Guanine nucleotide exchange factor-H1 signaling is critical for induction of anti-tumor immunity induced by microtubule-depolymerizing drugs

Kashyap AS[1,2], Zhao Y[2], Martin K[1], Omatsu T[2], Lloyd KG[3], Huang L[3], Reinecker HC[2], Zippelius A[1]

[1]Dept. of Biomedicine, University Hospital Basel, Switzerland; [2]Gastrointestinal Unit, Massachusetts General Hospital, USA; [3]BeyondSpring Pharmaceuticals, New York, USA

Introduction

❑ The immuno-suppressive tumor micro-environment poses a major hurdle for anti-tumor immunity and for the targeting of immune checkpoint inhibitory receptors such as CTLA-4 and PD1.

❑ We have shown that contrary to microtubule stabilizing agents (MSA) tubulin de-polymerizing agents (MDA) such as plinabulin (currently in Phase 3 in NSCLC), auristatins and maytansinoids reprogram the tumor immune environment by activating DCs and priming of anti-tumour T cells[1,2].

❑ MDAs were shown in pre-clinical models to boost the activity of anti-PD1 and anti-CTLA4[3].

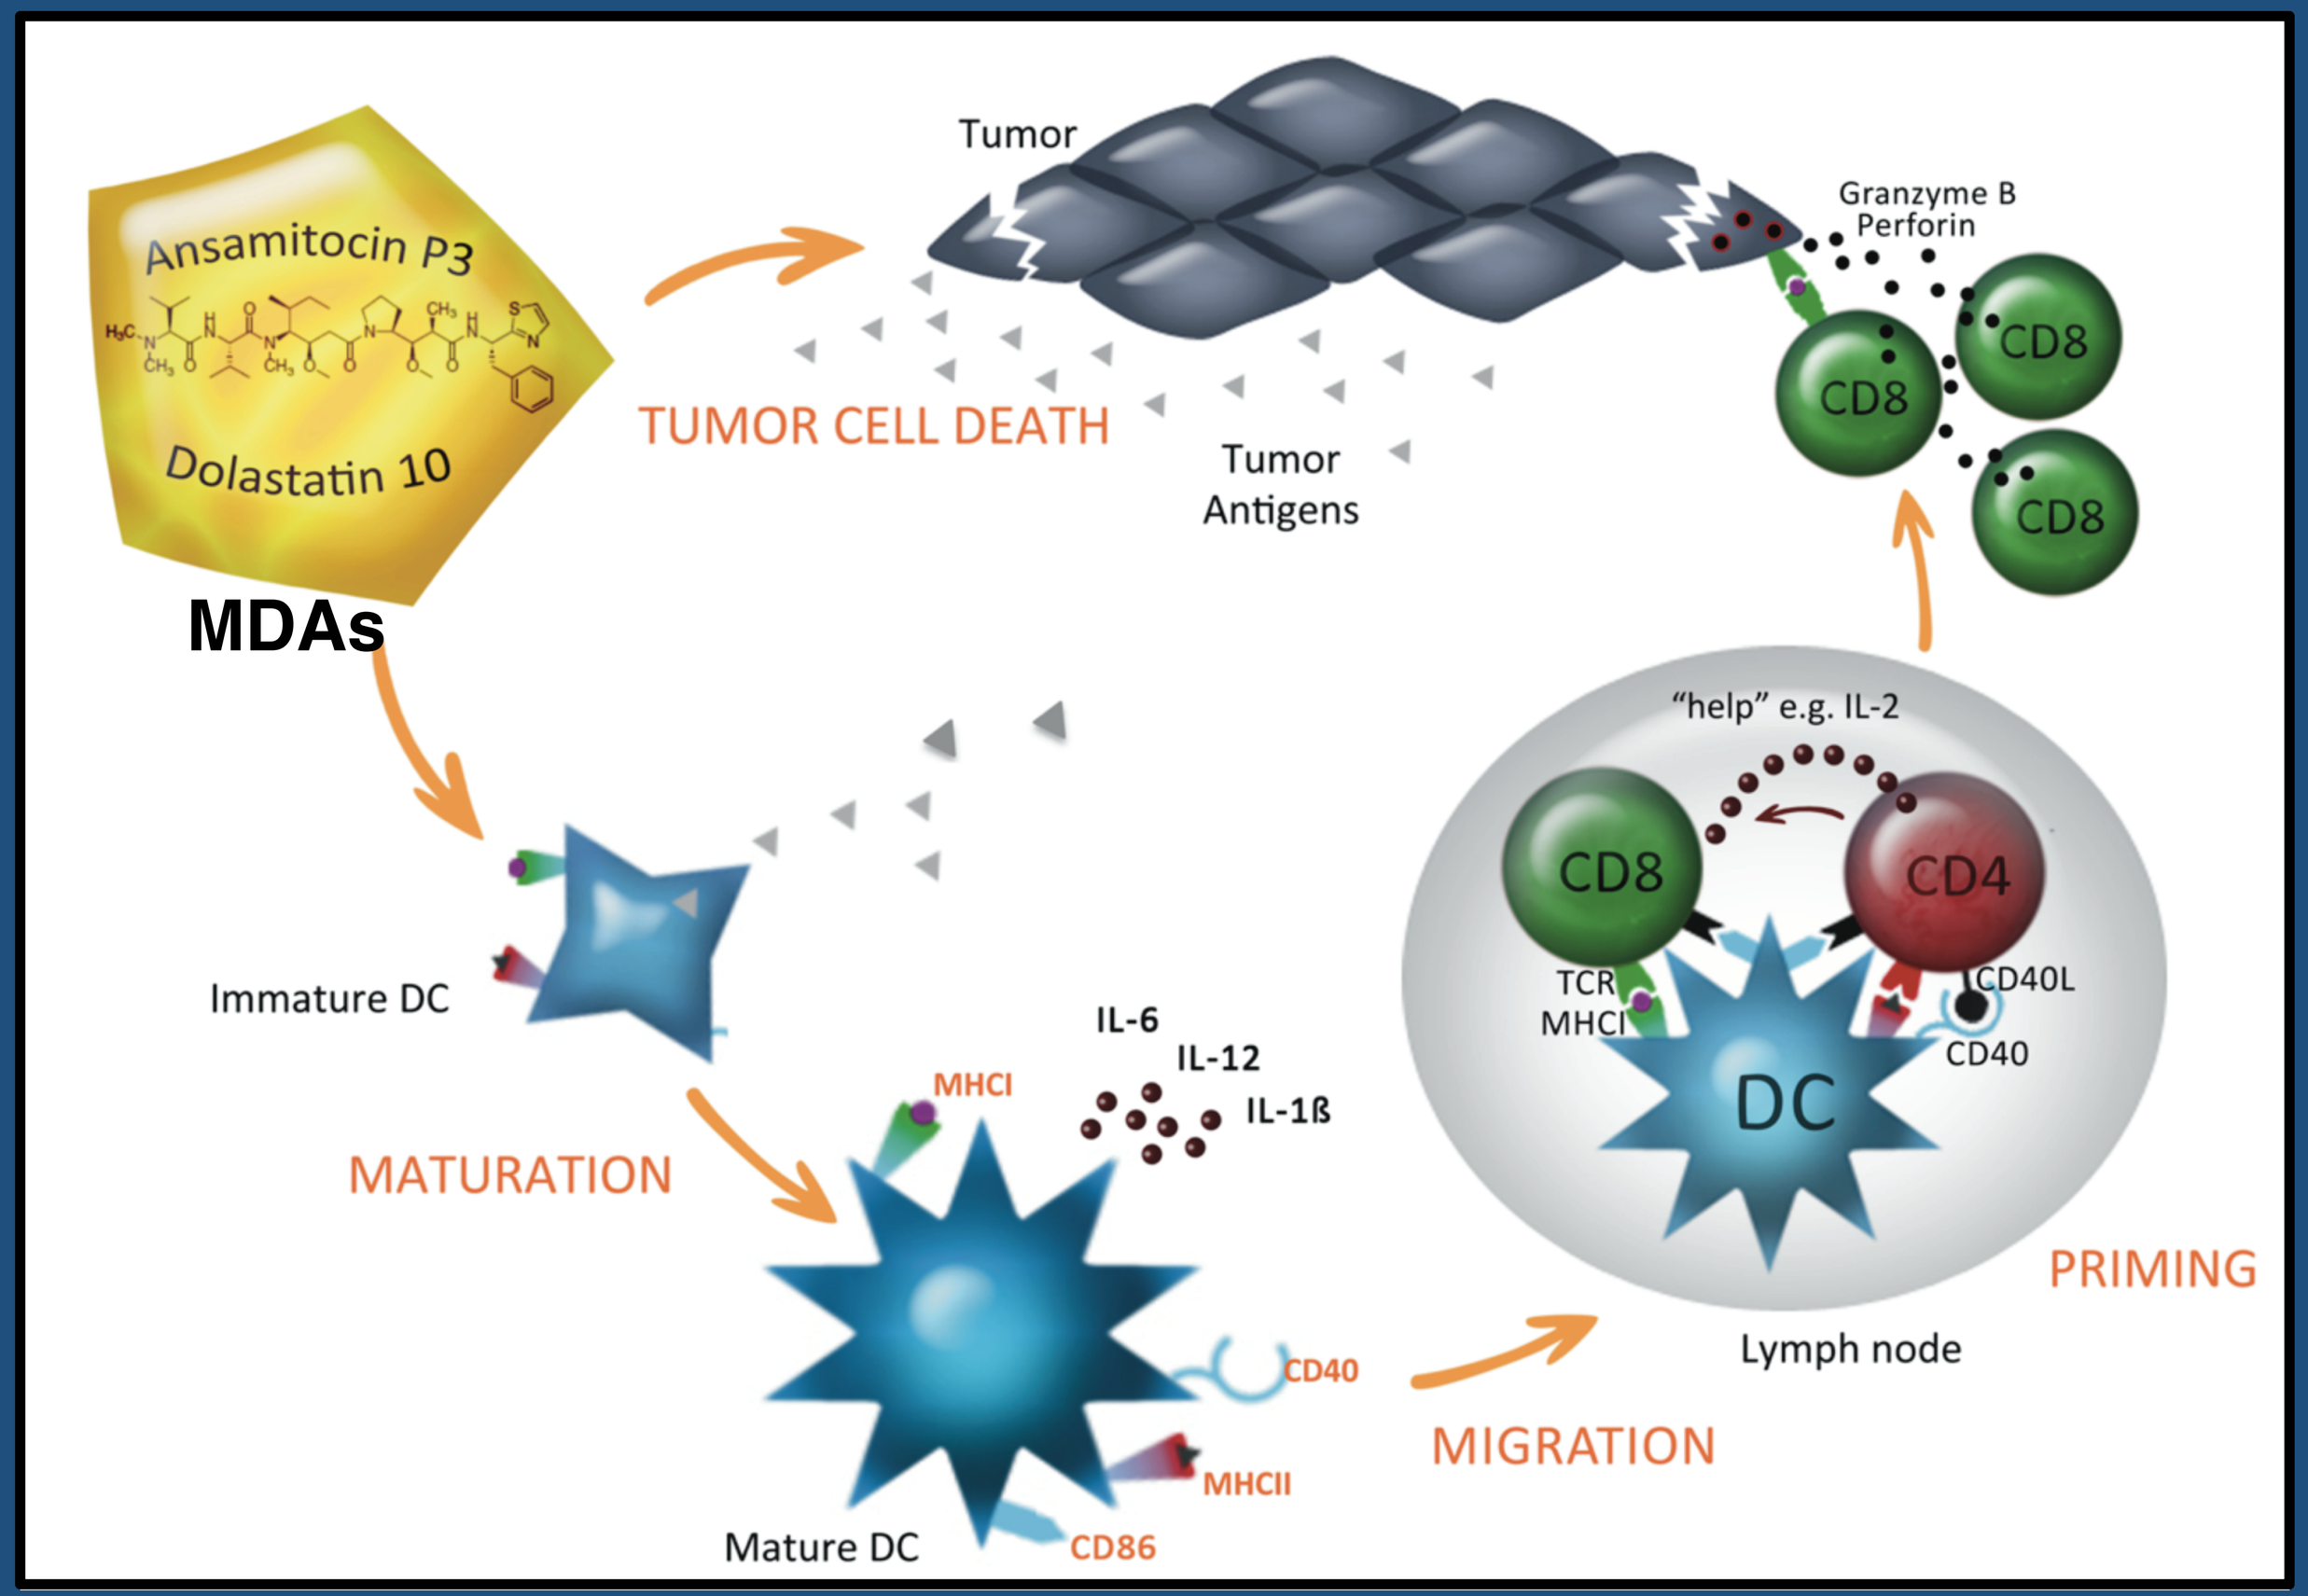
Ansamitocin P3
Dolastatin 10
MDAs
Tumor
TUMOR CELL DEATH
Tumor Antigens
Granzyme B Perforin
CD8
"help" e.g. IL-2
CD4
TCR
MHCI
CD40L
CD40
DC
Immature DC
MATURATION
IL-6
IL-12
IL-1ß
MHCI
MHCII
CD86
Mature DC
MIGRATION
Lymph node
PRIMING

