Plinabulin, a Novel Small Molecule in Development for Chemotherapy-Induced-Neutropenia (CIN) Prevention, Mobilizes CD34+ Cells through a Mechanism of Action Different from G-CSF and from CXCR4 Inhibition

Douglas W. Blayney, Stephan Ogenstad, Yuankai Shi, Lihua Du, Ken Lloyd, Lan Huang, Ramon W. Mohanlal: Stanford Cancer Institute, Stanford, CA; Statogen Consulting, LLC, Zebulon, NC; Department of Medical Oncology, Beijing Key Laboratory of Clinical Study on Anticancer Molecular Targeted Drugs, National Cancer Center/Cancer Hospital, Chinese Academy of Medical Sciences and Peking Union Medical College, Beijing, China; Wanchun Bulin Pharmaceuticals Limited, Dalian, China; BeyondSpring Pharmaceuticals, New York, NY

BeyondSpring PHARMACEUTICALS

Plinabulin Overview:
- **Small Molecule**
- **Given by IV infusion, on the same day of the chemotherapy**
- **More than 300 Patient Data from Phase I,II,III**
- **Currently in Phase III for CIN and NSCLC**

Plinabulin is a small molecule activator of GEFH1,and represents a novel signaling pathway leading up to activation of Dendritic Cells. Plinabulin is Developed for Chemotherapy-Induced Neutropenia (CIN) and as an anti-agent in NSCLC.

- **CD34+ mobilization is achieved with G-CSF and CXCR4-inhibitors (Plerixafor).**
- **Plinabulin is not a G-CSF and does not inhibit CXCR4 (19% inhibition at 10μM)**
- **We evaluated whether Plinabulin mobilizes CD34+ progenitor cells**

NSCLC Plinabulin Anti-Cancer Activity Study NPI-2358-101

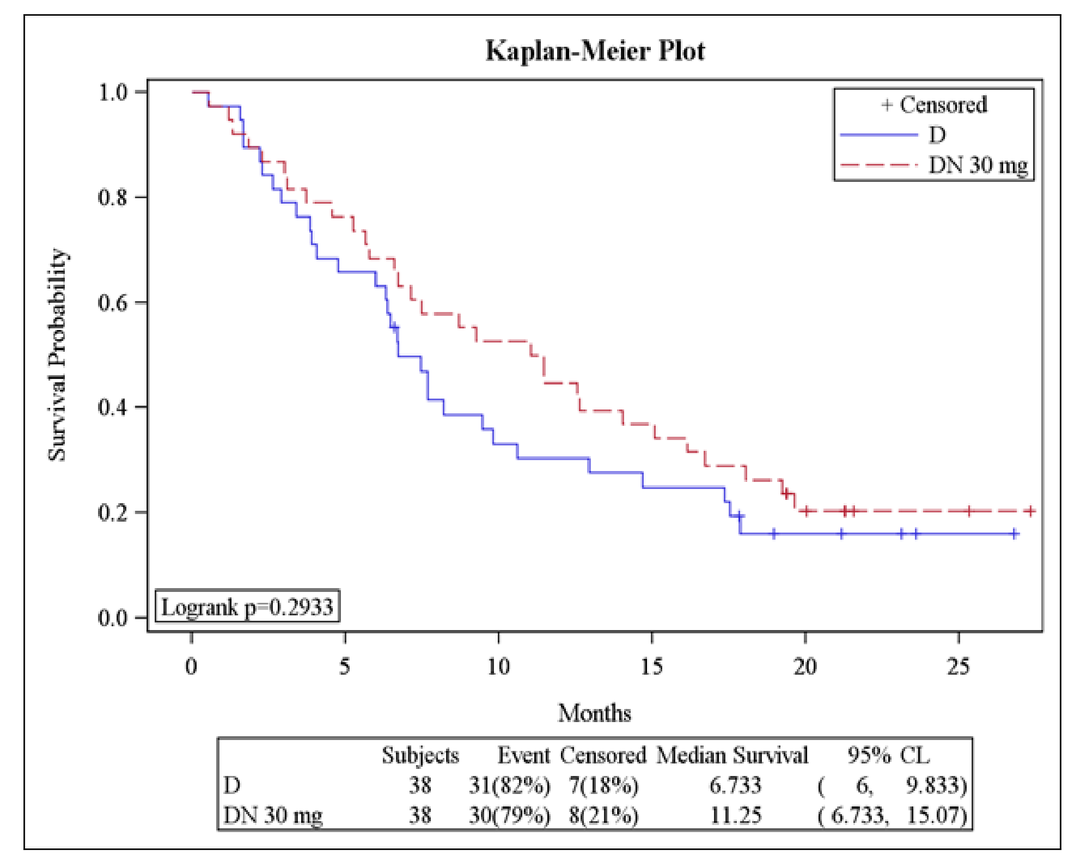
Kaplan-Meier Plot
+ Censored
D
DN 30 mg
Survival Probability
Logrank p=0.2933
Months
Subjects Event Censored Median Survival 95% CL
D 38 31(82%) 7(18%) 6.733 (6, 9.833)
DN 30 mg 38 30(79%) 8(21%) 11.25 (6.733, 15.07)

	Plinabulin + Docetaxel (DN)	Docetaxel alone (D)
	N=38	N=38
mOS	11.3 M	6.7 M
	P = 0.29	
DOR**	12.7 M	1.0 M
	P<0.05	
ORR	18.4%	10.5%
PFS	3.7 M	2.9 M

Plinabulin Mechanism of Action

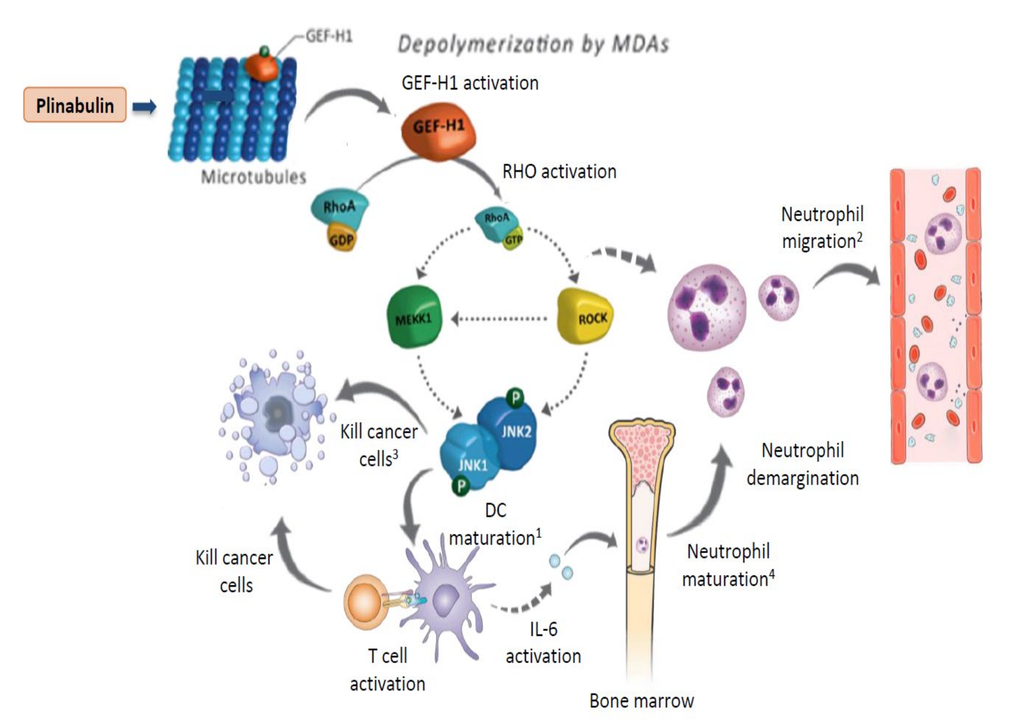
Plinabulin
Depolymerization by MDAs
GEF-H1 activation
Microtubules
RHO activation
Neutrophil migration
Kill cancer cells
DC maturation
Neutrophil demargination
Neutrophil maturation
T cell activation
IL-6 activation
Bone marrow

