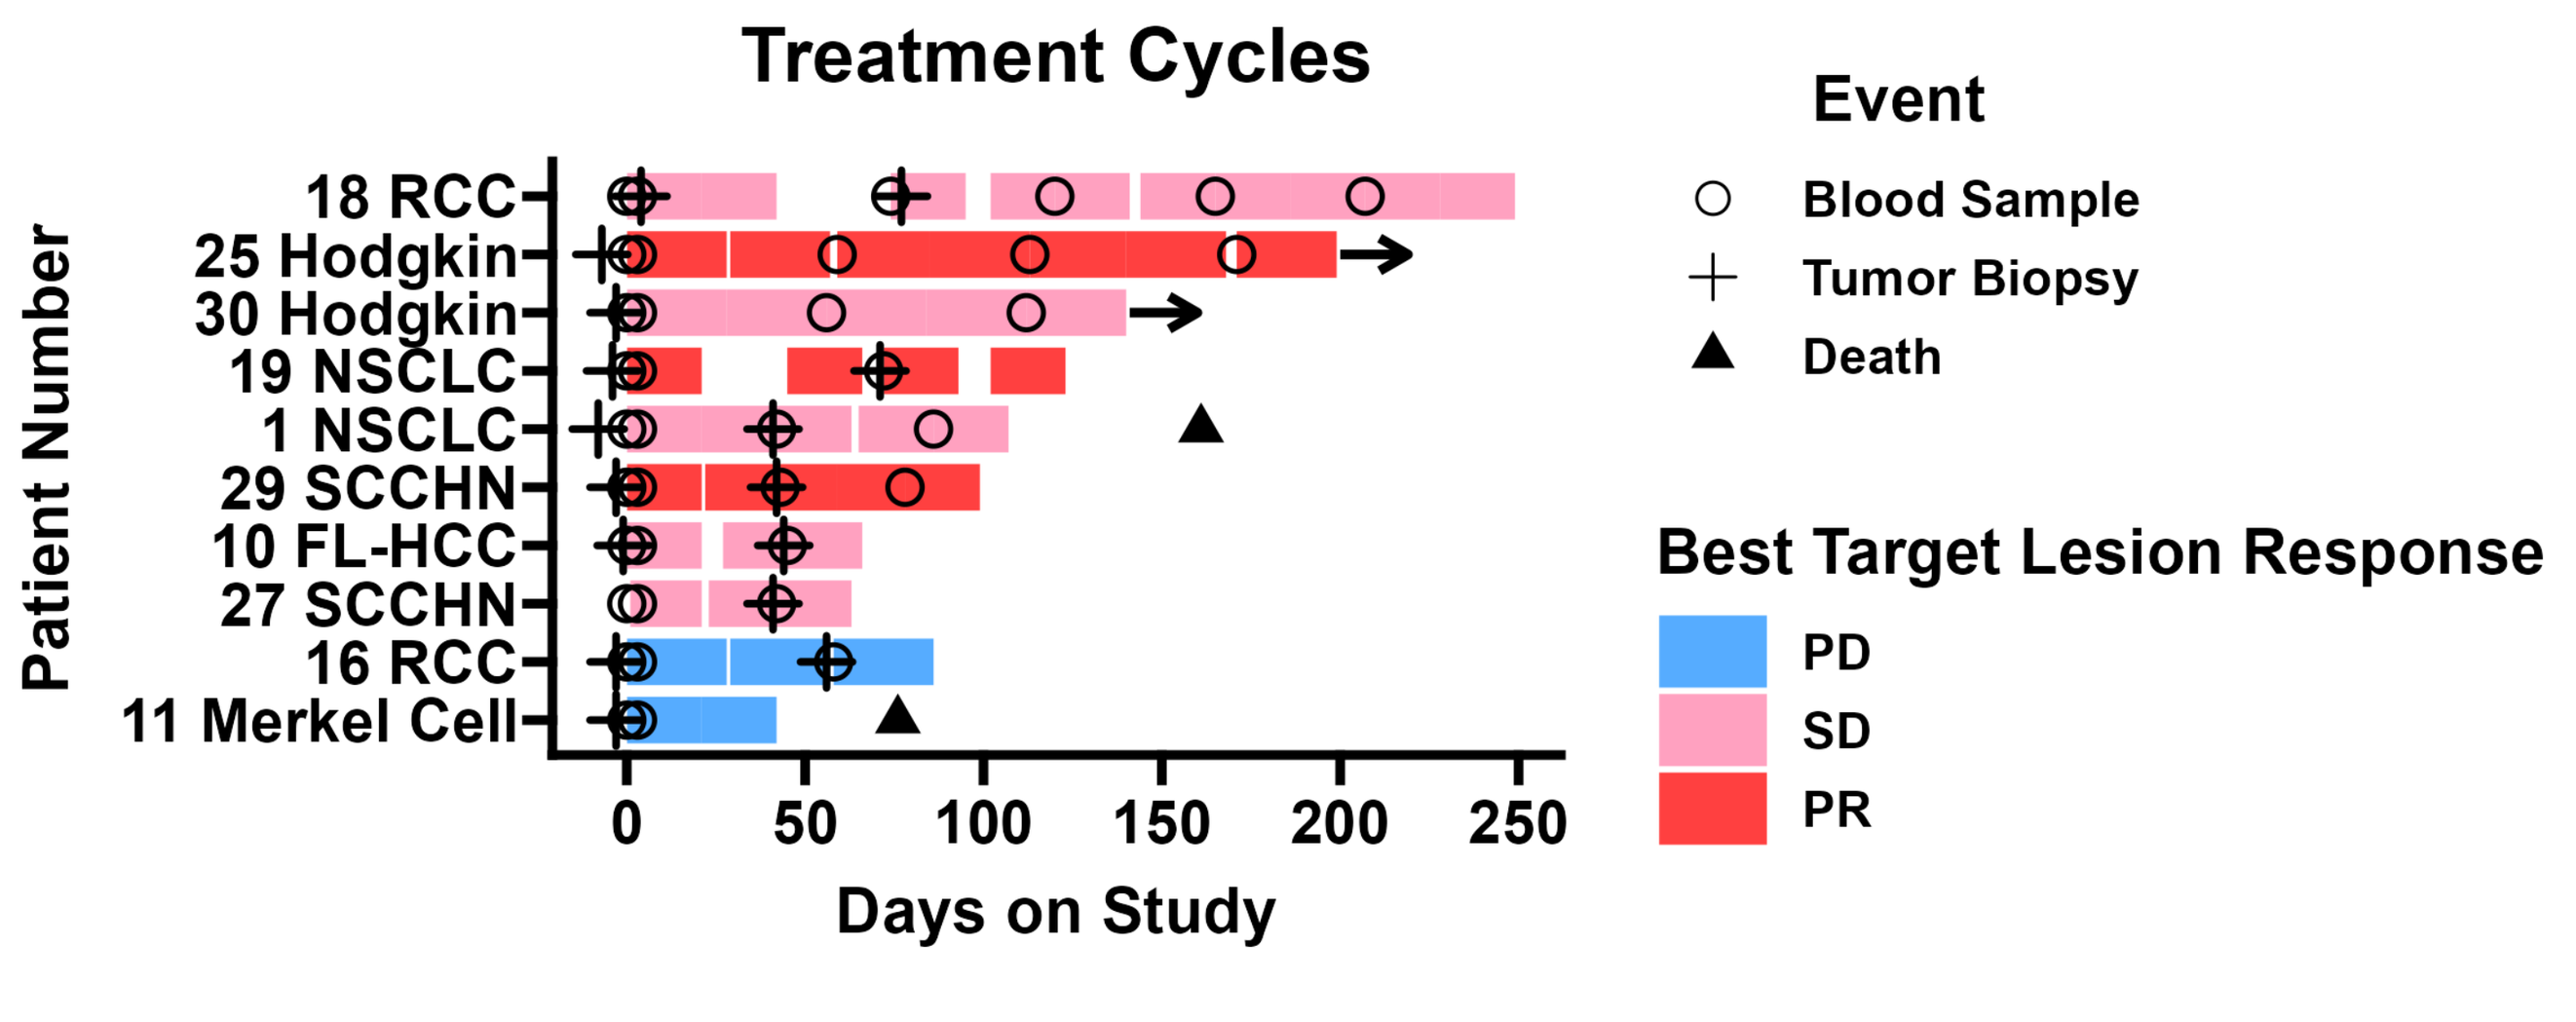
Treatment Cycles
Patient Number
18 RCC
25 Hodgkin
30 Hodgkin
19 NSCLC
1 NSCLC
29 SCCHN
10 FL-HCC
27 SCCHN
16 RCC
11 Merkel Cell
0 50 100 150 200 250
Days on Study
Event
Blood Sample
Tumor Biopsy
Death
Best Target Lesion Response
PD
SD
PR

Clinical Response to Triple Combination

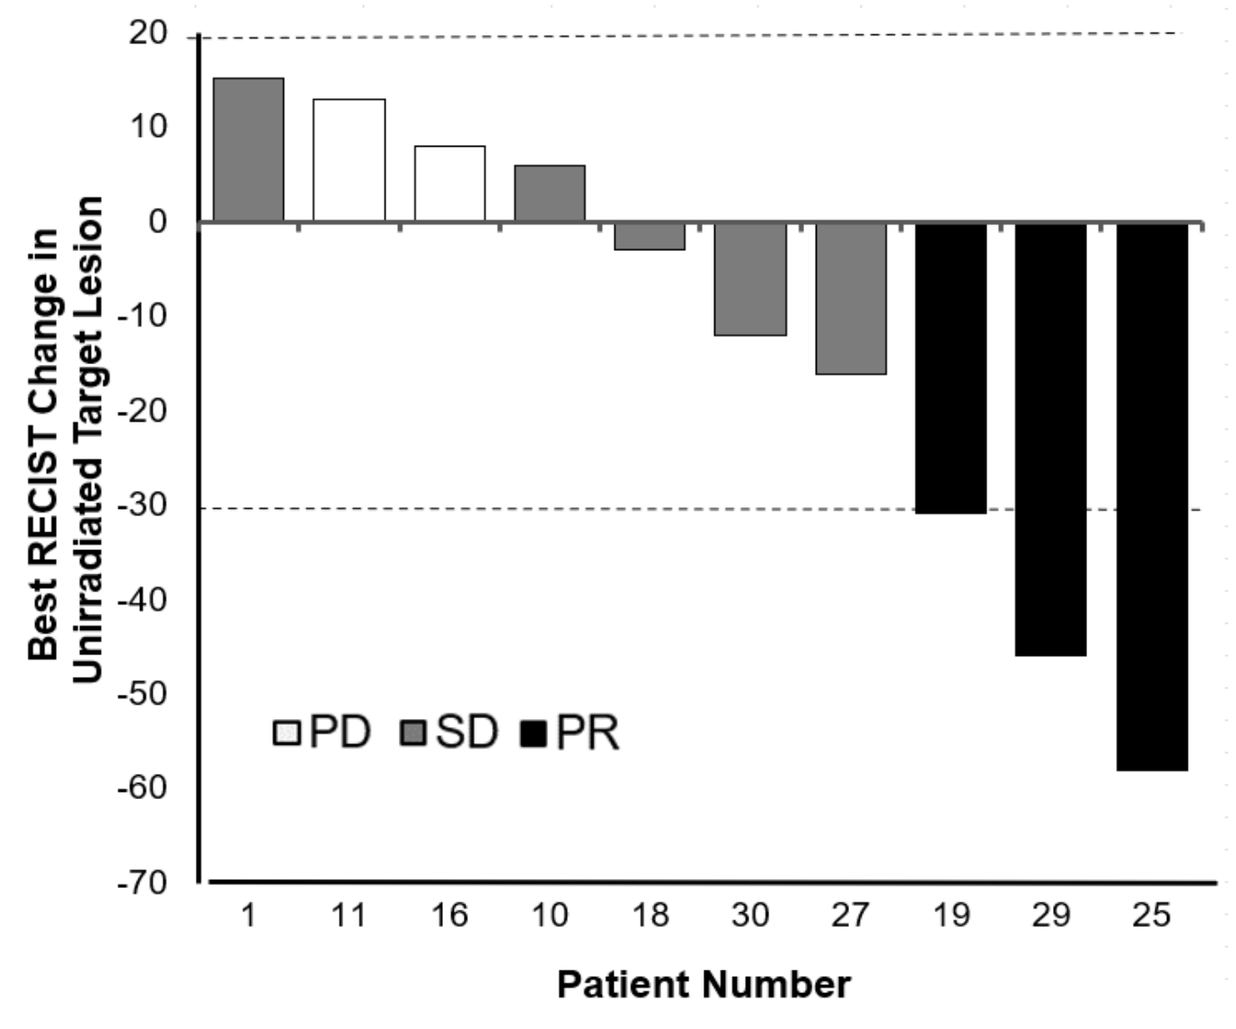
Best RECIST Change in Unirradiated Target Lesion
PD SD PR
1 11 16 10 18 30 27 19 29 25
Patient Number

Systemic abscopal response seen comparing baseline and C3D1 for one patient

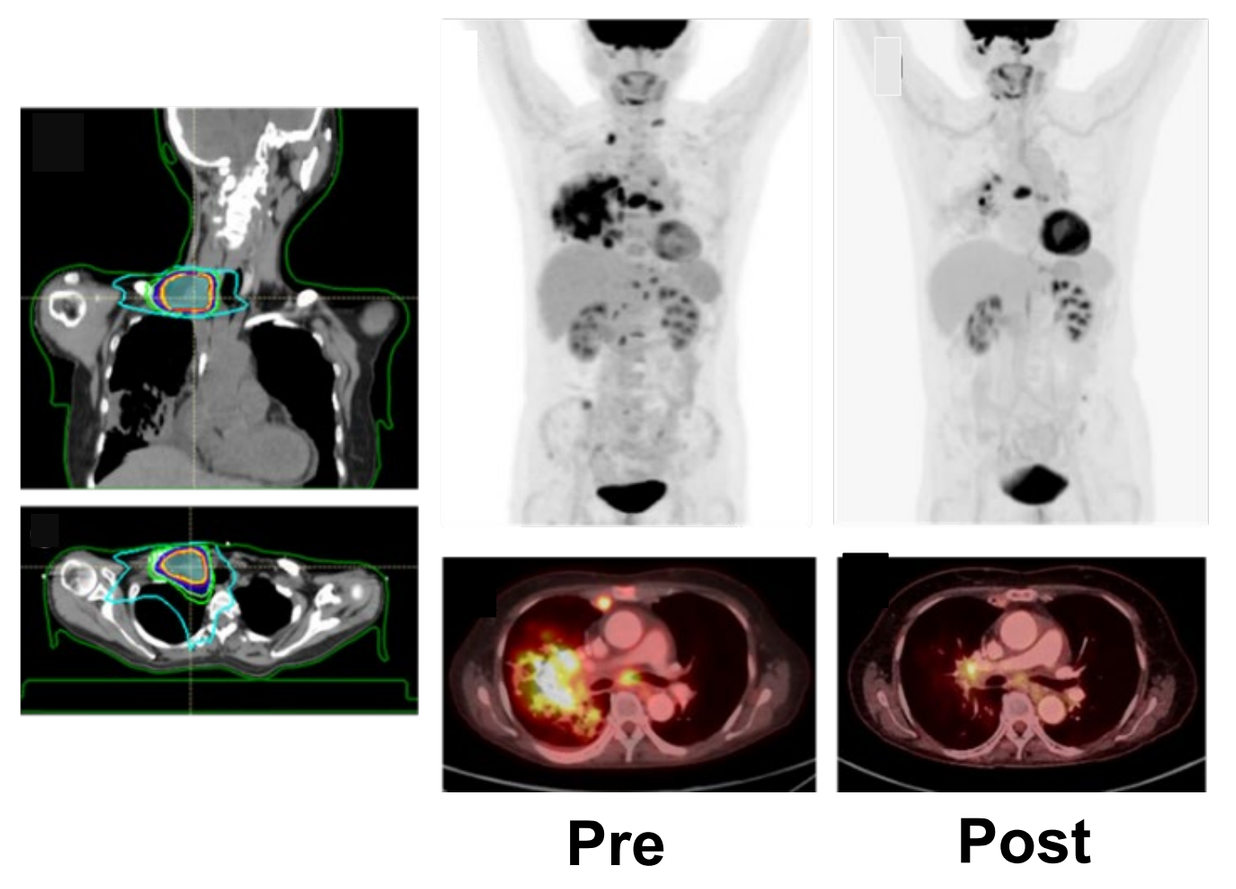