Fig 1: Model of MDA-induced anti-tumor immunity

❑ The mechanism behind MDA-induced dendritic cell (DC) activation remains unaddressed and is the focus of this study.

Hypothesis

DC activation induced by MDAs is independent of pattern-recognition receptors (PRR), but is rather dependent on the direct disruption of microtubules and the subsequent activation of downstream signaling cascades.

References

1. Mueller P, et al. 2014. *Cancer Immunol Res.*
2. Martin K, et al. 2014. *Cancer Immunol Immunother.*
3. Mueler P, et al. 2015. *Sci Trans Med.*

Results

MDAs and not MSAs stimulate DC activation: this is independent of microtubule binding site

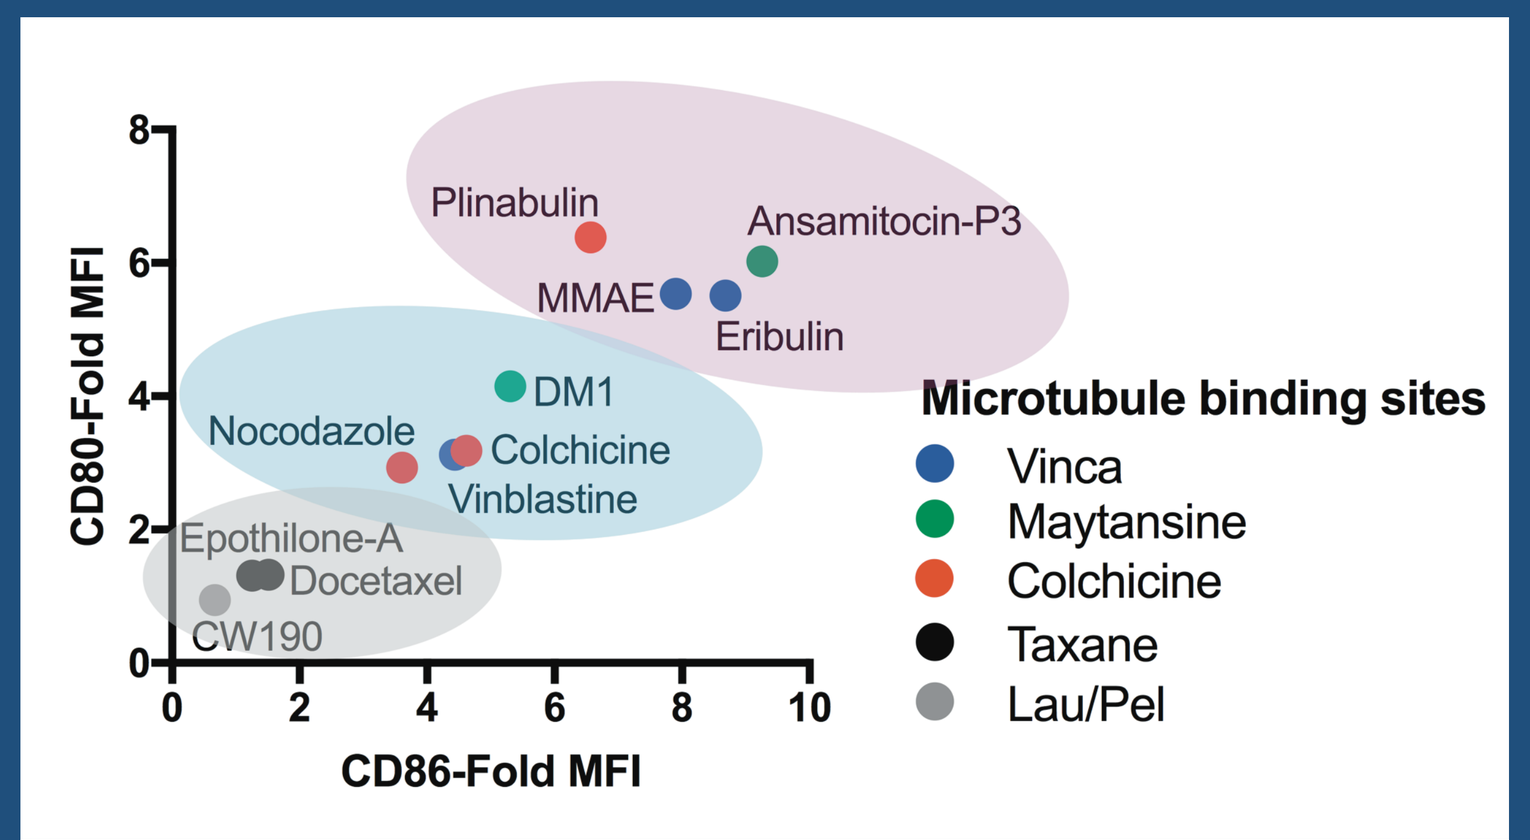
Plinabulin
Ansamitocin-P3
MMAE
Eribulin
DM1
Nocodazole
Colchicine
Vinblastine
Epothilone-A
Docetaxel
CW190
Microtubule binding sites
Vinca
Maytansine
Colchicine
Taxane
Lau/Pel
CD80-Fold MFI
CD86-Fold MFI

Fig 2: SP37A3 DC cells were treated with 1 µM of different MDAs and MSAs targeting distinct microtubule binding sites. CD86 and CD80 expression was assessed by FACS.