Plinabulin Activates Dendritic Cells

Methods

CIN Trials Study BPI-2358-105 (NCT03102606) and BPI-2358-106 (NCT03294577).

Study BPI-2358-105 (NCT03102606):
Phase 2/3, Multicenter, Randomized, Double Blind Study to Evaluate Duration of Severe Neutropenia with Plinabulin Versus Pegfilgrastim in Patients with Solid Tumors Receiving Docetaxel Myelosuppressive Chemotherapy (Protective 1)

Study BPI-2358-106 (NCT03294577):
Phase 2/3, Multicenter, Randomized Study to Evaluate Plinabulin versus Pegfilgrastim in Reducing the Duration of Severe Neutropenia in Breast Cancer Patients Receiving Myelosuppressive Chemotherapy with Docetaxel, Doxorubicin, and Cyclophosphamide (TAC) (Protective 2)

Assessments:

- Absolute Neutrophil Count (ANC) at predose and multiple timepoints postdose with Chemotherapy +/- Plinabulin in Cycle 1
- Bone Pain was assessed with a validated questionnaire (Bone Pain Inventory (Short Form) on predose and multiple timepoints postdose with Chemotherapy +/- Plinabulin in Cycle 1
- Blood CD34+ cell counts were measured at predose and multiple timepoints postdose with Docetaxel +/- Plinabulin
 - CD34+ measurements were obtained in at least 9 pts on both D0 and D8 for each Plin dose. No blood draws for CD34+ cells were taken after D8.

Study Design:

These were phase 2 portions of Phase 2/3 Studies, and were designed as a multicenter, open label, randomized study.
Study 105: A total of N=55 NSCLC patients were randomly assigned to the following arms:
Arm 1: Docetaxel (75 mg/m²) + Pegfilgrastim (6 mg)
Arm 2: Docetaxel (75 mg/m²) + Plinabulin (20 mg/m²)
Arm 3: Docetaxel (75 mg/m²) + Plinabulin (10 mg/m²)
Arm 4: Docetaxel (75 mg/m²) + Plinabulin (5 mg/m²)
Study 106: A total of N=72 BC patients were randomly assigned to the following combination arms:
Arm a: TAC + Pegfilgrastim **(6 mg)**
Arm b: TAC + Pegfilgrastim **(6 mg)** + Plinabulin (20 mg/m²)
Arm c: TAC +Pegfilgrastim **(3 mg)** + Plinabulin (20 mg/m²)
Arm d: TAC+ Pegfilgrastim **(1.5 mg)** + Plinabulin (20 mg/m²)

Target Patient Population:
Study 105: Patients with advanced or metastatic non-Small Cell Lung Cancer (NSCLC) after failing platinum-based therapy.
Study 106: Breast Cancer (BC) stage I,II,III patients who are candidate for adjuvant or neoadjuvant TAC

Results

Study 105

Figure 1. D0 & D8 CD34+ count with Plinabulin Doses

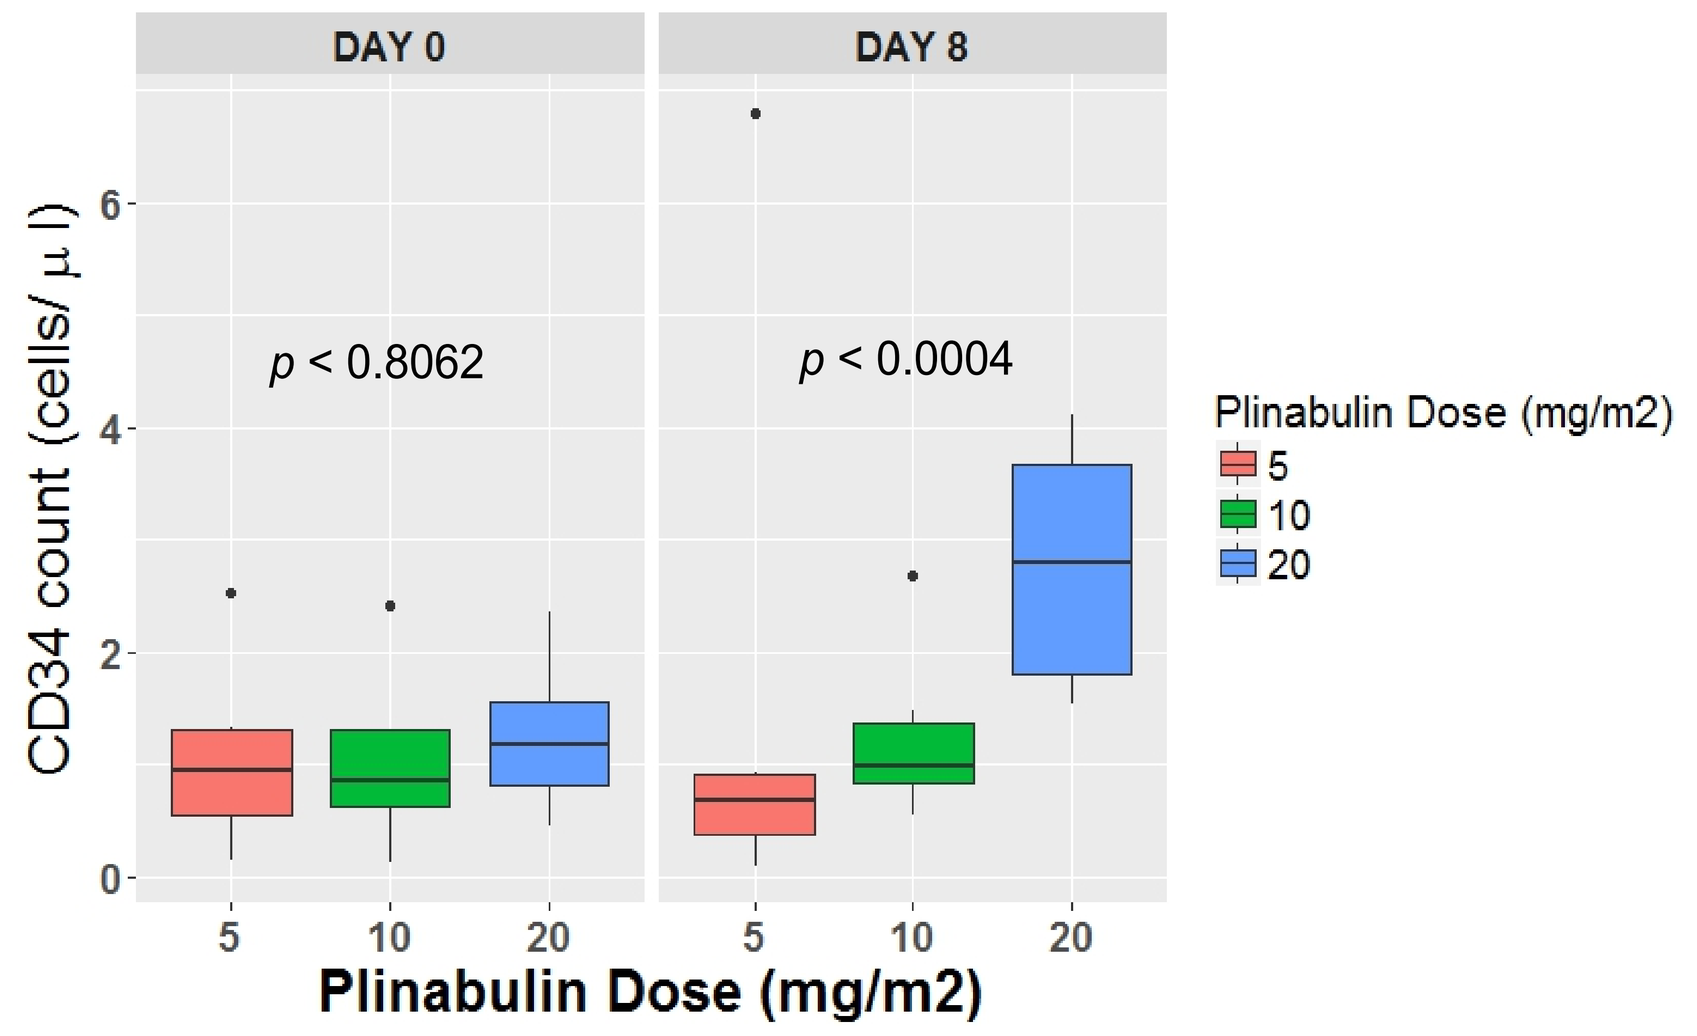
Figure 1
DAY 0
DAY 8
p < 0.8062
p < 0.0004
CD34 count (cells/ μ l)
Plinabulin Dose (mg/m2)
5
10
20
Plinabulin Dose (mg/m2)