Pre
Post

Peripheral DC and Monocyte Phenotypic Changes

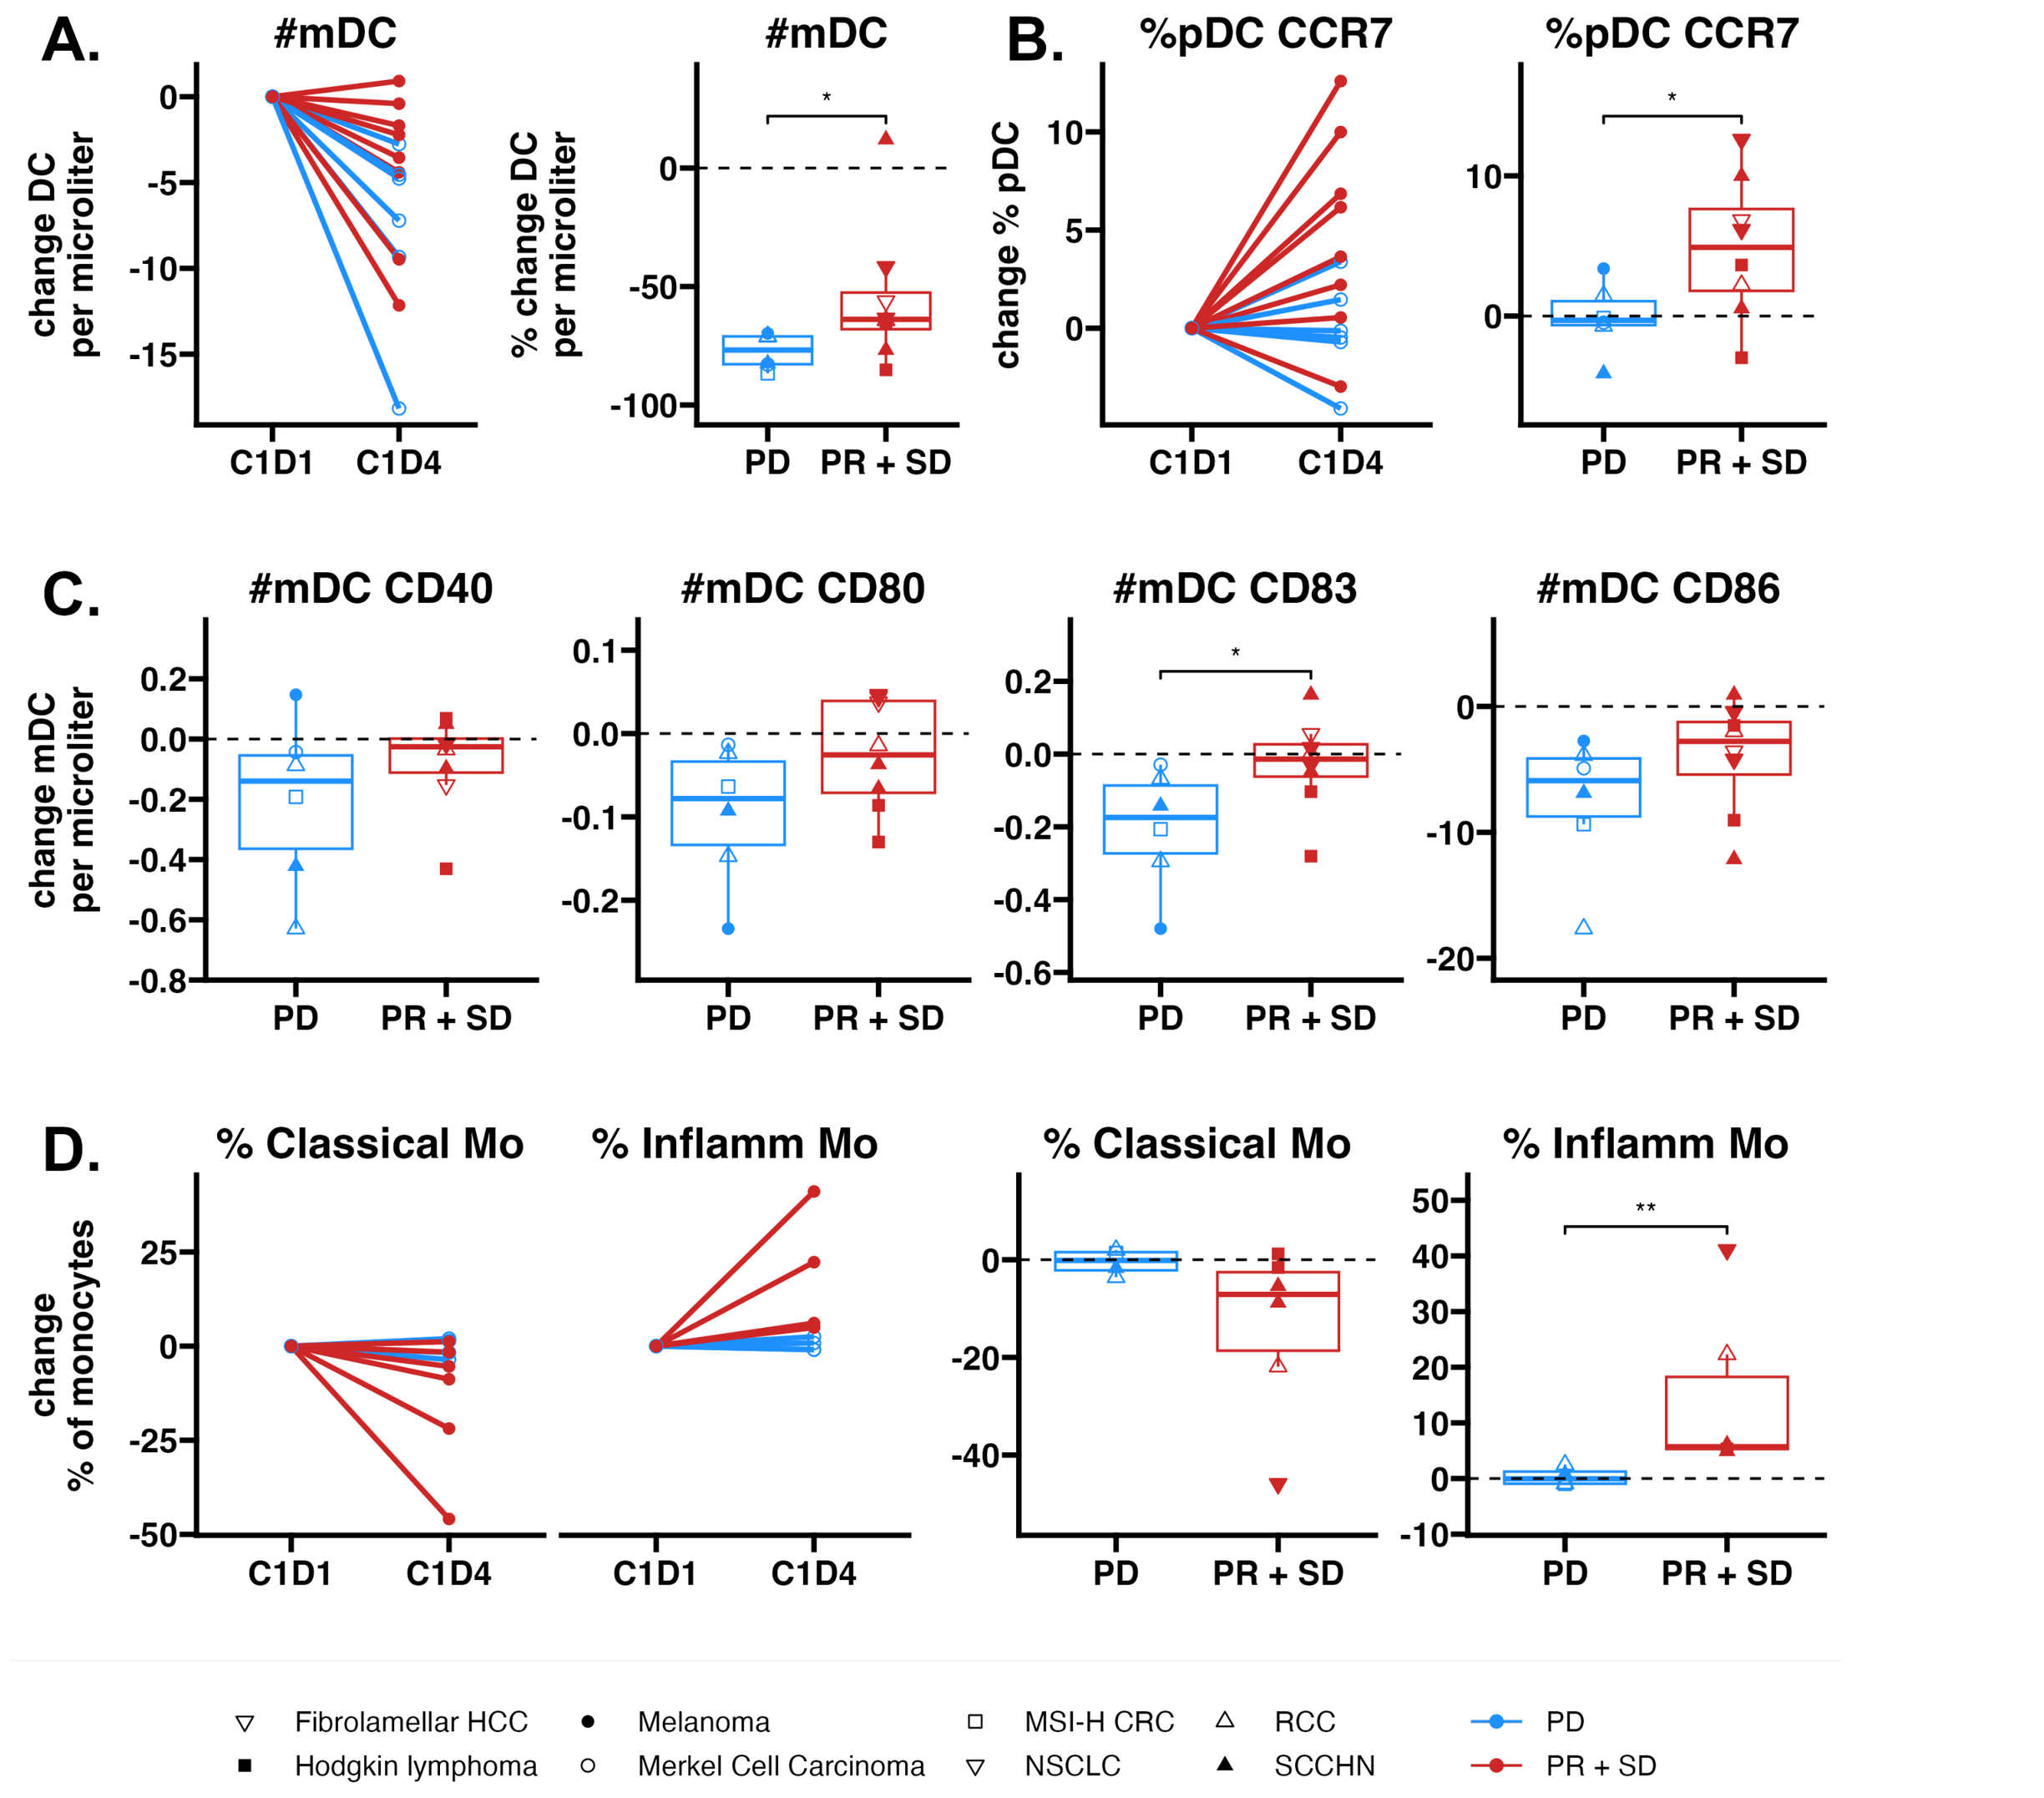
A. #mDC
#mDC
B. %pDC CCR7
%pDC CCR7
C. #mDC CD40
#mDC CD80
#mDC CD83
#mDC CD86
D. % Classical Mo
% Inflamm Mo
% Classical Mo
% Inflamm Mo
C1D1 C1D4
PD PR + SD
Fibrolamellar HCC
Hodgkin lymphoma
Melanoma
Merkel Cell Carcinoma
MSI-H CRC
NSCLC
RCC
SCCHN
PD
PR + SD

Combinatorial effect of plinabulin with radiation and immune checkpoint blockade on early immune activation and dendritic cell maturation in peripheral blood. Fresh whole blood samples were