MDA-induced DC activation is independent of PRRs

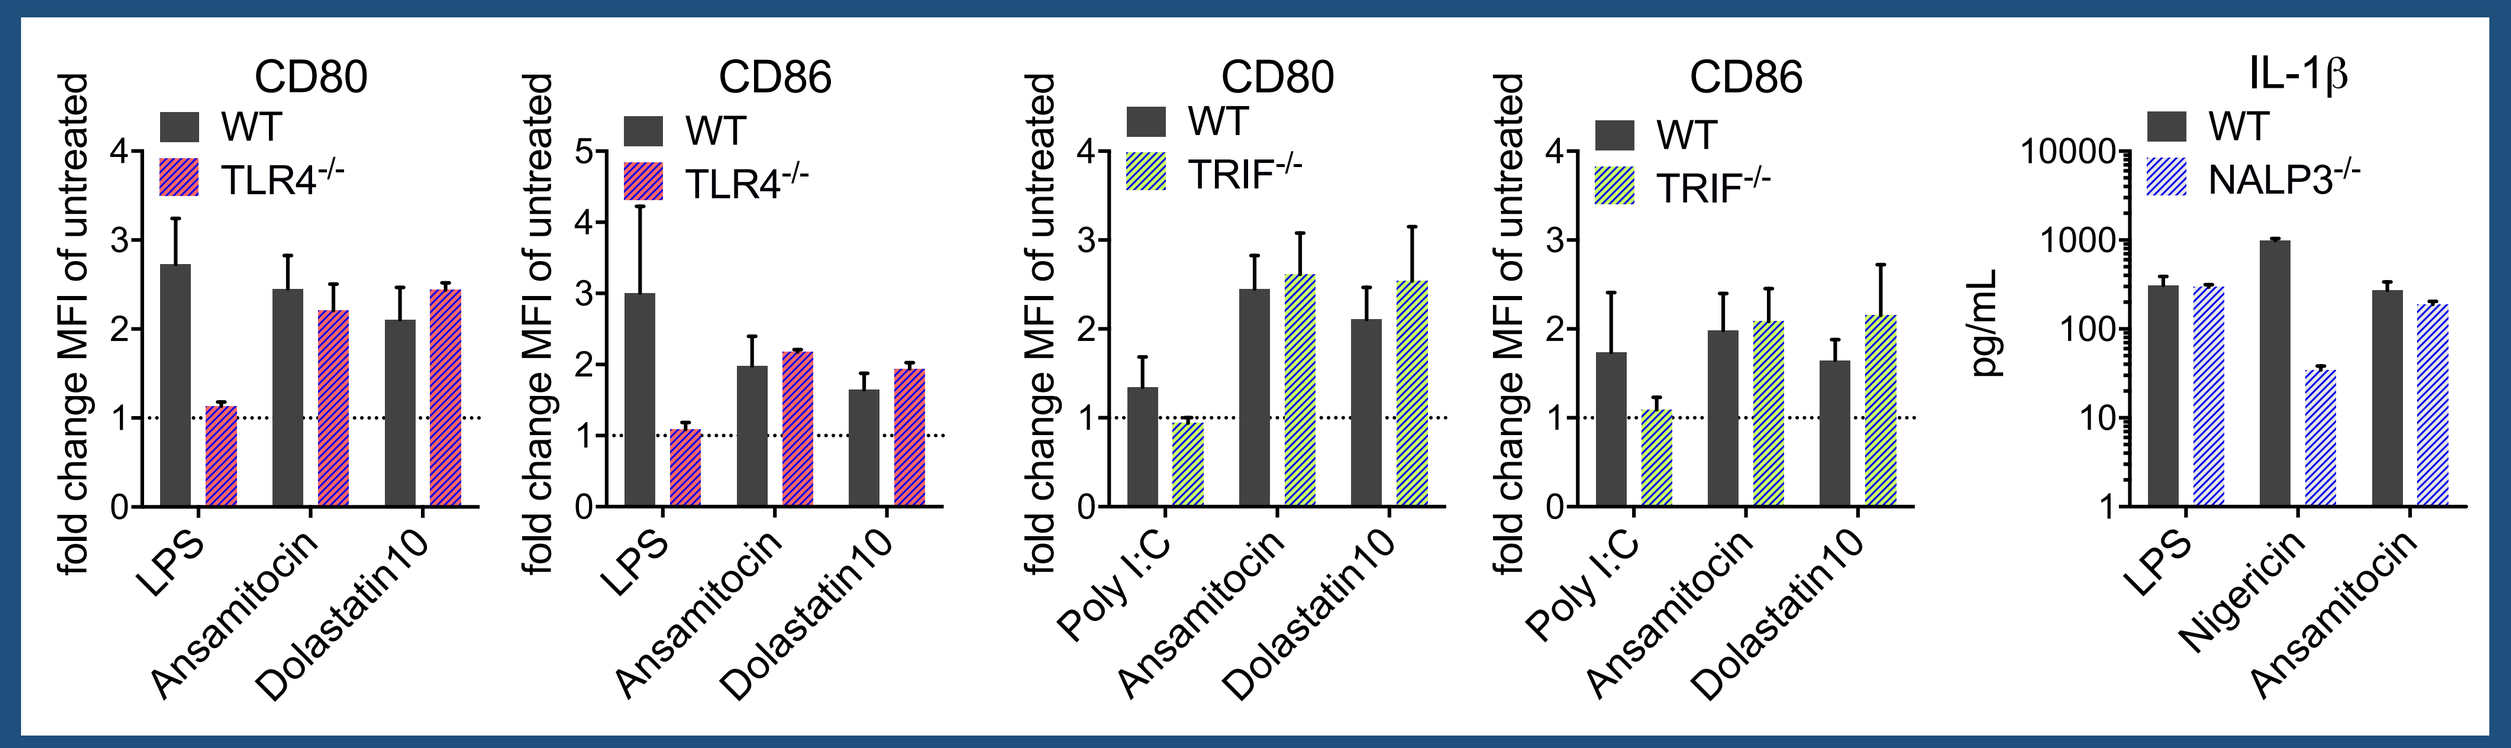
CD80
CD86
WT
TLR4-/-
TRIF-/-
IL-1β
NALP3-/-
fold change MFI of untreated
pg/mL
LPS
Poly I:C
Ansamitocin
Dolastatin10
Nigericin

Fig 3: BMDCs from WT/KO mice were treated with MDAs (100 nM) or controls for 24h. CD80 and CD86 expression was assessed by FACS and IL1β measured by ELISA. MDA-induced DC activation remained unaffected in the KO mice

Microtubule associated Guanine nucleotide exchange factor (GEFH1) is released from microtubules upon treatment with Ansamitocin-P3

A

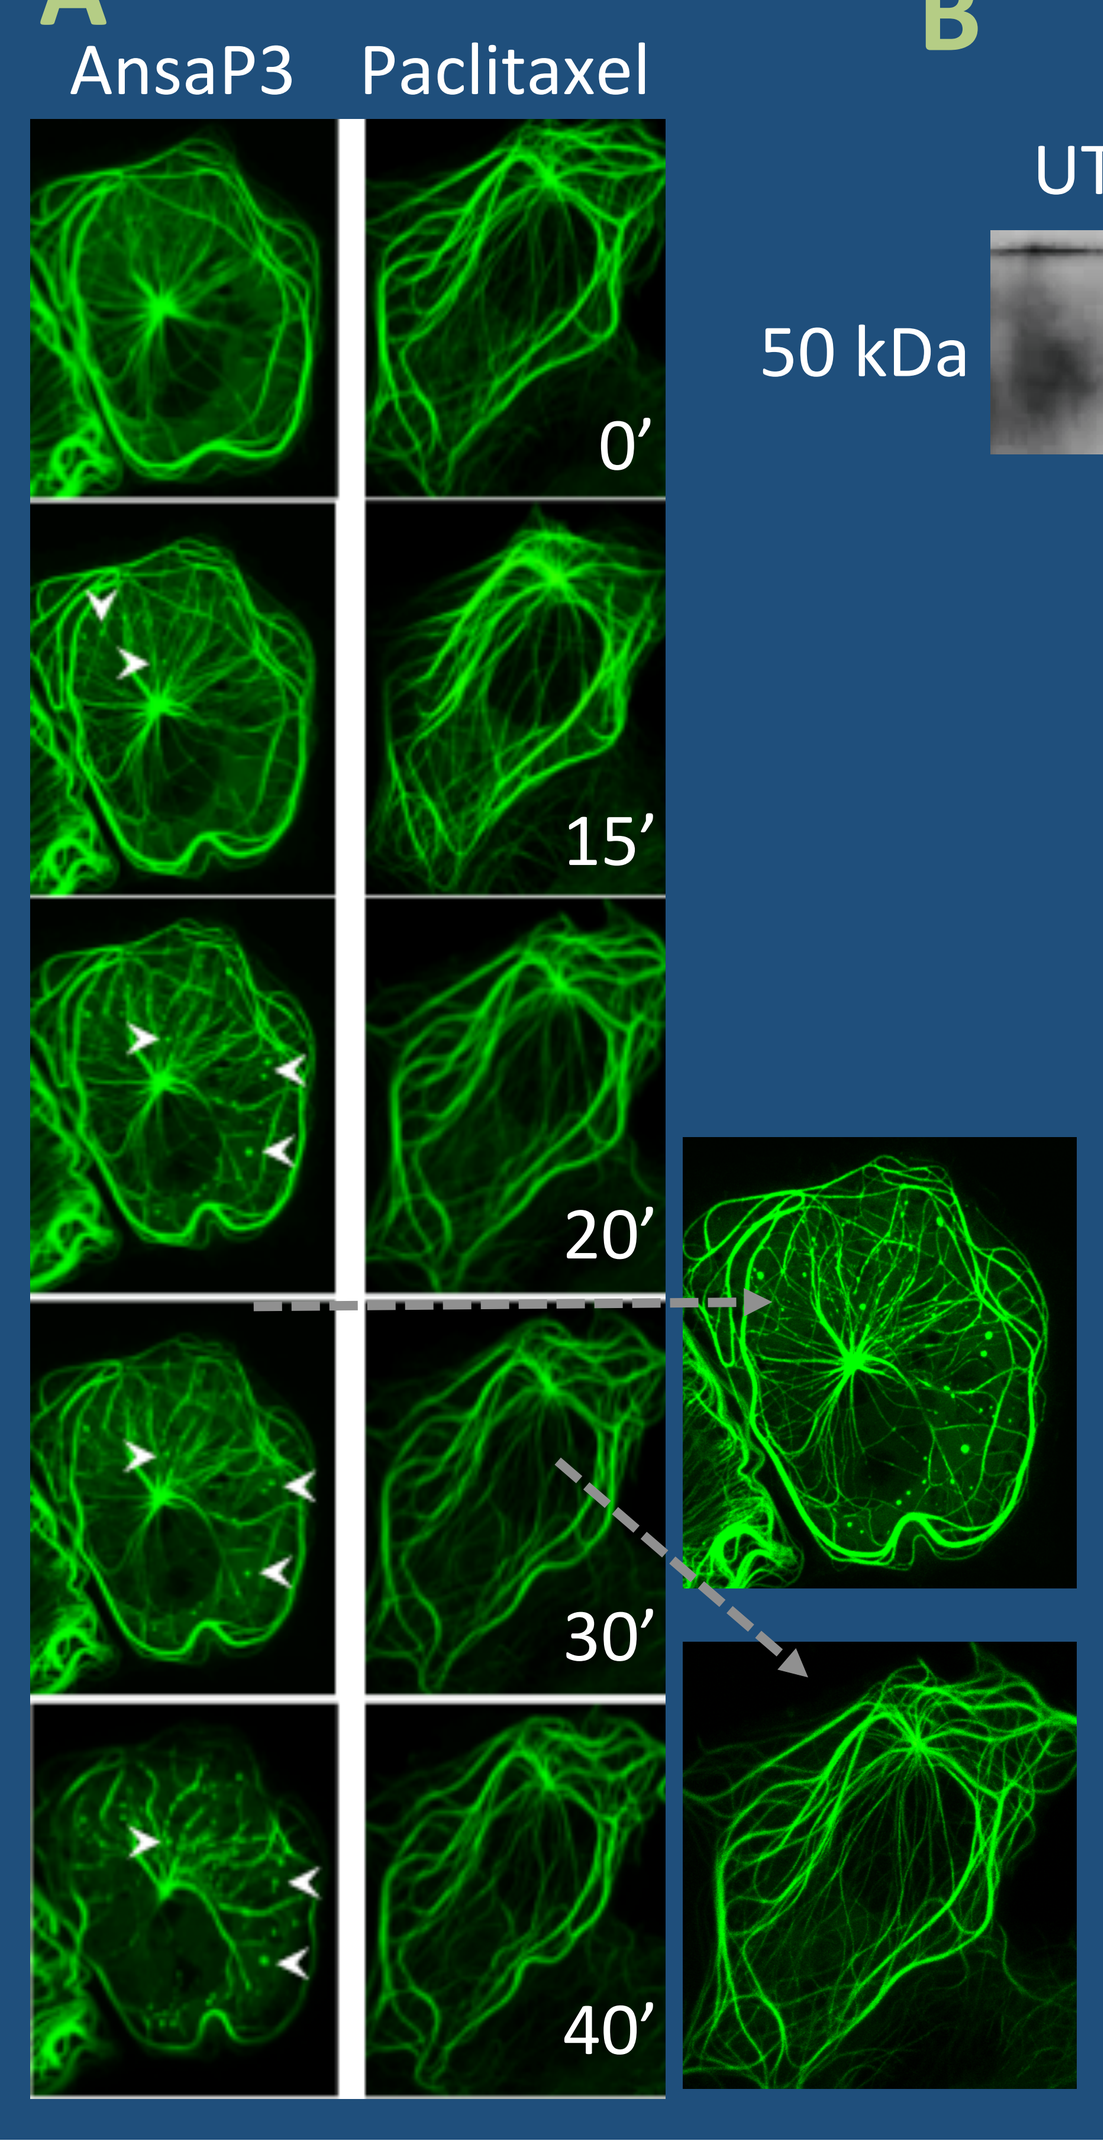
AnsaP3
Paclitaxel
0'
15'
20'
30'
40'