Figure 2 Neutropenia by Grade (95% CI) (Cycle 1)

Key Finding:

1. Plinabulin and Pegfilgrastim: Equally Effective for Gr 4 Neutropenia

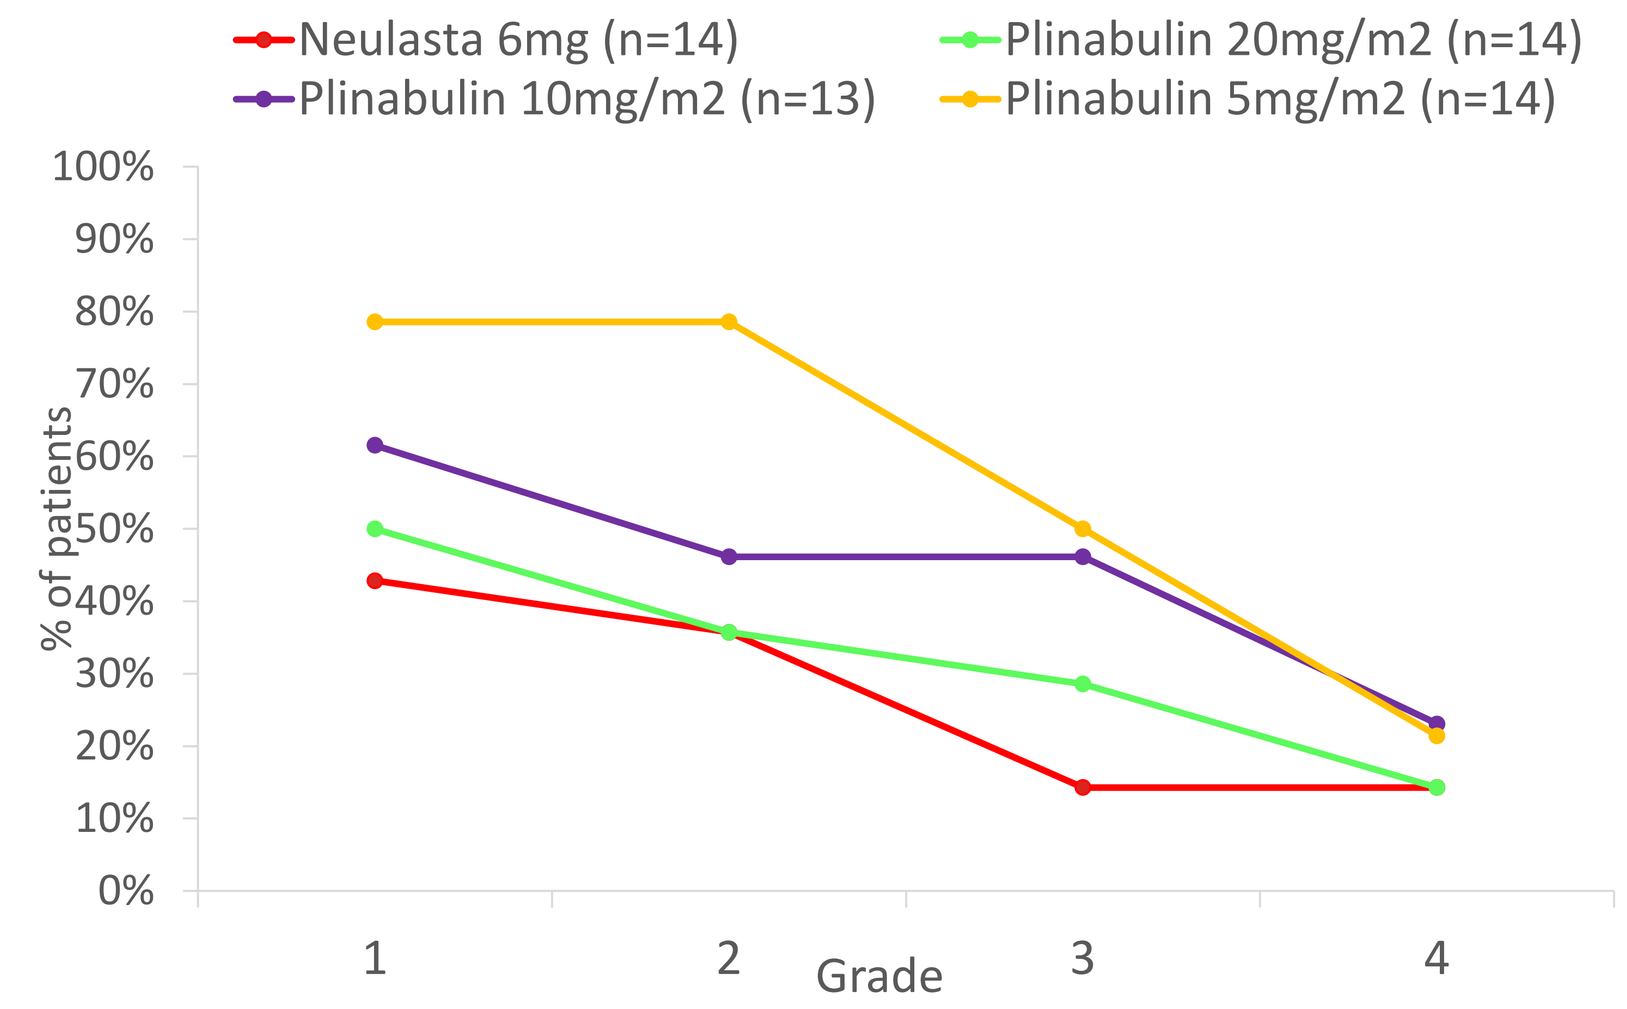
Neulasta 6mg (n=14)
Plinabulin 20mg/m2 (n=14)
Plinabulin 10mg/m2 (n=13)
Plinabulin 5mg/m2 (n=14)
% of patients
Grade