evaluated by flow cytometry at baseline and C1D4. **A**, Changes (left, all patient) and percent changes (right, PD vs. PR+SD) in #mDC per microliter. **B**, Changes in %pDC expressing CCR7. **C**, Changes in #mDC that express costimulatory molecules CD40, CD80, CD83 and CD86. **D**, Changes in %blood monocytes (CD14+HLA-DR+) shifting from classical (CD14+CD16-) to inflammatory (CD14dimCD16bright) phenotypes. Box plots showing different cancer types for the 14 evaluable patients. *P <0.05; **P <0.01 determined by Wilcoxon tests in two group comparisons.

Tumor scRNA Sequencing Analysis

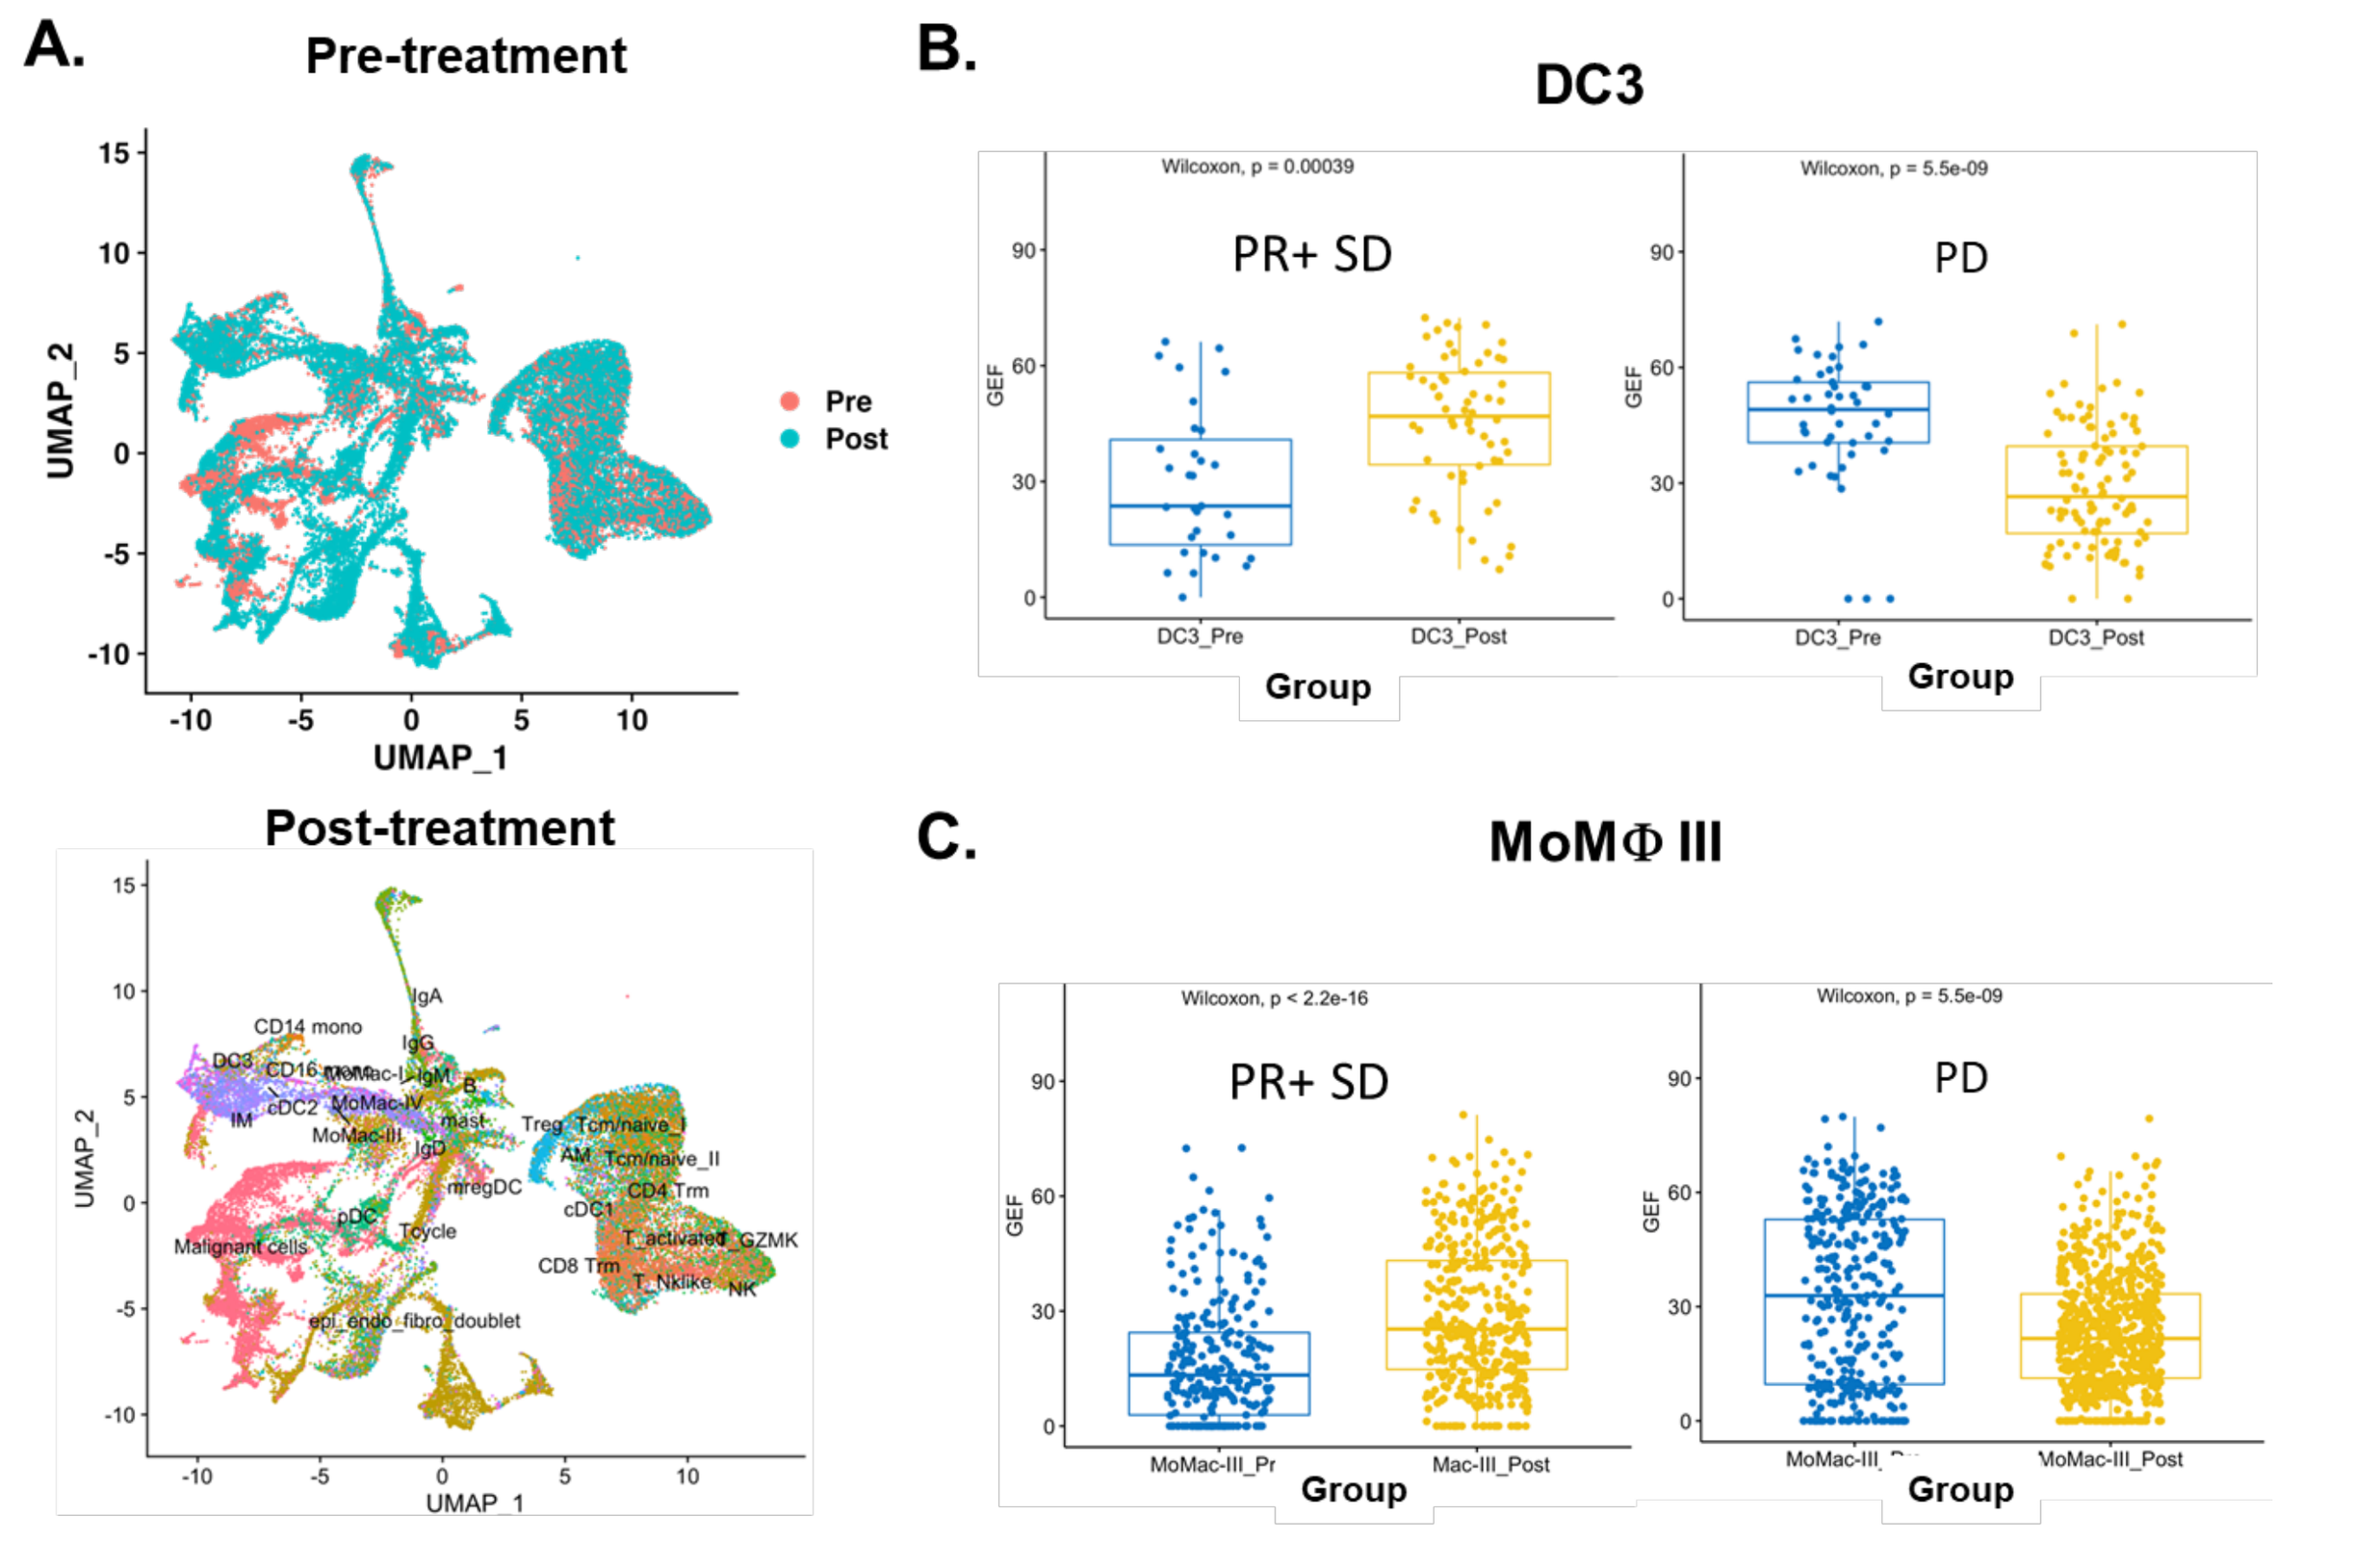
A. Pre-treatment
Post-treatment
UMAP_1
UMAP_2
Pre
Post
B. DC3
PR+ SD
PD
Group
C. MoMΦ III
PR+ SD
PD
Group

scRNAseq analysis of GEF-H1 immune score in matching pre- and post-treatment biopsies of unirradiated tumor lesions. A, UMAP plots of the scRNA-seq data from all patients grouped by pre and post treatment. **B**, Boxplots comparing GEF-H1 scores in DC3 (a CD1C+ subset of DC) of pre/post tumor samples from patients with (PR+SD, N=4) and without (PD, N=2) clinical benefits to the triple