B

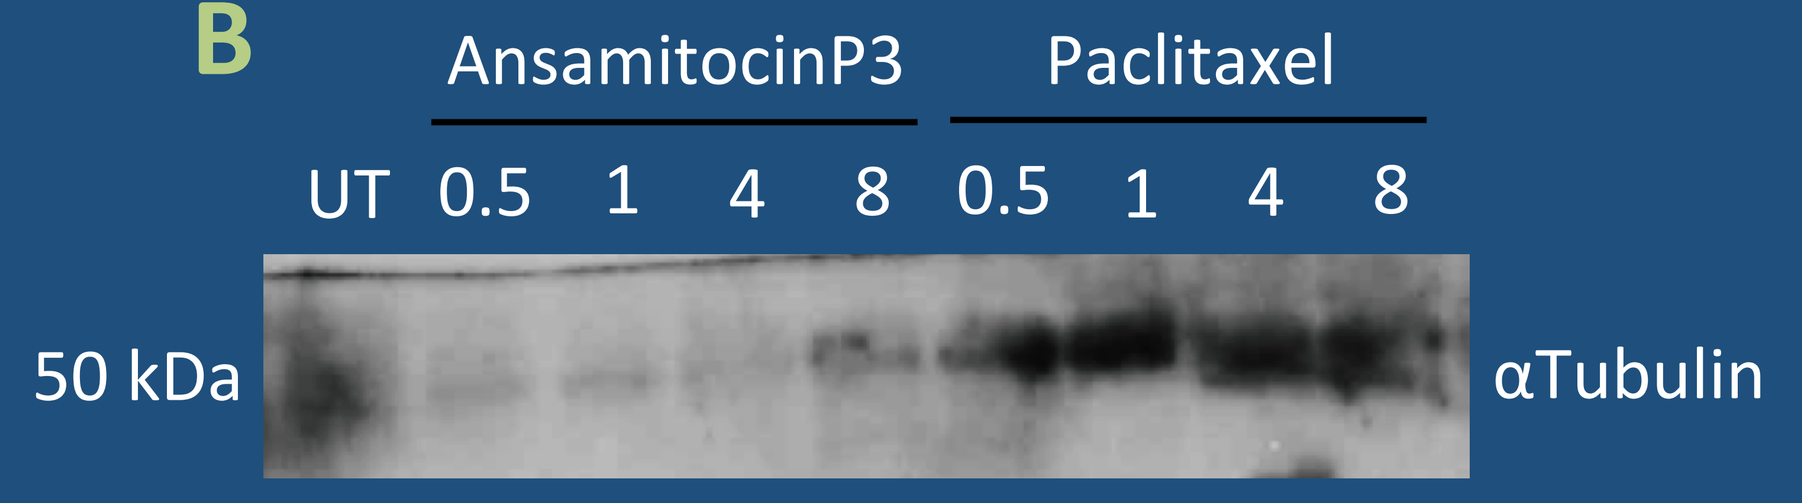
AnsamitocinP3
Paclitaxel
UT 0.5 1 4 8 0.5 1 4 8
50 kDa
αTubulin

Fig 4: A) Within 15 minutes changes in GEFH1 localisation was observed with AnsaP3-treated Cos7 fibroblasts expressing GEFH1-GFP. No such changes were observed with Paclitaxel. **B)** Immuno-precipitated (IP) GEFH1 samples from WT BMDCs were probed for αTubulin. As opposed to Paclitaxel treatment αTubulin was undetected in GEFH1 IP samples after AnsaP3 treatment.

MDAs (and not MSAs) trigger GEFH1 activation

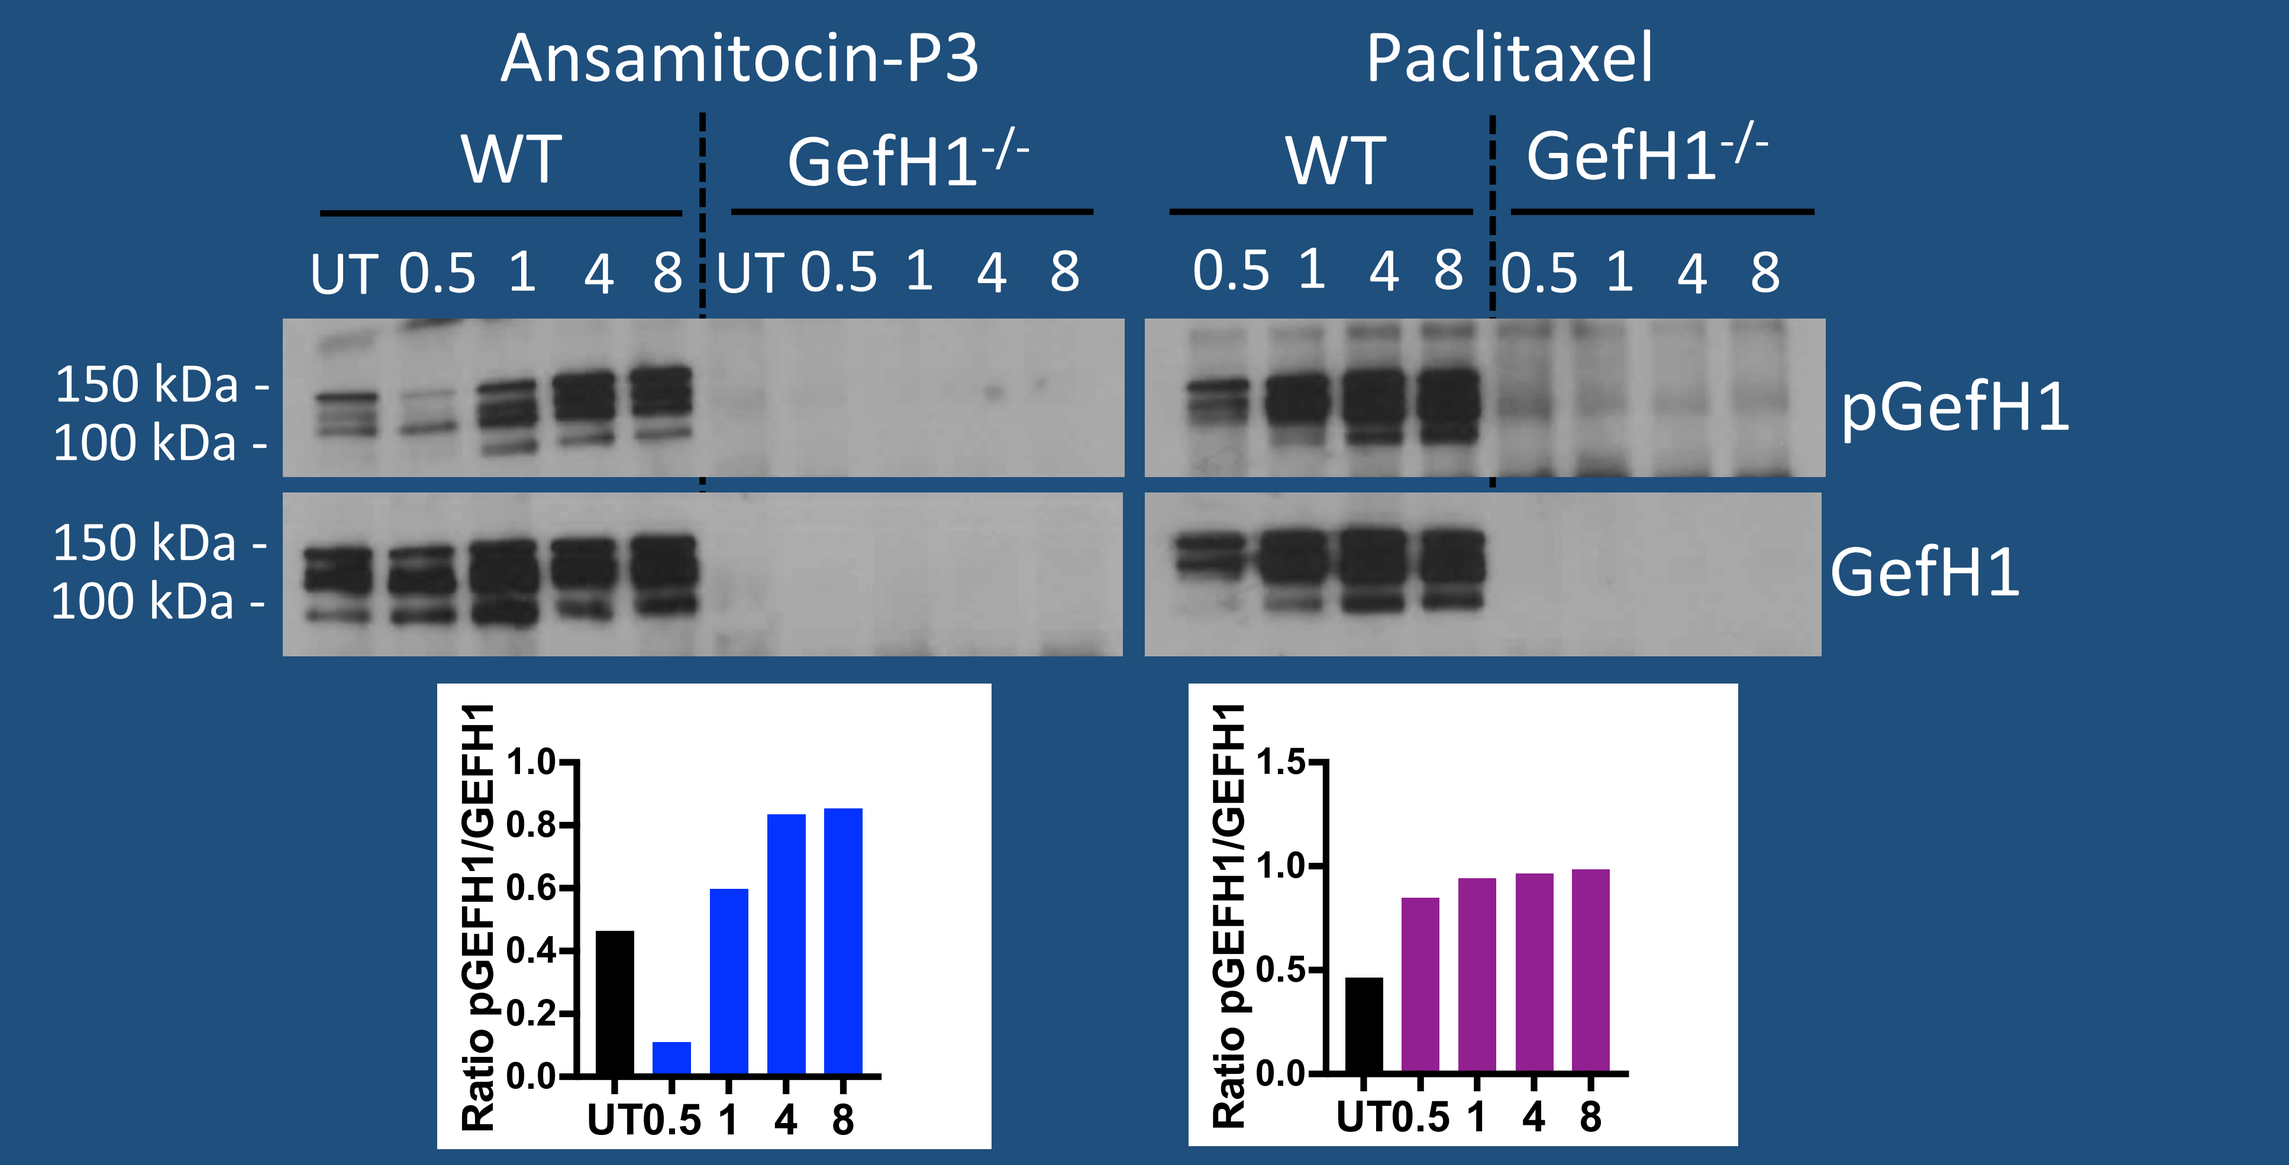
Ansamitocin-P3
Paclitaxel
WT
GefH1-/-
UT 0.5 1 4 8
0.5 1 4 8
150 kDa -
100 kDa -
pGefH1
GefH1
Ratio pGEFH1/GEFH1
UT0.5 1 4 8