Figure 3. Absolute Neutrophil Count (95% CI) (Cycle 1)

Key Findings:

1. Plinabulin: Mean Absolute Count Remained in Normal Range
2. Pegfilgrastim: Mean Absolute Neutrophil Counts Overshoots Upper Limit of Normal

Neutrophil Count Cycle 1

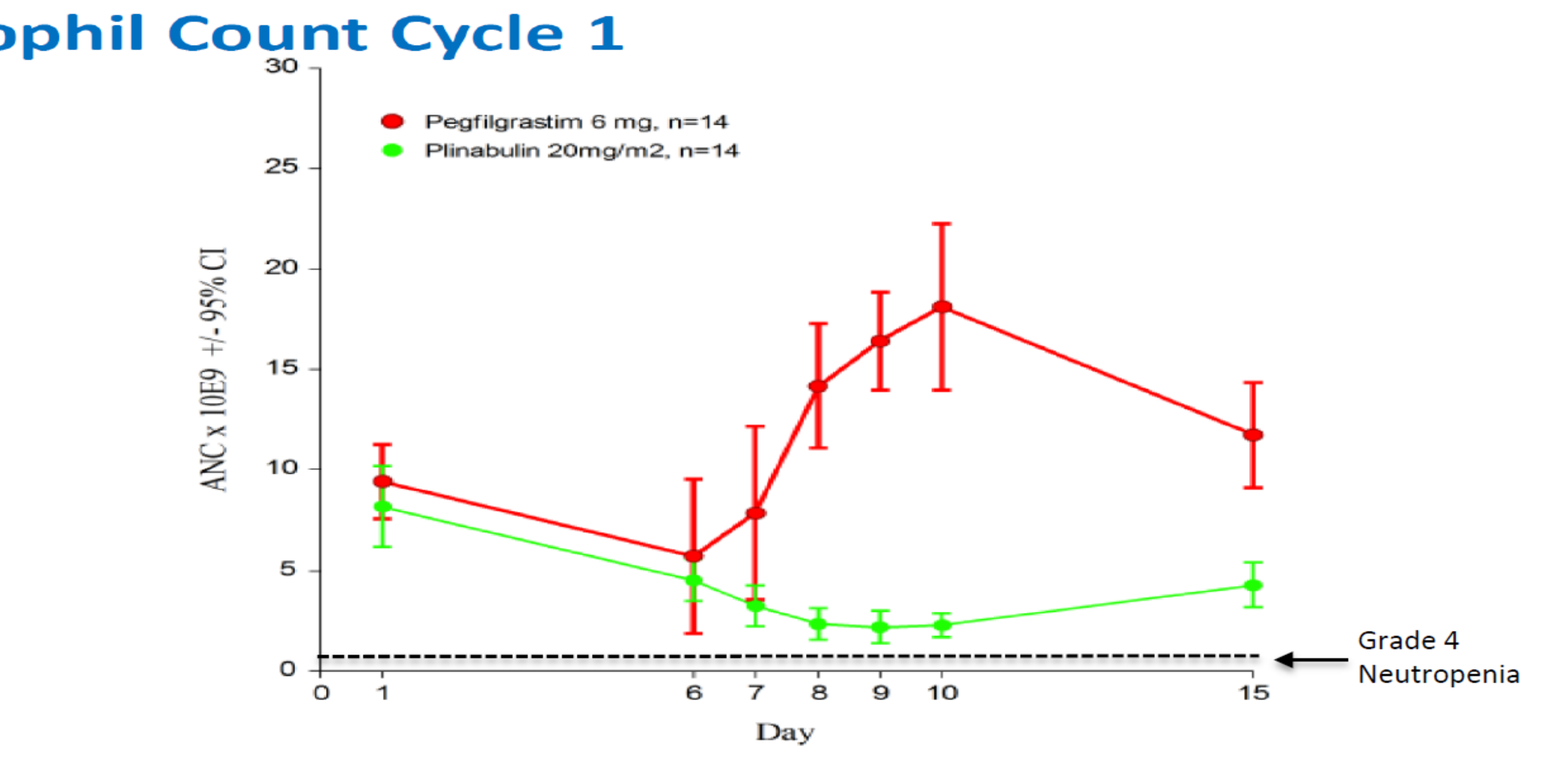
Pegfilgrastim 6 mg, n=14
Plinabulin 20mg/m2, n=14
ANC x 10E9 +/- 95% CI
Grade 4 Neutropenia
Day

	Baseline	D6	D7	D8	D9	D10	D15
Peg	9.436	5.717	7.853	14.169	16.410	18.122	11.746
Plin 20mg/m2	8.185	4.512	3.233	2.335	2.183	2.274	4.267
P-value Peg vs. 20mg/m2	NS	NS	NS	<0.0001	<0.0001	<0.0001	0.0001

Figure 4. Bone Pain with Plinabulin and Pegfilgrastim.