combination regimen. **C**, Boxplots comparing GEF-H1 scores in MoMϕ III (a subtype of monocyte-derived macrophages). P values shown are obtained using Wilcoxon tests in two group comparisons.

Conclusions

- Triple combination of plinabulin with irradiation and anti-PD-1 mAb enhances DC activation, T-cell proliferation, and abscopal effect in preclinical models.
- The combination is safe and yields good overall response (3 PR; 5 SD; 3 PD, among 10 ICI-refractory and 1 ICI-naïve patients) with 2 Hodgkin lymphoma patients demonstrating enduring responses.
- In patients progressing on ICI, the addition of plinabulin after RT initiation (3-6 hours apart) induces an early systemic immune response detectable in subsets of DC and monocytes in the peripheral blood 3 days later.
- scRNAseq analysis of tumor biopsies of patients with clinical benefit (PR+SD), indicates an increased GEF-H1-dependent immune signature score in subsets of tumor-derived DCs and monocyte-derived macrophages. The opposite was seen in patients with PD.
- Additional biomarker analyses are underway.

References

1. La Sala, G., et al. Thermodynamics, and Kinetics of Plinabulin Binding to Two Tubulin Isotypes. *Chem* **5**, 1-18. 2019
2. Chen, Y., et al. SBRT combined with PD-1/PD-L1 inhibitors in NSCLC treatment: a focus on the mechanisms, advances, and future challenges. *J Hematol Oncol* **13**, 105, 2020.
3. Cimino, P.J., et al. Plinabulin, an inhibitor of tubulin polymerization, targets KRAS signaling through disruption of endosomal recycling. *Biomed Rep* **10**, 218-224, 2019
4. Del Prete, A., et al. Dendritic cell subsets in cancer immunity and tumor antigen sensing. *Cell Mol Immunol* **20**, 432-447, 2023.
5. Kashyap, A.S., et al. GEF-H1 Signaling upon Microtubule Destabilization Is Required for Dendritic Cell Activation and Specific Anti-tumor Responses. *Cell Rep* **28**, 3367-3380 e3368, 2019
6. Leader, A.M., et al. Single-cell analysis of human non-small cell lung cancer lesions refines tumor classification and patient stratification. *Cancer Cell* **39**, 1594-1609 e1512, 2021
7. Natoli, M., et al. Plinabulin, a Distinct Microtubule-Targeting Chemotherapy, Promotes M1-Like Macrophage Polarization and Anti-tumor Immunity. *Front Oncol* **11**, 644608, 2021