Fig 5: GEFH1 was immunoprecipitated from WT and GEFH1-/- BMDCs treated with Ansamitocin-P3 or Paclitaxel (100 nM) for indicated time-points. No GEFH1 was detected in GEFH1-/- BMDCs. Active GEFH1 (de-phosphorylated) was detected with Ansamitocin-P3 at 30 mins (blots and densitometry graphs). GEFH1 remained inactive (phosphorylated) with Paclitaxel.

GEFH1 release and activation triggers conversion RhoA-GDP to RhoA-GTP

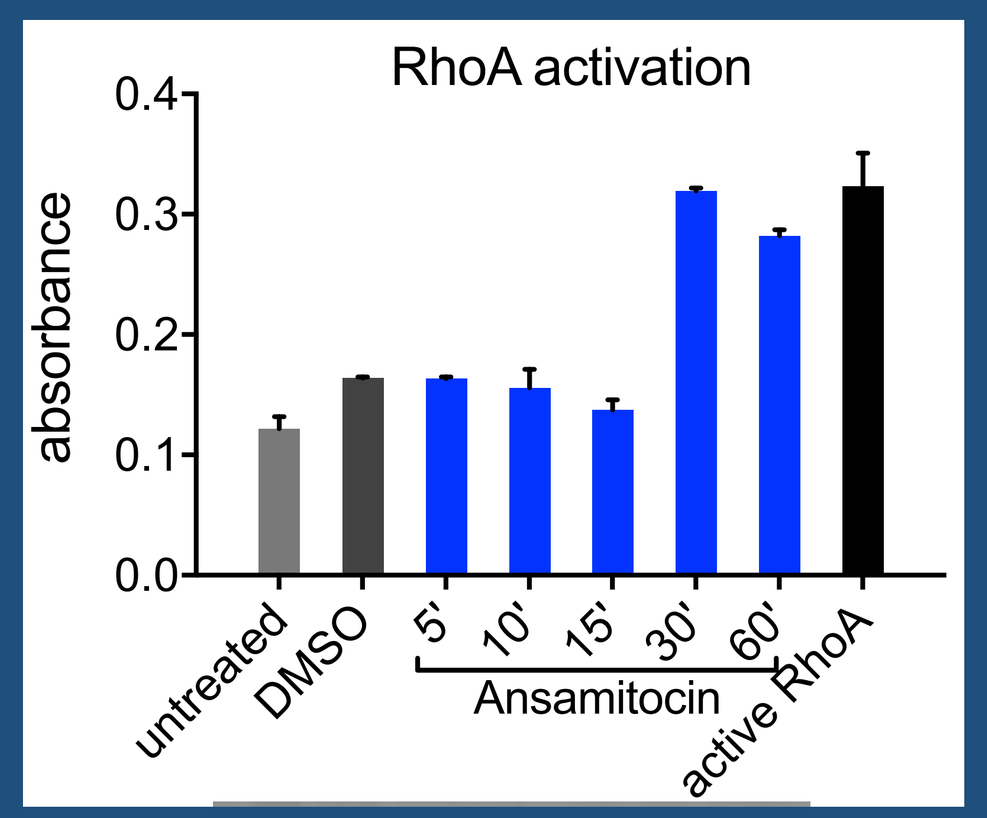
RhoA activation
absorbance
untreated
DMSO
5' 10' 15' 30' 60'
Ansamitocin
active RhoA

Fig 6: Total protein from SP37A3 DCs treated with Ansamitocin-P3 was added to G-LISA plates and Rho-GTP was measured. Rho-GTP was observed within 30 mins after treatment.

MDAs recruit the MKK4/JNK/cJun signaling axis to induce DC activation in a GEFH1-dependent manner

A

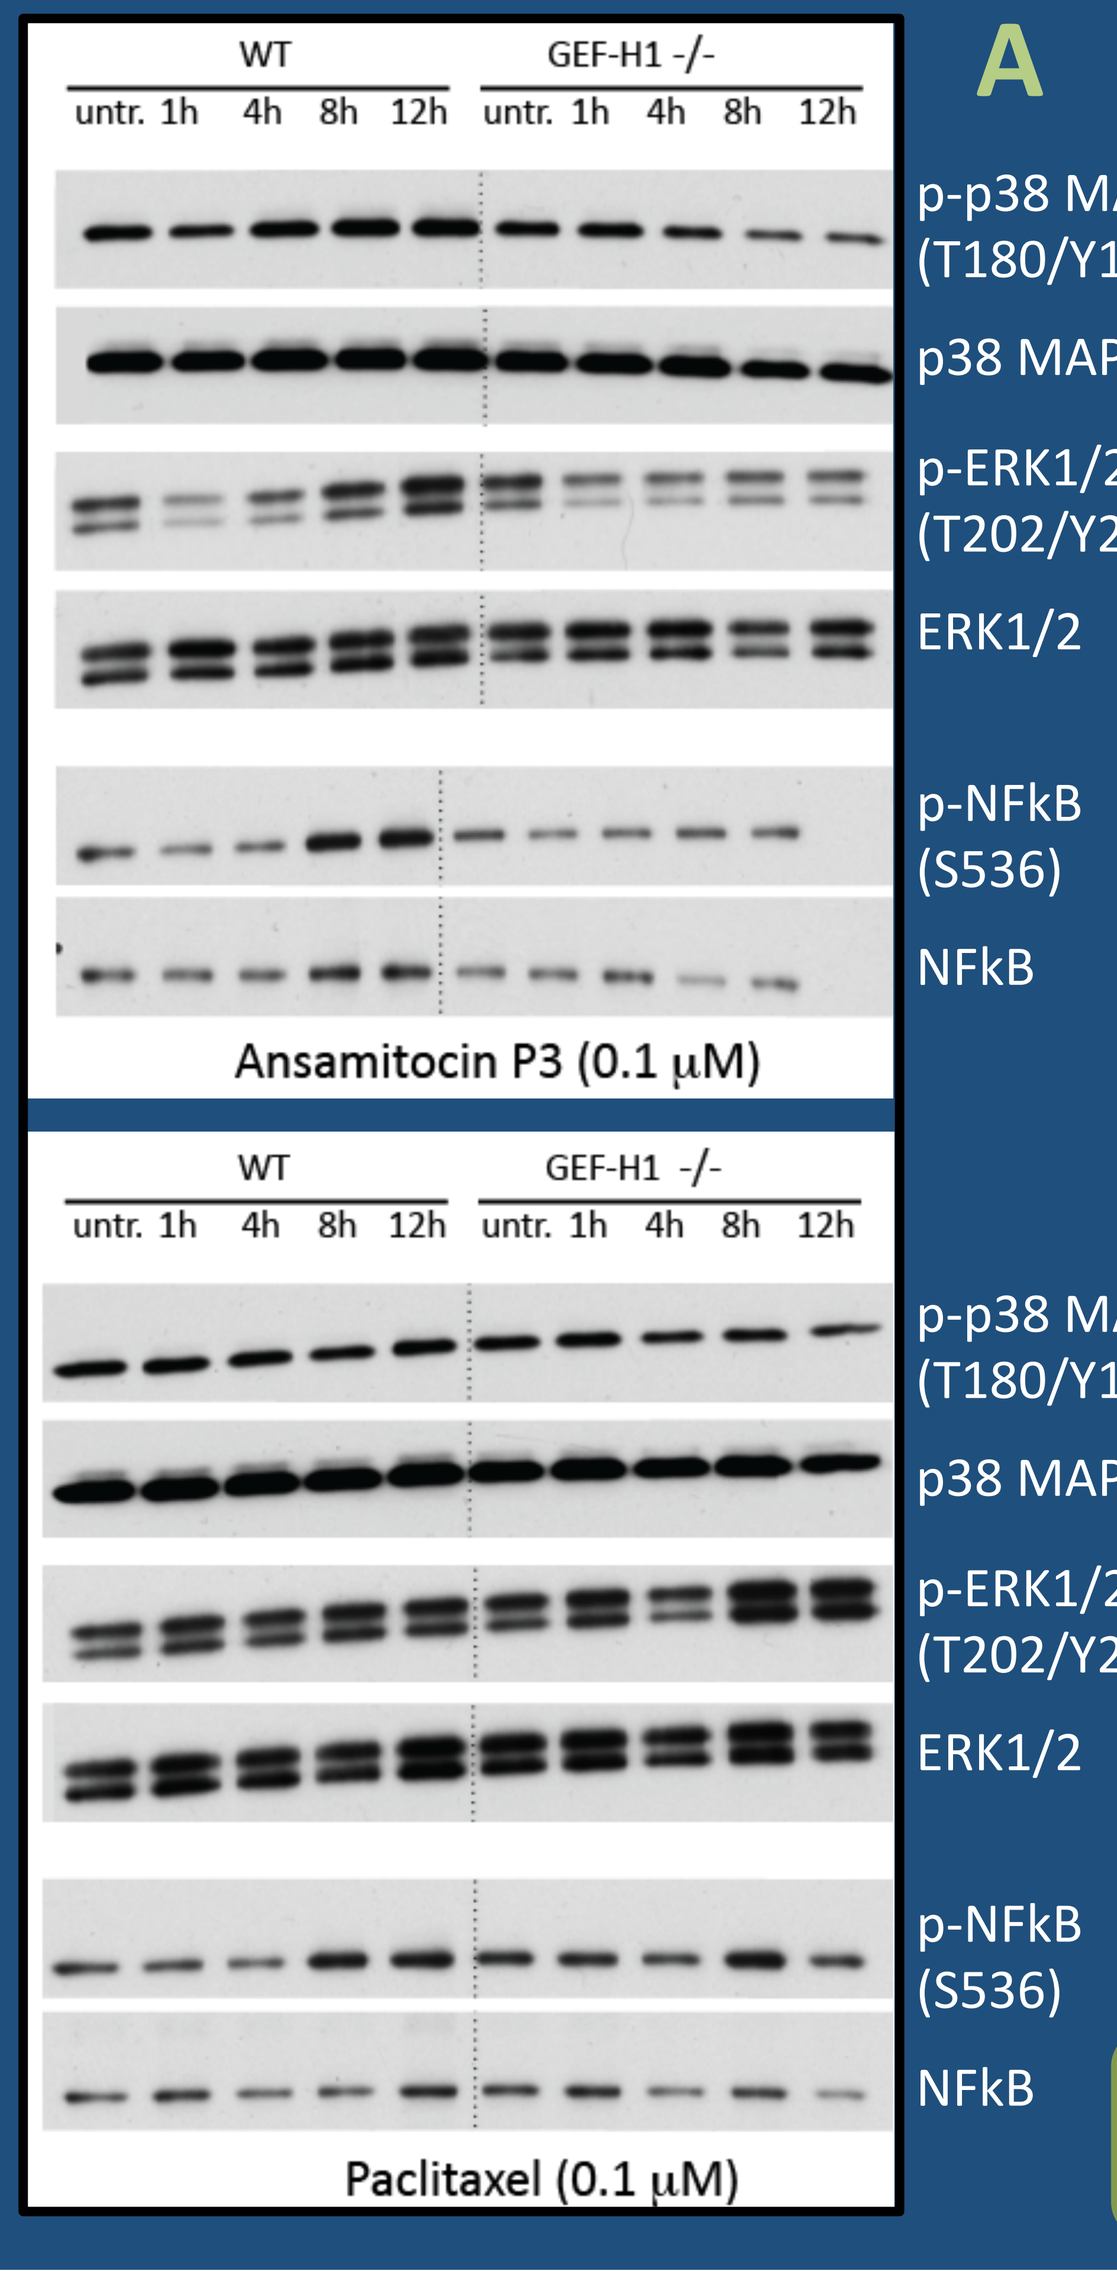
WT
GEF-H1 -/-
untr. 1h 4h 8h 12h
p-p38 MAPK (T180/Y182)
p38 MAPK
p-ERK1/2 (T202/Y204)
ERK1/2
p-NFkB (S536)
NFkB
Ansamitocin P3 (0.1 µM)
Paclitaxel (0.1 µM)