Key Finding:

1. Plinabulin caused less Bone Pain vs Pegfilgrastim

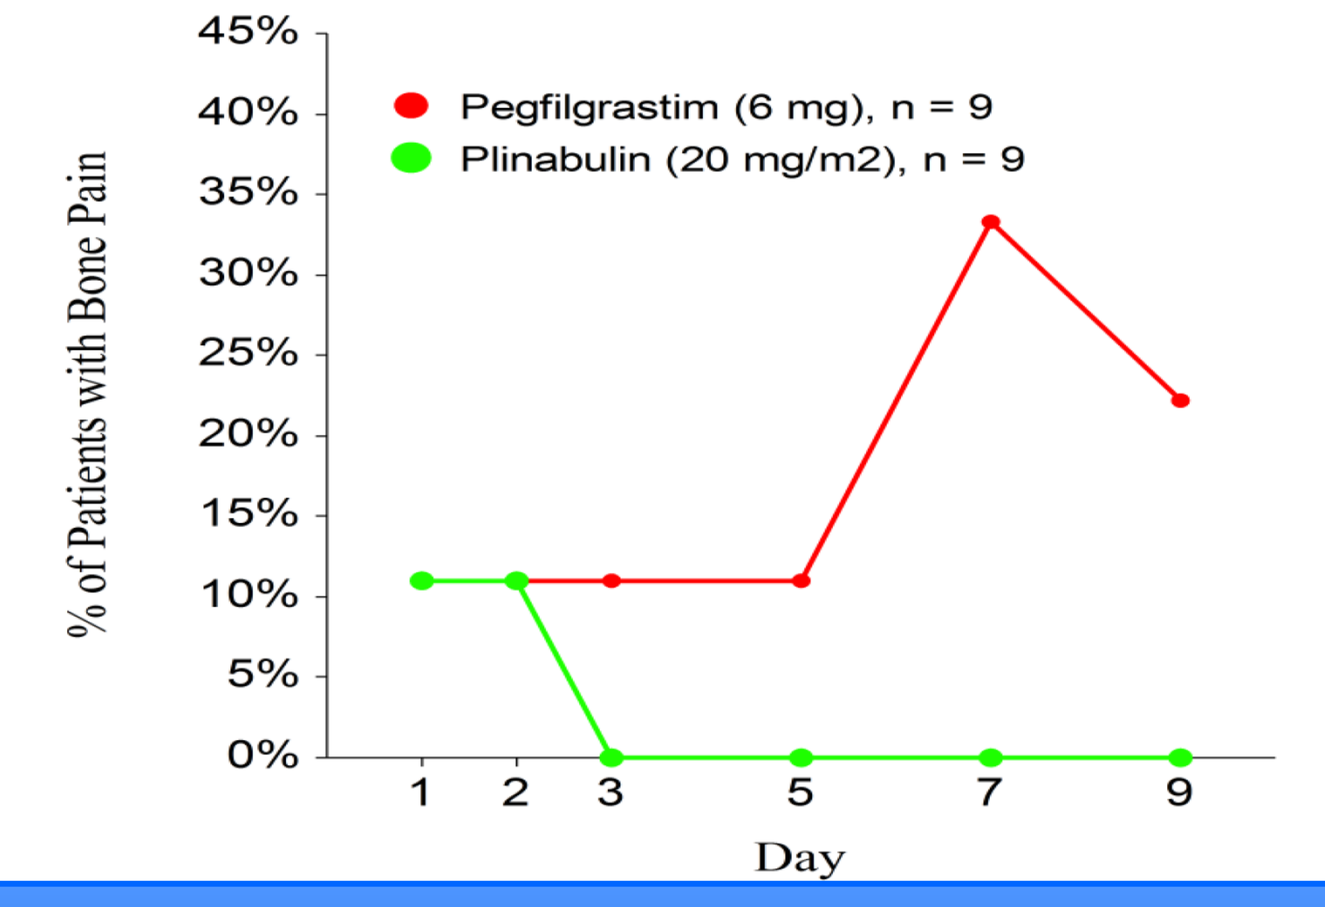
Pegfilgrastim (6 mg), n = 9
Plinabulin (20 mg/m2), n = 9
% of Patients with Bone Pain
Day

Study 106

Figure 5. Plinbulin and Neulasta Have Complimentary Protection against TAC-Induced Neutropenia

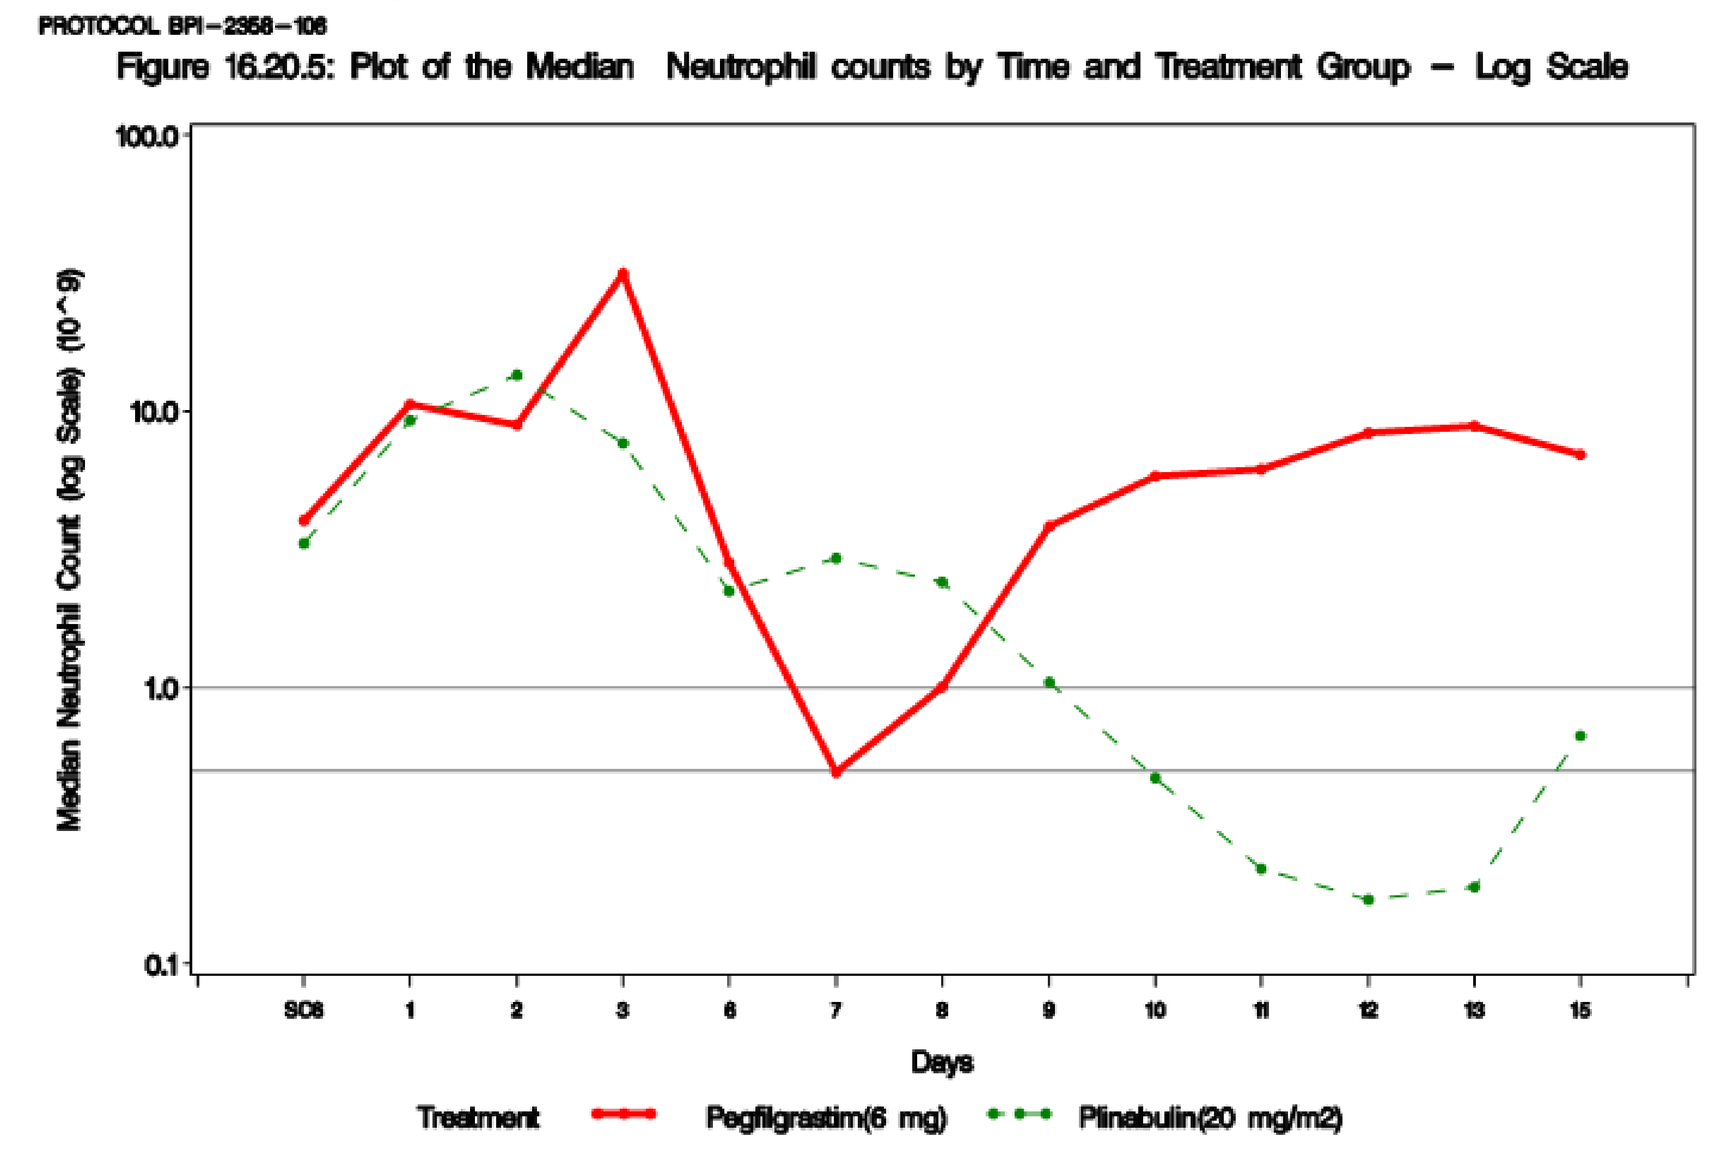
PROTOCOL BPI-2358-106
Figure 16.20.5: Plot of the Median Neutrophil counts by Time and Treatment Group – Log Scale
Median Neutrophil Count (log Scale) (10^9)
Days
Treatment
Pegfilgrastim(6 mg)
Plinabulin(20 mg/m2)

Figure 6. Plinabulin Combined with Neulasta Show Enhanced Protection against TAC-Induced Neutropenia