B

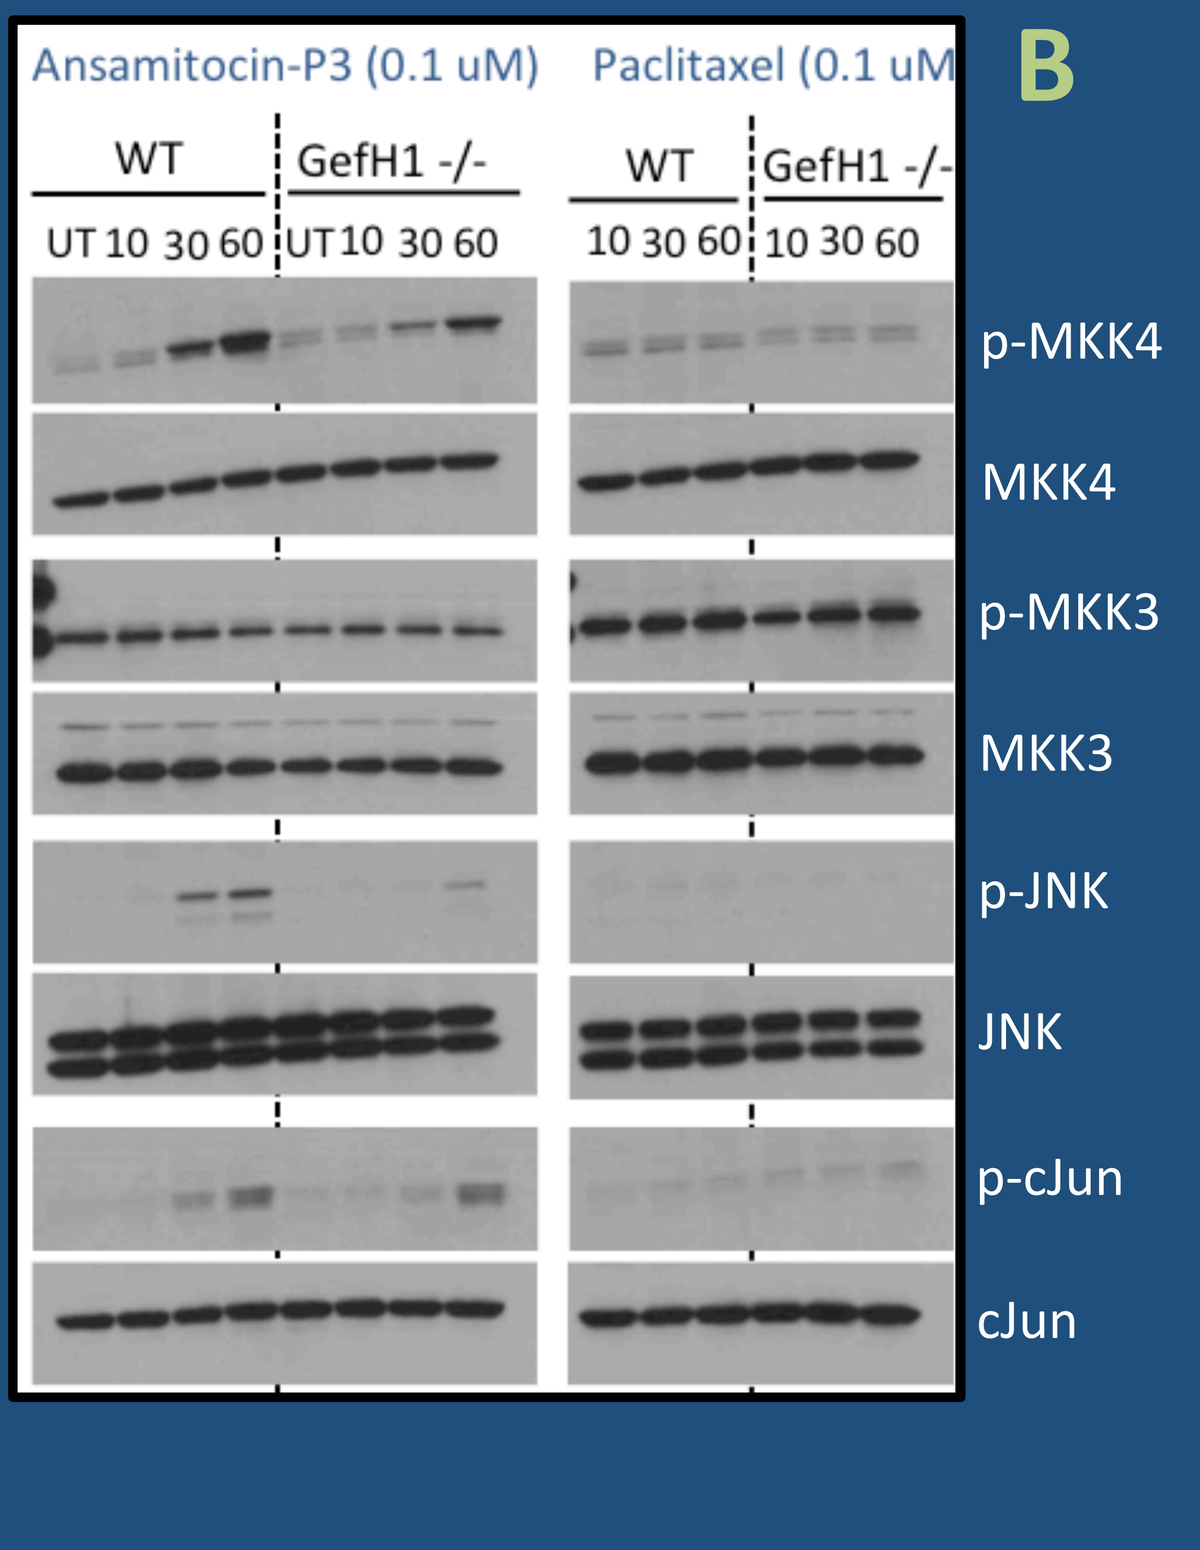
Ansamitocin-P3 (0.1 uM)
Paclitaxel (0.1 uM)
WT
GefH1 -/-
UT 10 30 60
p-MKK4
MKK4
p-MKK3
MKK3
p-JNK
JNK
p-cJun
cJun

Fig 7: Lysates from WT or GEFH1-/- BMDCs treated with AnsaP3 or Paclitaxel were probed for phospho or total protein. Although ERK and NFkB activation was GEFH1 dependent **(A)** it was observed in both AnsaP3 and Paclitaxel. However, MKK4, JNK and cJun remained uniquely activated in AnsaP3 **(B)** and was GEFH1 dependent.