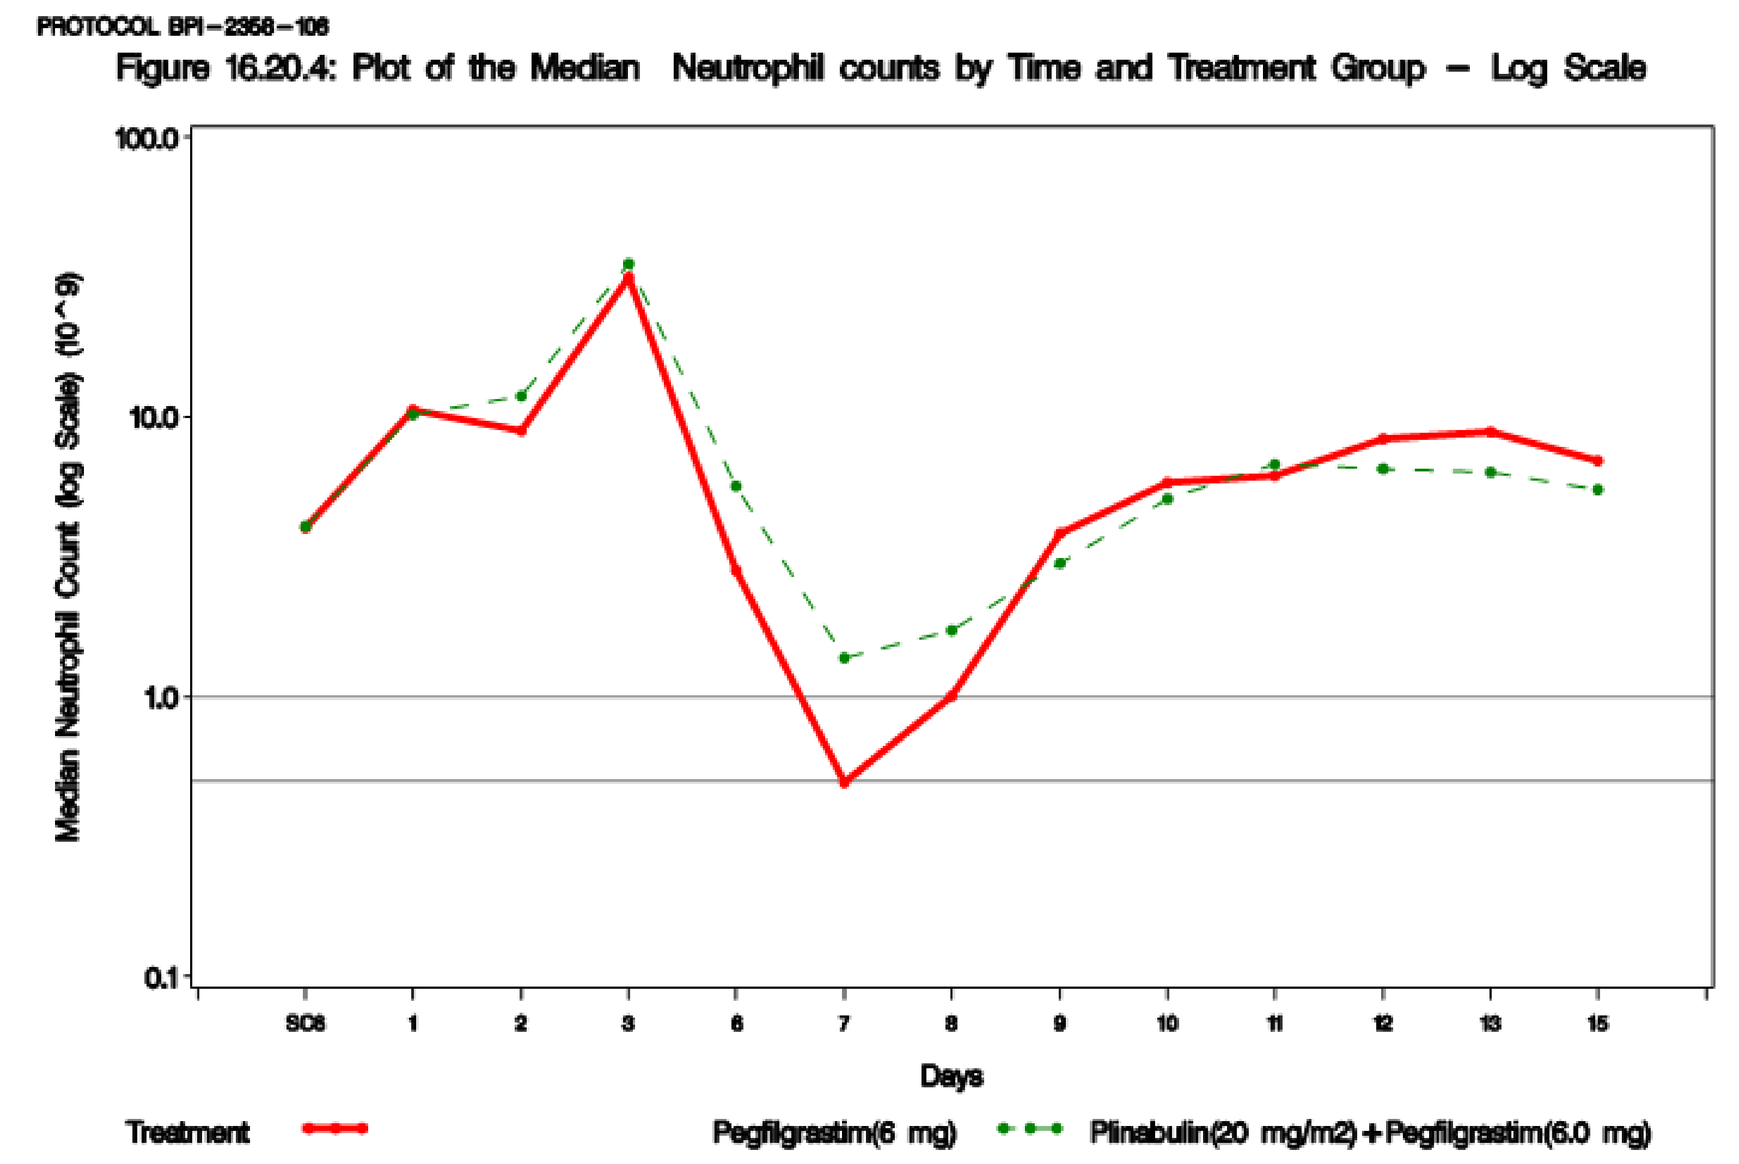
PROTOCOL BPI-2358-106
Figure 16.20.4: Plot of the Median Neutrophil counts by Time and Treatment Group – Log Scale
Median Neutrophil Count (log Scale) (10^9)
Days
Treatment
Pegfilgrastim(6 mg)
Plinabulin(20 mg/m2)+Pegfilgrastim(6.0 mg)

Figure 7. Plinabulin Combined with Neulasta almost Eradicated Bone Pain Induced by Pegfilgrastim

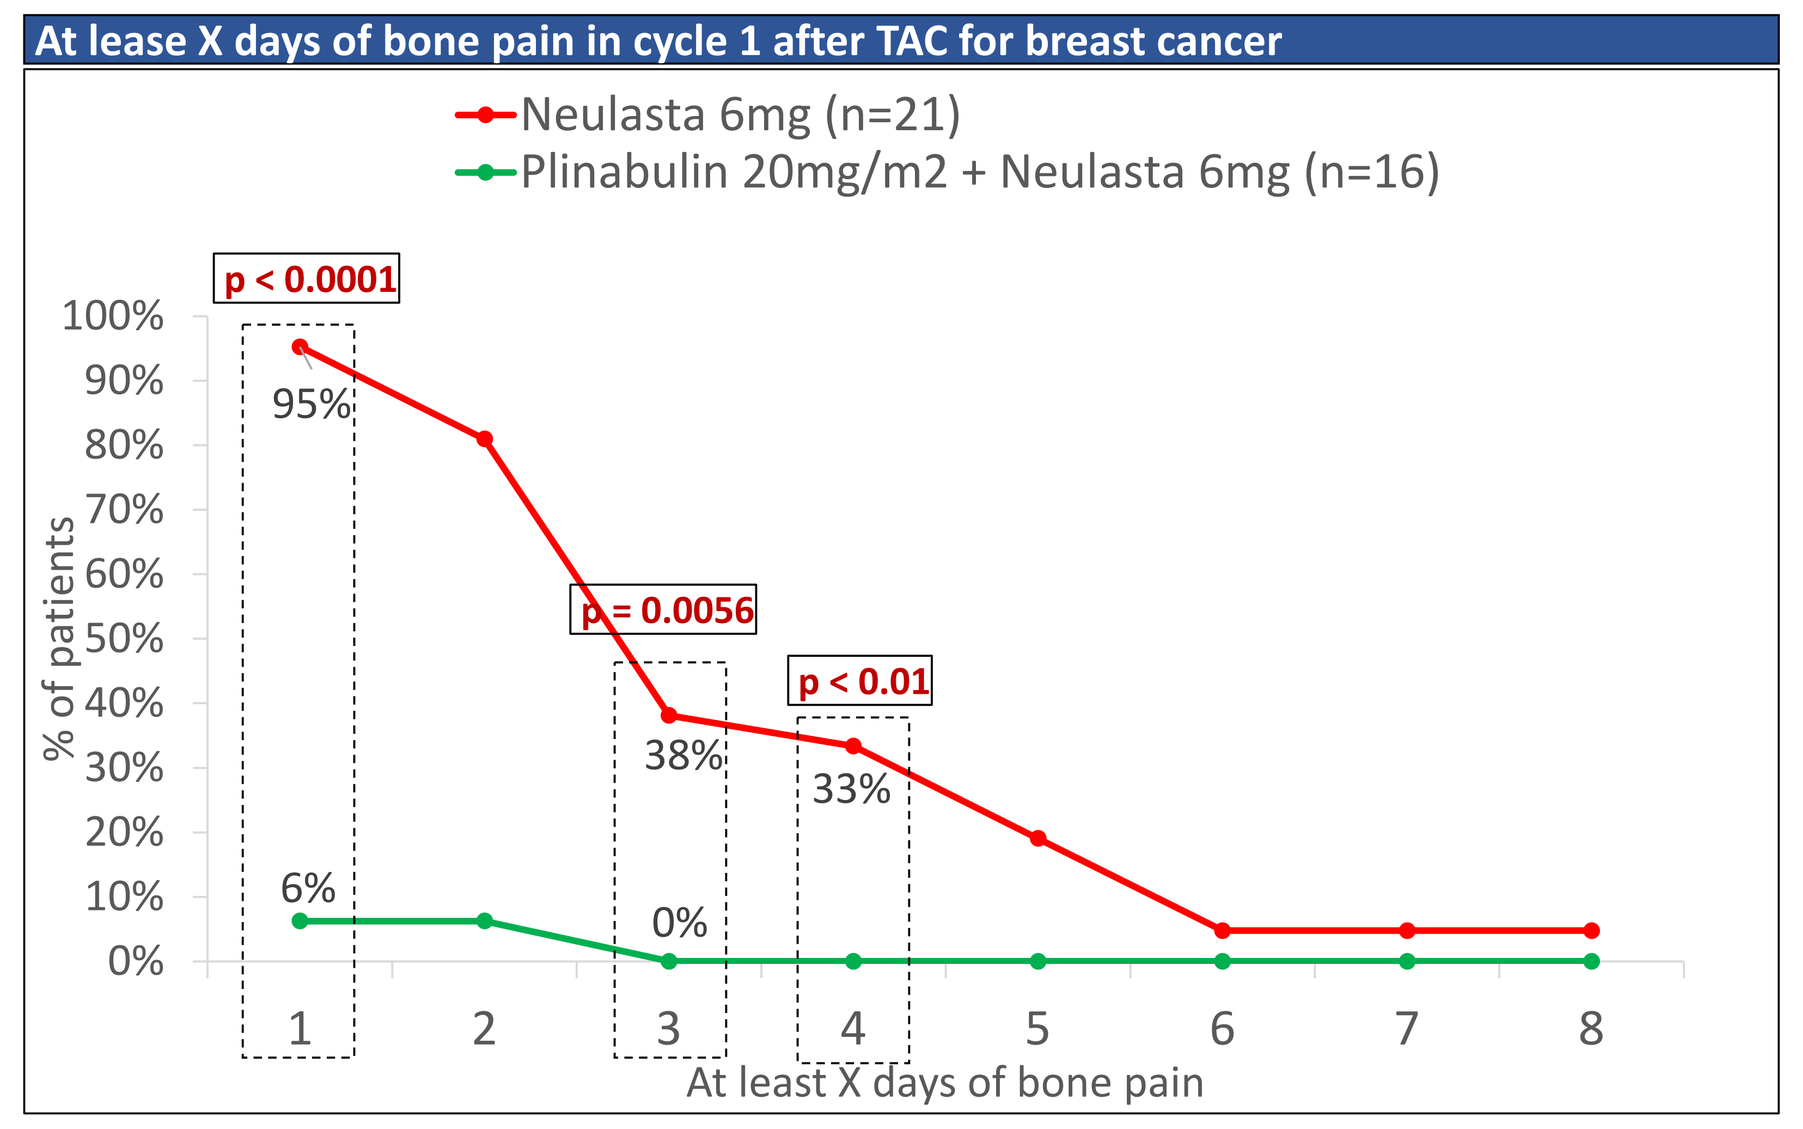
At lease X days of bone pain in cycle 1 after TAC for breast cancer
Neulasta 6mg (n=21)
Plinabulin 20mg/m2 + Neulasta 6mg (n=16)
p < 0.0001
p = 0.0056
p < 0.01
95%
38%
33%
6%
0%
% of patients
At least X days of bone pain

Table 1. Neulasta Alone, and Plinabulin / Neulasta Combination CIN and Bone Pain Summary Results

	Peg 6mg N=21	Peg 6mg+Plin N=16	Peg 3mg+Plin N=21	Peg 1.5mg+Plin N=14
Gr 3/4 frequency	81%	50%*	57%	86%
Gr 4 frequency	57%	38%	52%	57%
DSMN mean (±SD)	1.4 (±1) day	0.9 (±1.1) day	1.6 (±1.6) day	2.6 (±1.6) day
DSMN median	1 day	0.5 day	2 days	3 days
Nadir mean (±SD)	0.77 (±0.90) μ/L	1.15 (±0.94) μ/L	0.88 (±1.02) μ/L	0.76 (±0.97) μ/L
Bone Pain ≥1 day	95%	6%**	33%**	36%**
Bone Pain ≥4 day	33%	0%***	9.5%	7.1%

Plinabulin vs. Pegfilgrastim

Table 2. Plinabulin Superior Profile compared with Pegfilgrastim

Study 105 (phase 2): Plinabulin demonstrated a clear superiority profile against Neulasta, standard of care

Plinabulin demonstrates a clear superiority profile in cycle 1 after Docetaxel for NSCLC

	Neulasta 6mg	Plinabulin 20mg/m²
DSN (grade 4)	0.5 day	0.5 day
% neutropenia (grade 4)	14%	14%
% bone pain	Yes	No from day 3
Thrombocytopenia	Yes	No
Immune suppression	Yes	No
Anti-cancer	No	Yes

Study 106 (phase 2): Plinabulin / Neulasta Combo demonstrates a clear superiority profile against Neulasta, standard of care

Plinabulin / Neulasta Combo demonstrates a clear superiority profile after TAC for breast cancer

	Neulasta	Plinabulin / Neulasta Combo
DSN (grade 3/4)	Over 1 day	Less than 1 day
% neutropenia (grade 3/4)	High (> 80%)	Low (50%)
Median ANC nadir (10^9 cells/L)	0.47 (> 50% with grade 4 neutropenia)	1.00 (> 50% avoid grade 3/4 neutropenia)
% bone pain	Almost all	Limited
Immune suppression	Yes	Limited
Anti-cancer	No	Yes

Conclusion

- **Plinabulin Monotherapy is an Equally Effective Single-Dose-per-Cycle Agent as Pegfilgrastim for CIN**
- **With Combining Plinabulin to Pegfilgrastim:**
 - **Superior Efficacy for Neutropenia**
 - **Almost Eradicates Pegfilgrastim-Induced Bone Pain**
- **Plinabulin Mobilizes CD34+ Progenitor Cells through a MoA Independent from G-CSF of CXCR4**
- **Plinabulin can Potentially Mobilize CD34+ Cells from the Bone Marrow prior to HCT, in Particular in Patients Failing to Respond to G-CSF or G-CSF/Plerixafor**

Clinical trial supported by BeyondSpring, Inc., presented at ASH 2018

***Contact:* dblayney@stanford.edu**
rmohanlal@beyondspringpharma.com