GEFH1 is critically involved in MDA-induced DC activation and function

A B C D

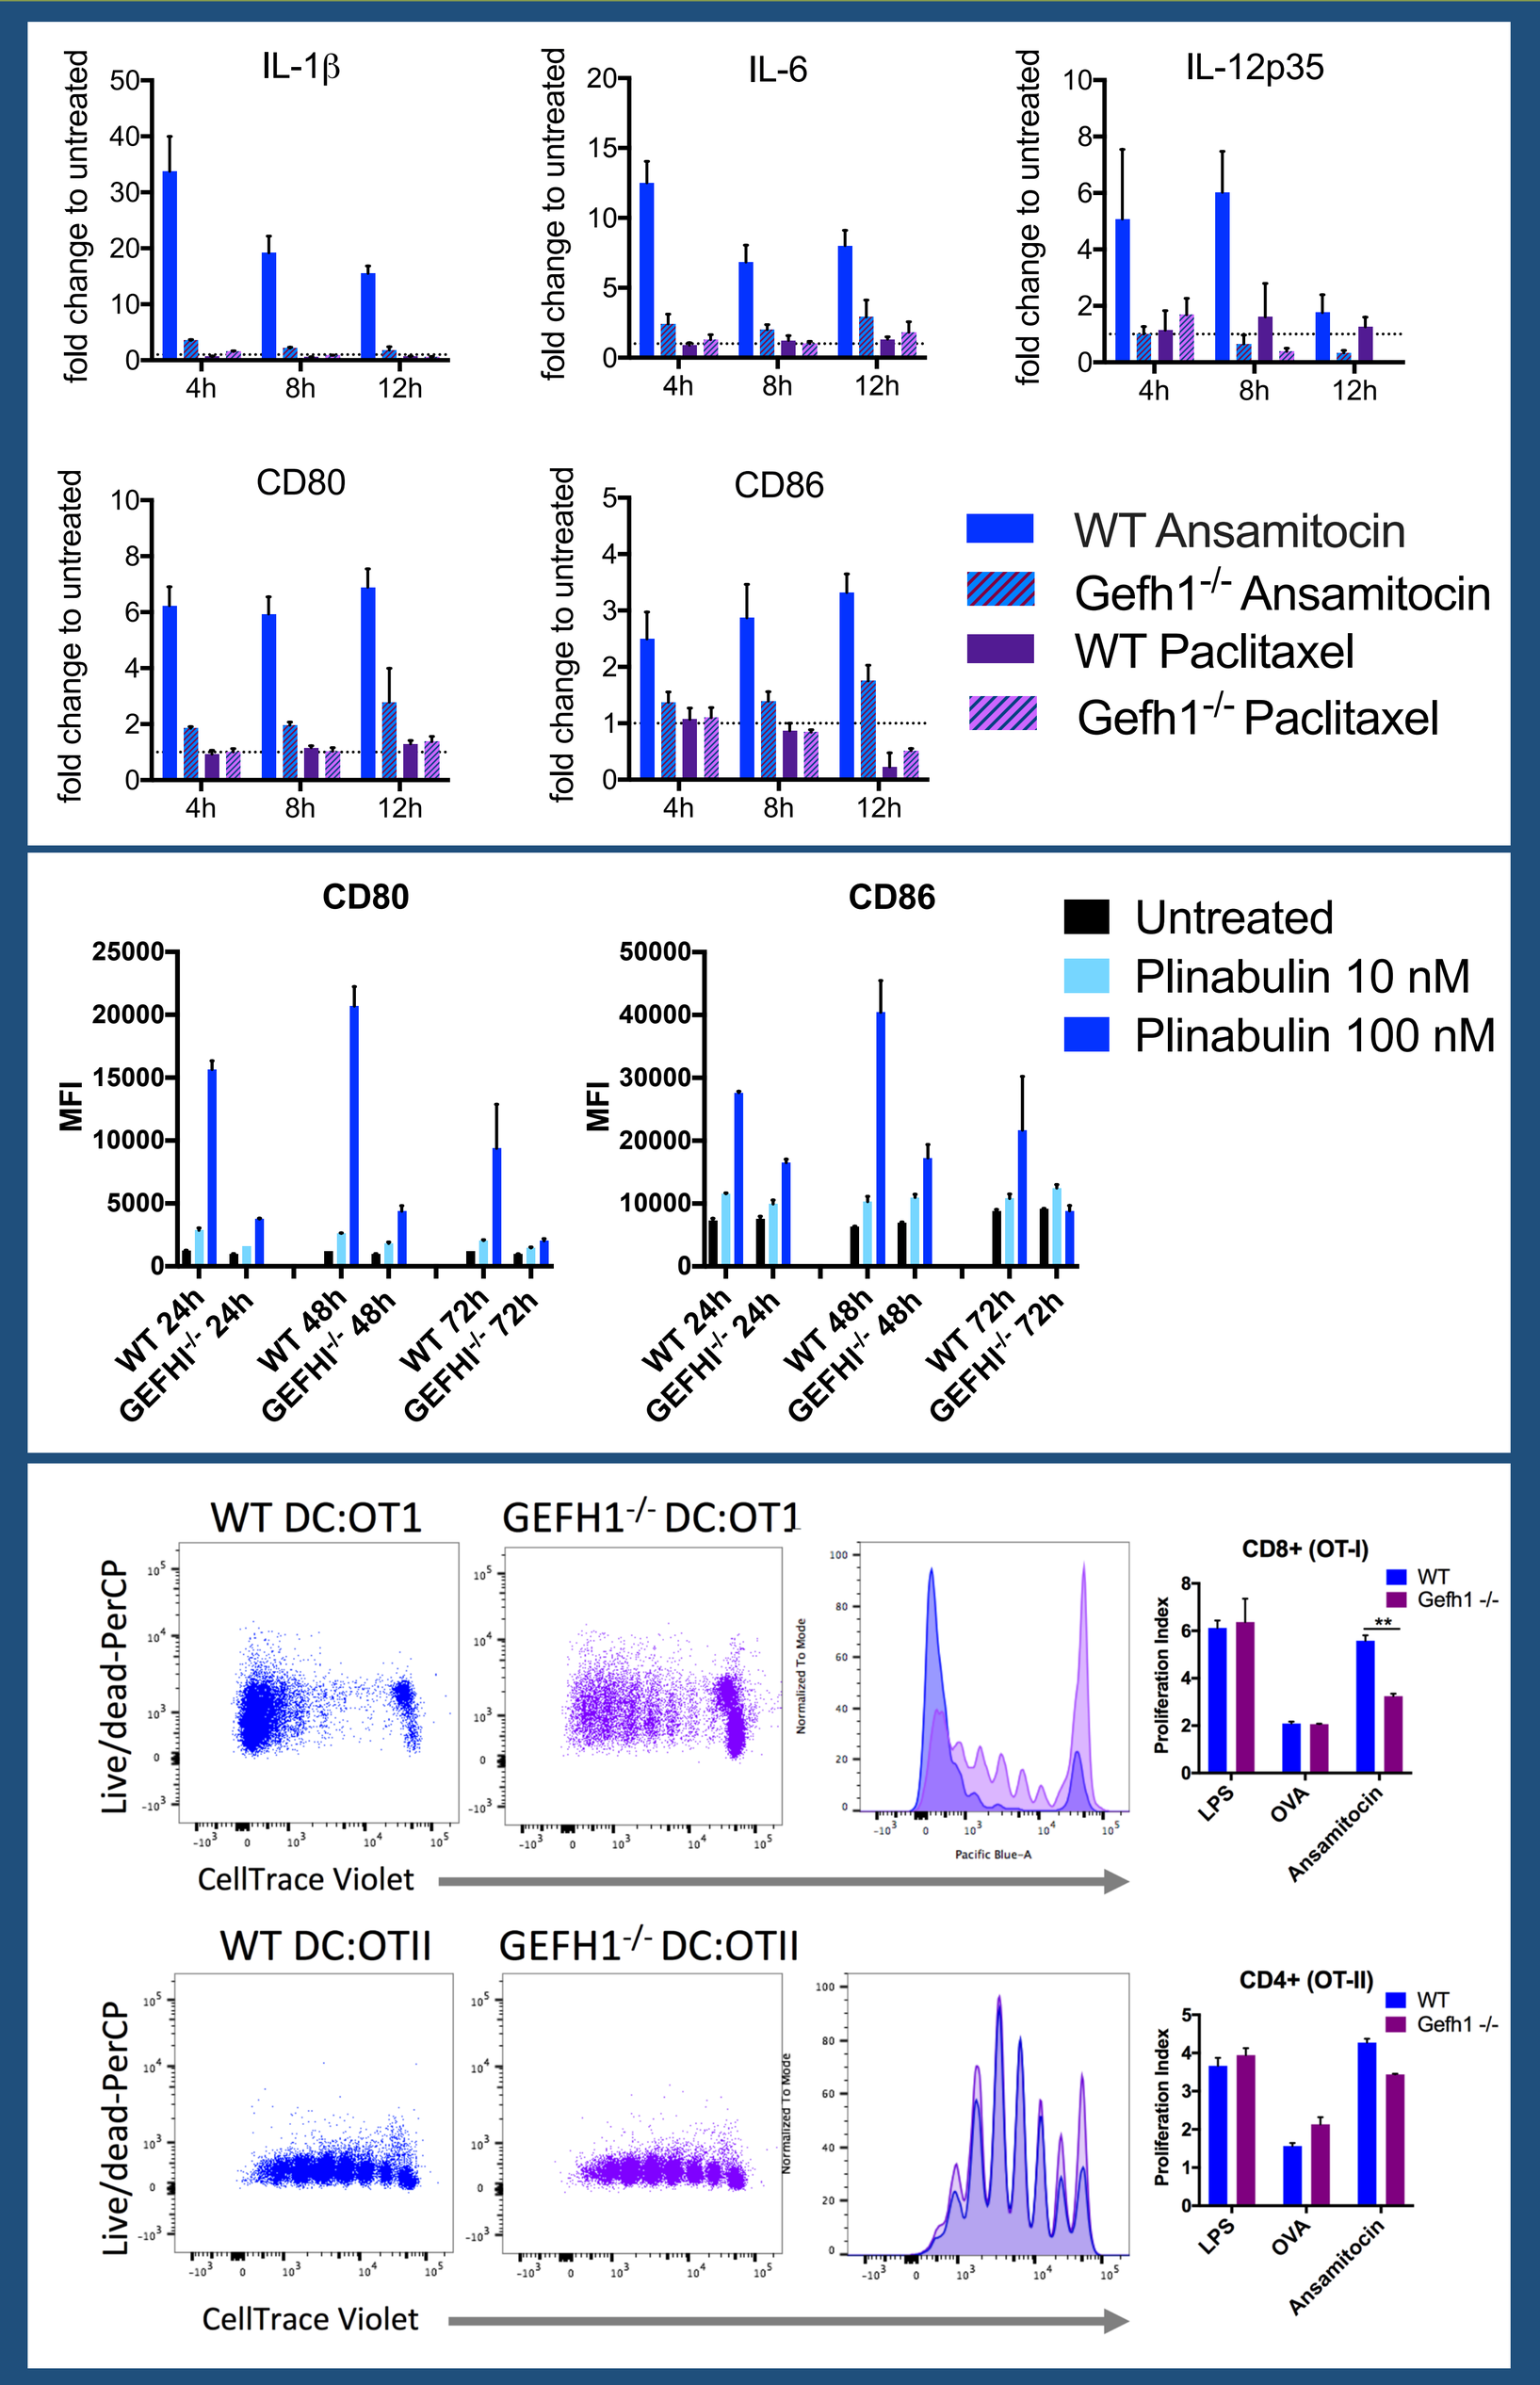
IL-1β
IL-6
IL-12p35
CD80
CD86
fold change to untreated
4h 8h 12h
WT Ansamitocin
Gefh1-/- Ansamitocin
WT Paclitaxel
Gefh1-/- Paclitaxel
Untreated
Plinabulin 10 nM
Plinabulin 100 nM
MFI
WT 24h
GEFHI-/- 24h
WT 48h
GEFHI-/- 48h
WT 72h
GEFHI-/- 72h
WT DC:OT1
GEFH1-/- DC:OT1
WT DC:OTII
GEFH1-/- DC:OTII
Live/dead-PerCP
CellTrace Violet
CD8+ (OT-I)
CD4+ (OT-II)
Proliferation Index
LPS
OVA
Ansamitocin

Fig 8: In GEFH1-/- BMDCs **(A)** and XS106 DC cells **(B)** the MDAs AnsaP3 and Plinabulin were unable to induce DC activation. Reduced activation of GEFH1-/- BMDCs lead to suboptimal expansion of OVA-specific CD8 T cells **(C)**. No effect on CD4 T cells was observed **(D)**. Together, this suggests that GEFH1 is critical for MDA-induced DC activation and function.

Model - MoA

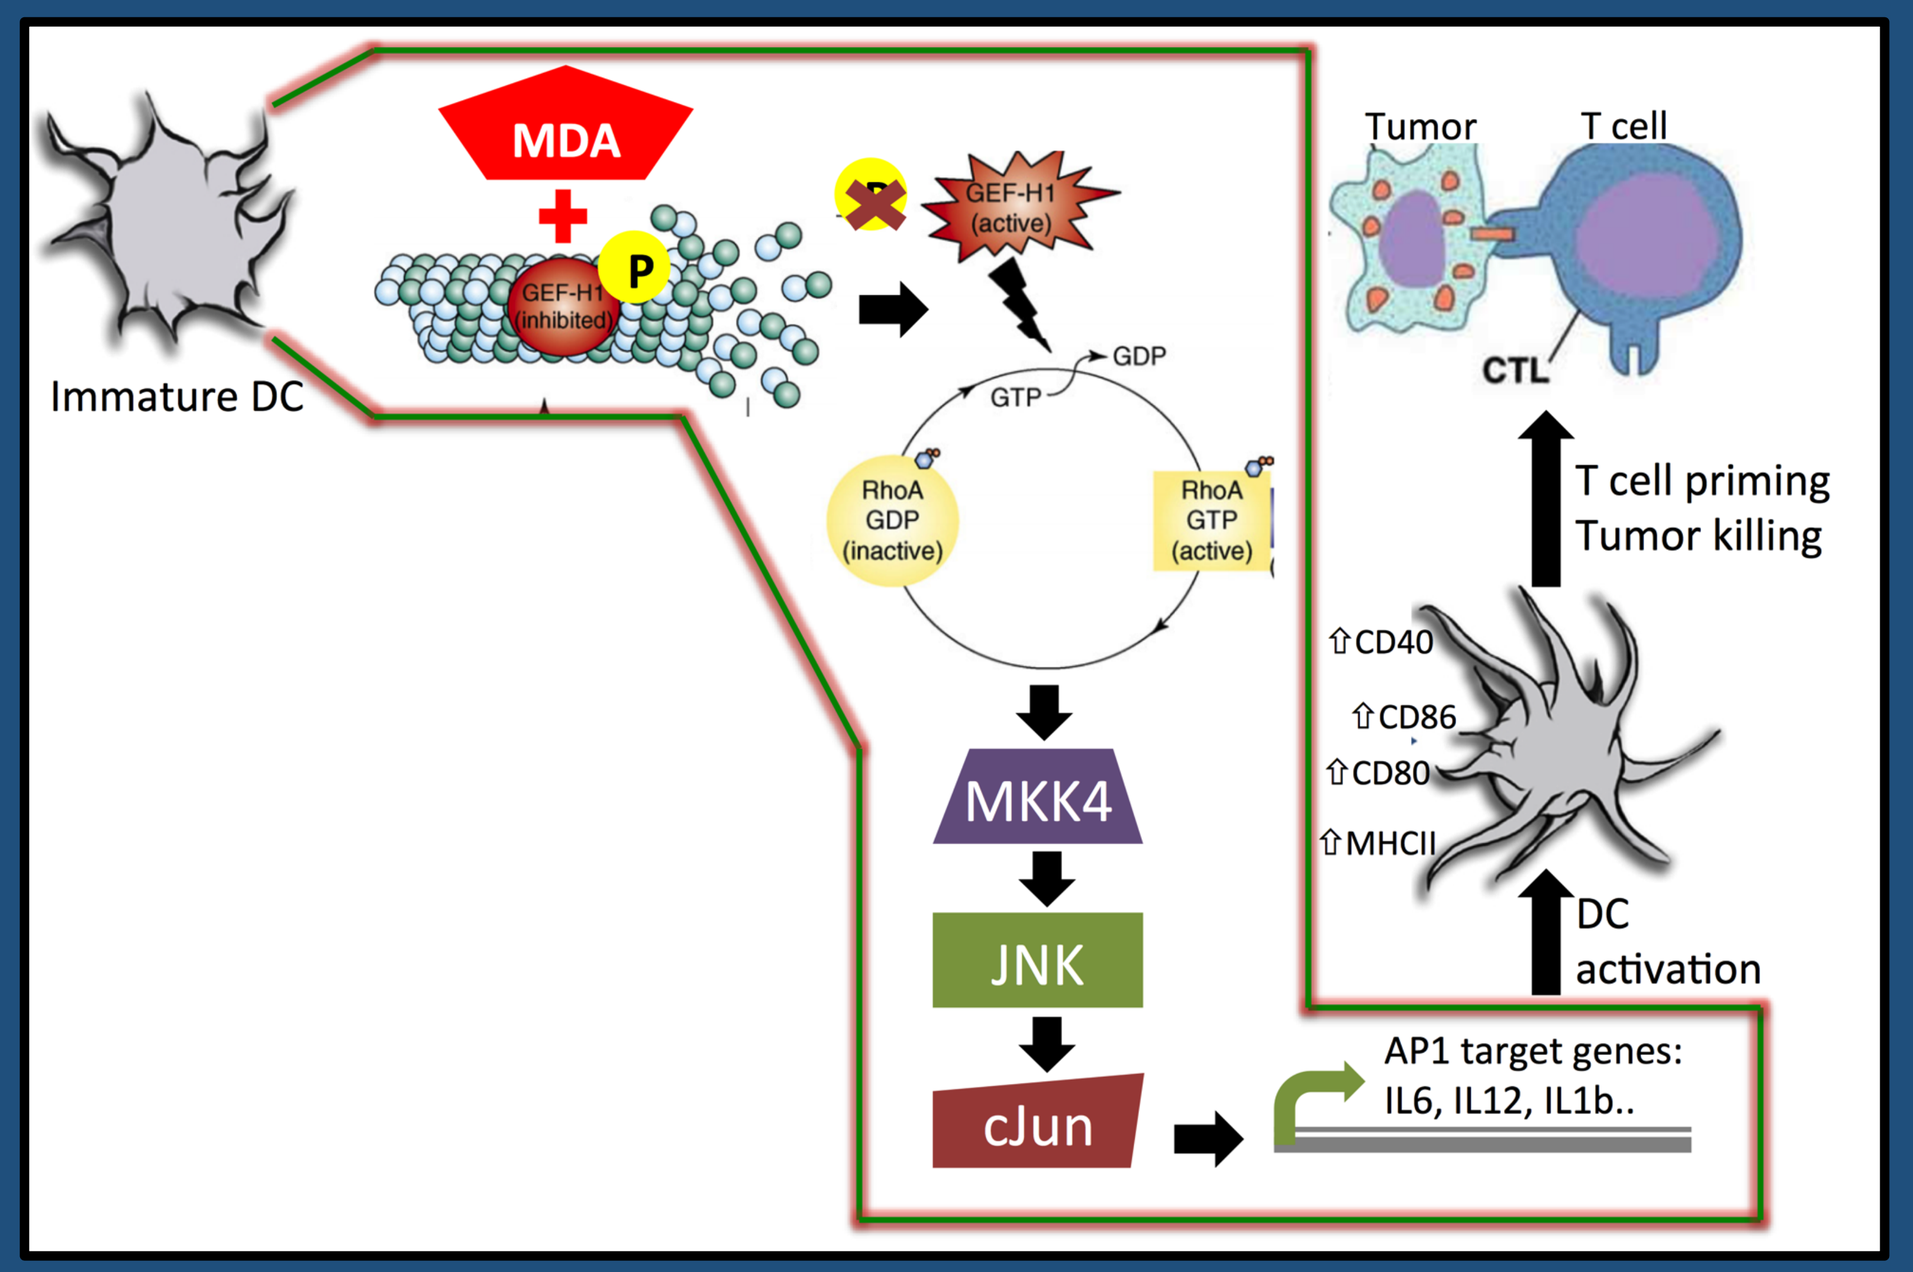
MDA
GEF-H1 (inhibited)
P
GEF-H1 (active)
Immature DC
GTP
GDP
RhoA GDP (inactive)
RhoA GTP (active)
MKK4
JNK
cJun
AP1 target genes: IL6, IL12, IL1b..
Tumor
T cell
CTL
T cell priming
Tumor killing
CD40
CD86
CD80
MHCII
DC activation

Acknowledgements: *The work is supported by BeyondSpring Pharmaceuticals, Wilhelm Sander Stiftung, Swiss National Fund International Travel grant, Huggenberger-Bischoffs Stiftung and Bio-Techne travel grant.* ***Contact:*** *abhishek.kashyap@unibas.ch*